3936

Efficacy of single agent radium-223 in the syngeneic MBT-2 bladder cancer bone growth model in mice

Tiina E. Kähkönen[1], Mari I. Suominen[1], Jenni H.E. Mäki-Jouppila[1], Birgitta Sjöholm[2], Ilmari Ahonen[3], Dominik Mumberg[4], Karl Ziegelbauer[4], Jussi M. Halleen[1], Sanna-Maria Käkönen[2], Arne Scholz[4]

[1] Pharmatest Services Ltd., Turku, Finland; [2]Aurexel Life Sciences Ltd., Askainen, Finland; [3]Vincit Oy, Turku, Finland; [4]Bayer AG, Pharmaceuticals, Berlin, Germany

INTRODUCTION

- Radium-223 dichloride (radium-223, Xofigo®) is a targeted alpha therapy which prolongs the survival of castration-resistant prostate cancer (CRPC) patients with symptomatic bone metastases without known visceral metastases.[1]
- As a calcium-mimetic, radium-223 selectively binds to hydroxyapatite and targets areas of high bone turnover such as bone metastases.
- Radium-223 inhibits disease progression by reducing tumor growth and tumor-induced pathological bone reaction in breast and prostate cancer mouse models (**Fig. 1**).[2, 3]
- The direct cytotoxic effects of radium-223 on local trabecular meshwork (TM) cells and disease-driving osteoblasts and osteoclasts have been established in immunodeficient models.[3]
- The effects of radium-223 on local immune cells are unknown, and these cells may also be involved in the antitumor efficacy of radium-223. However, investigation of the potential immune mode-of-action component of radium-223 requires successful development of immunocompetent TM models. Furthermore, these models would be needed for the testing of radium-223 in combination with immuno-oncological drugs.
- Here, we report the effects of radium-223 on the development and progression of osteolytic bone lesions and on survival in the syngeneic intratibial MBT-2 murine bladder cancer model in immunocompetent mice.

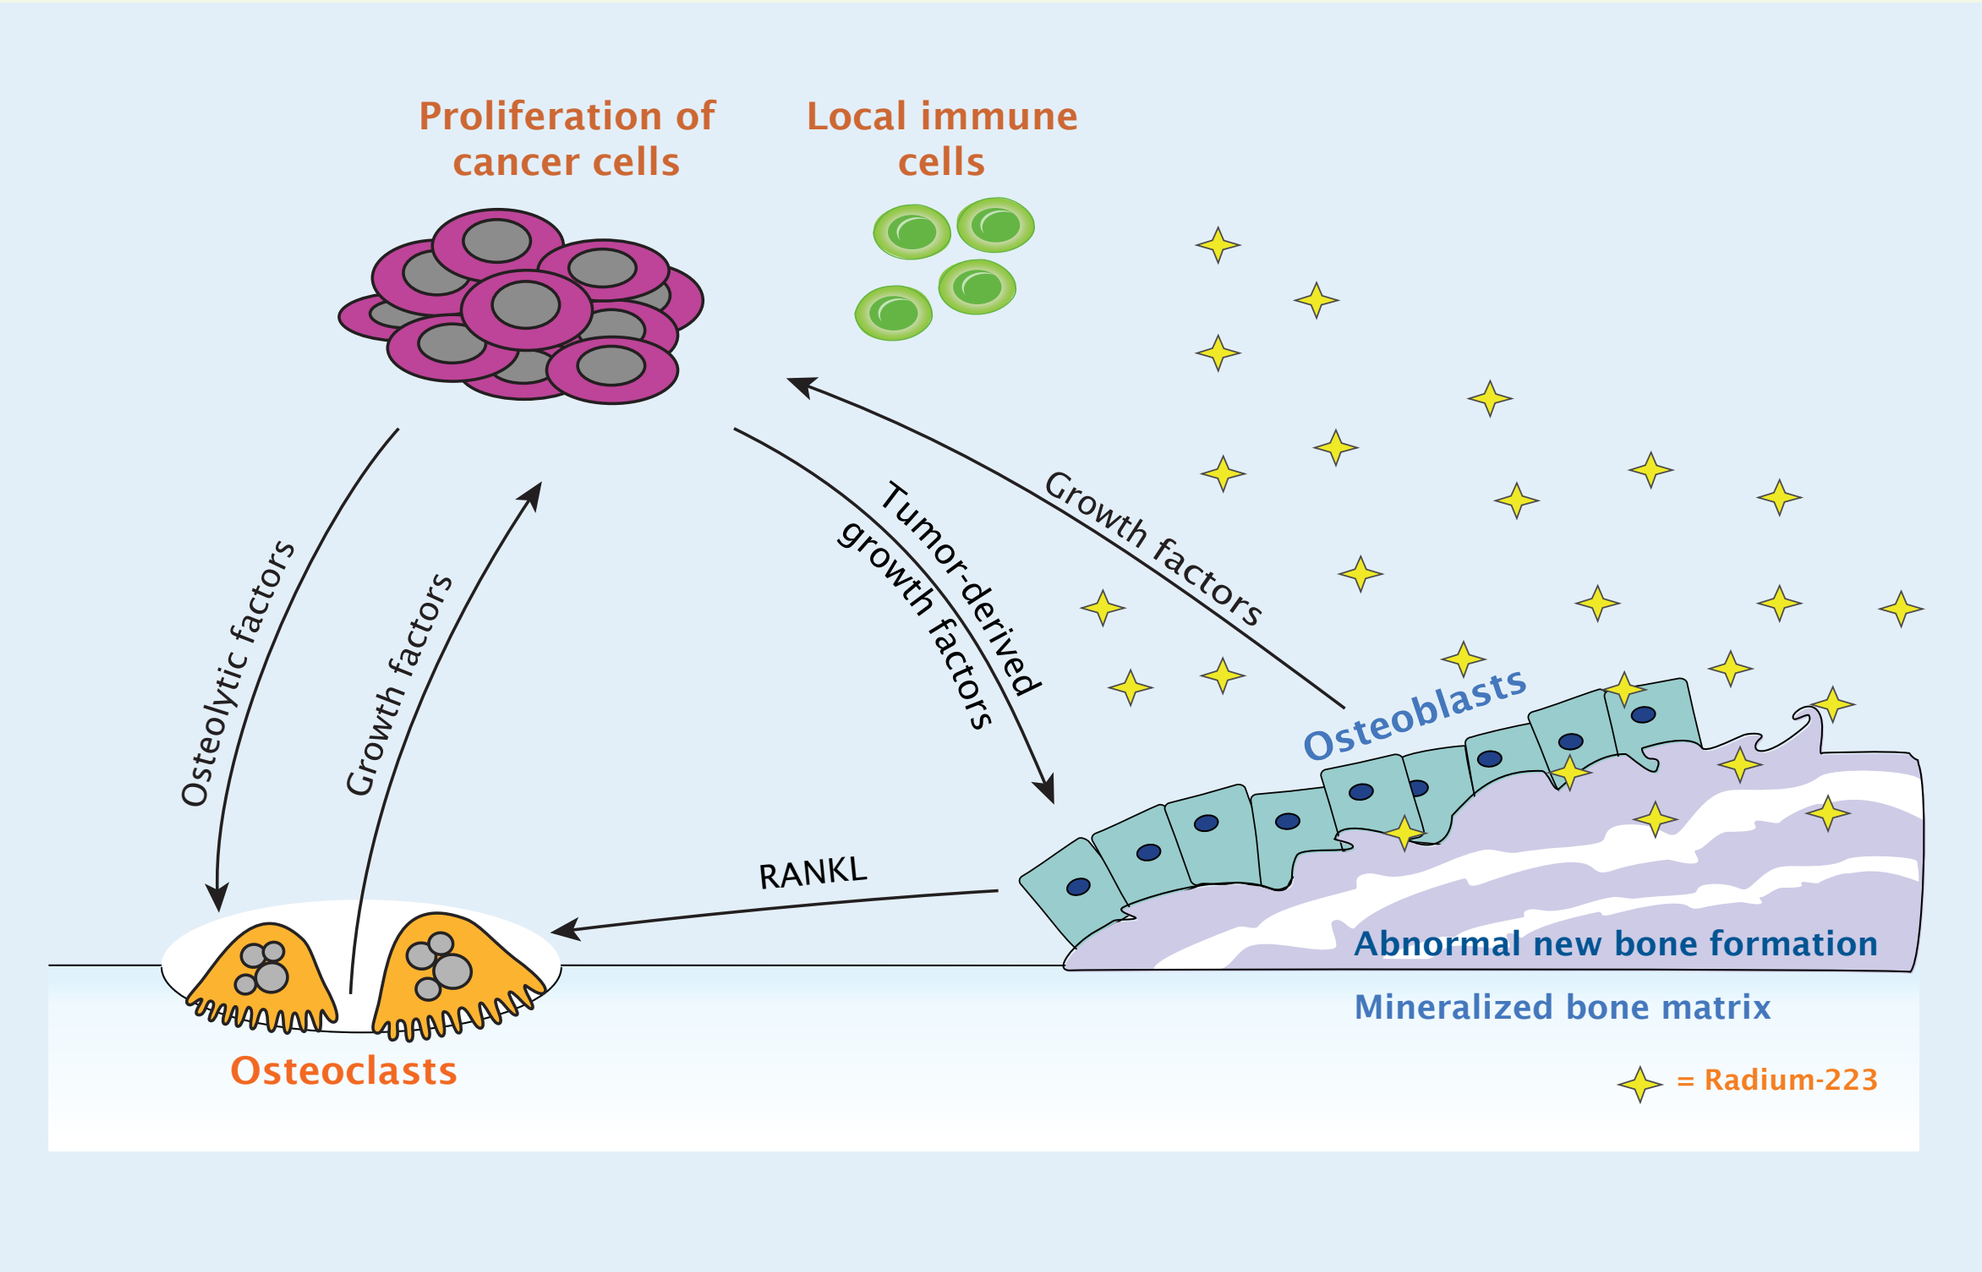
Proliferation of cancer cells
Local immune cells
Osteolytic factors
Growth factors
Tumor-derived growth factors
Growth factors
RANKL
Osteoblasts
Abnormal new bone formation
Mineralized bone matrix
Osteoclasts
= Radium-223

Figure 1. Radium-223 therapy exhibits a dual targeting mode-of-action that destroys tumor cells and inhibits tumor-induced pathologic bone reaction. The effects of radium-223 on local immune cells have not been characterized so far (modified from Suominen *et al.*[2]).

RESULTS

Radium-223 inhibits cancer growth in bone and tumor-induced bone destruction

- Around day 25, when mice with extensive bone lesion were sacrificed, the tumor growth in bone was stabilized in the control group.

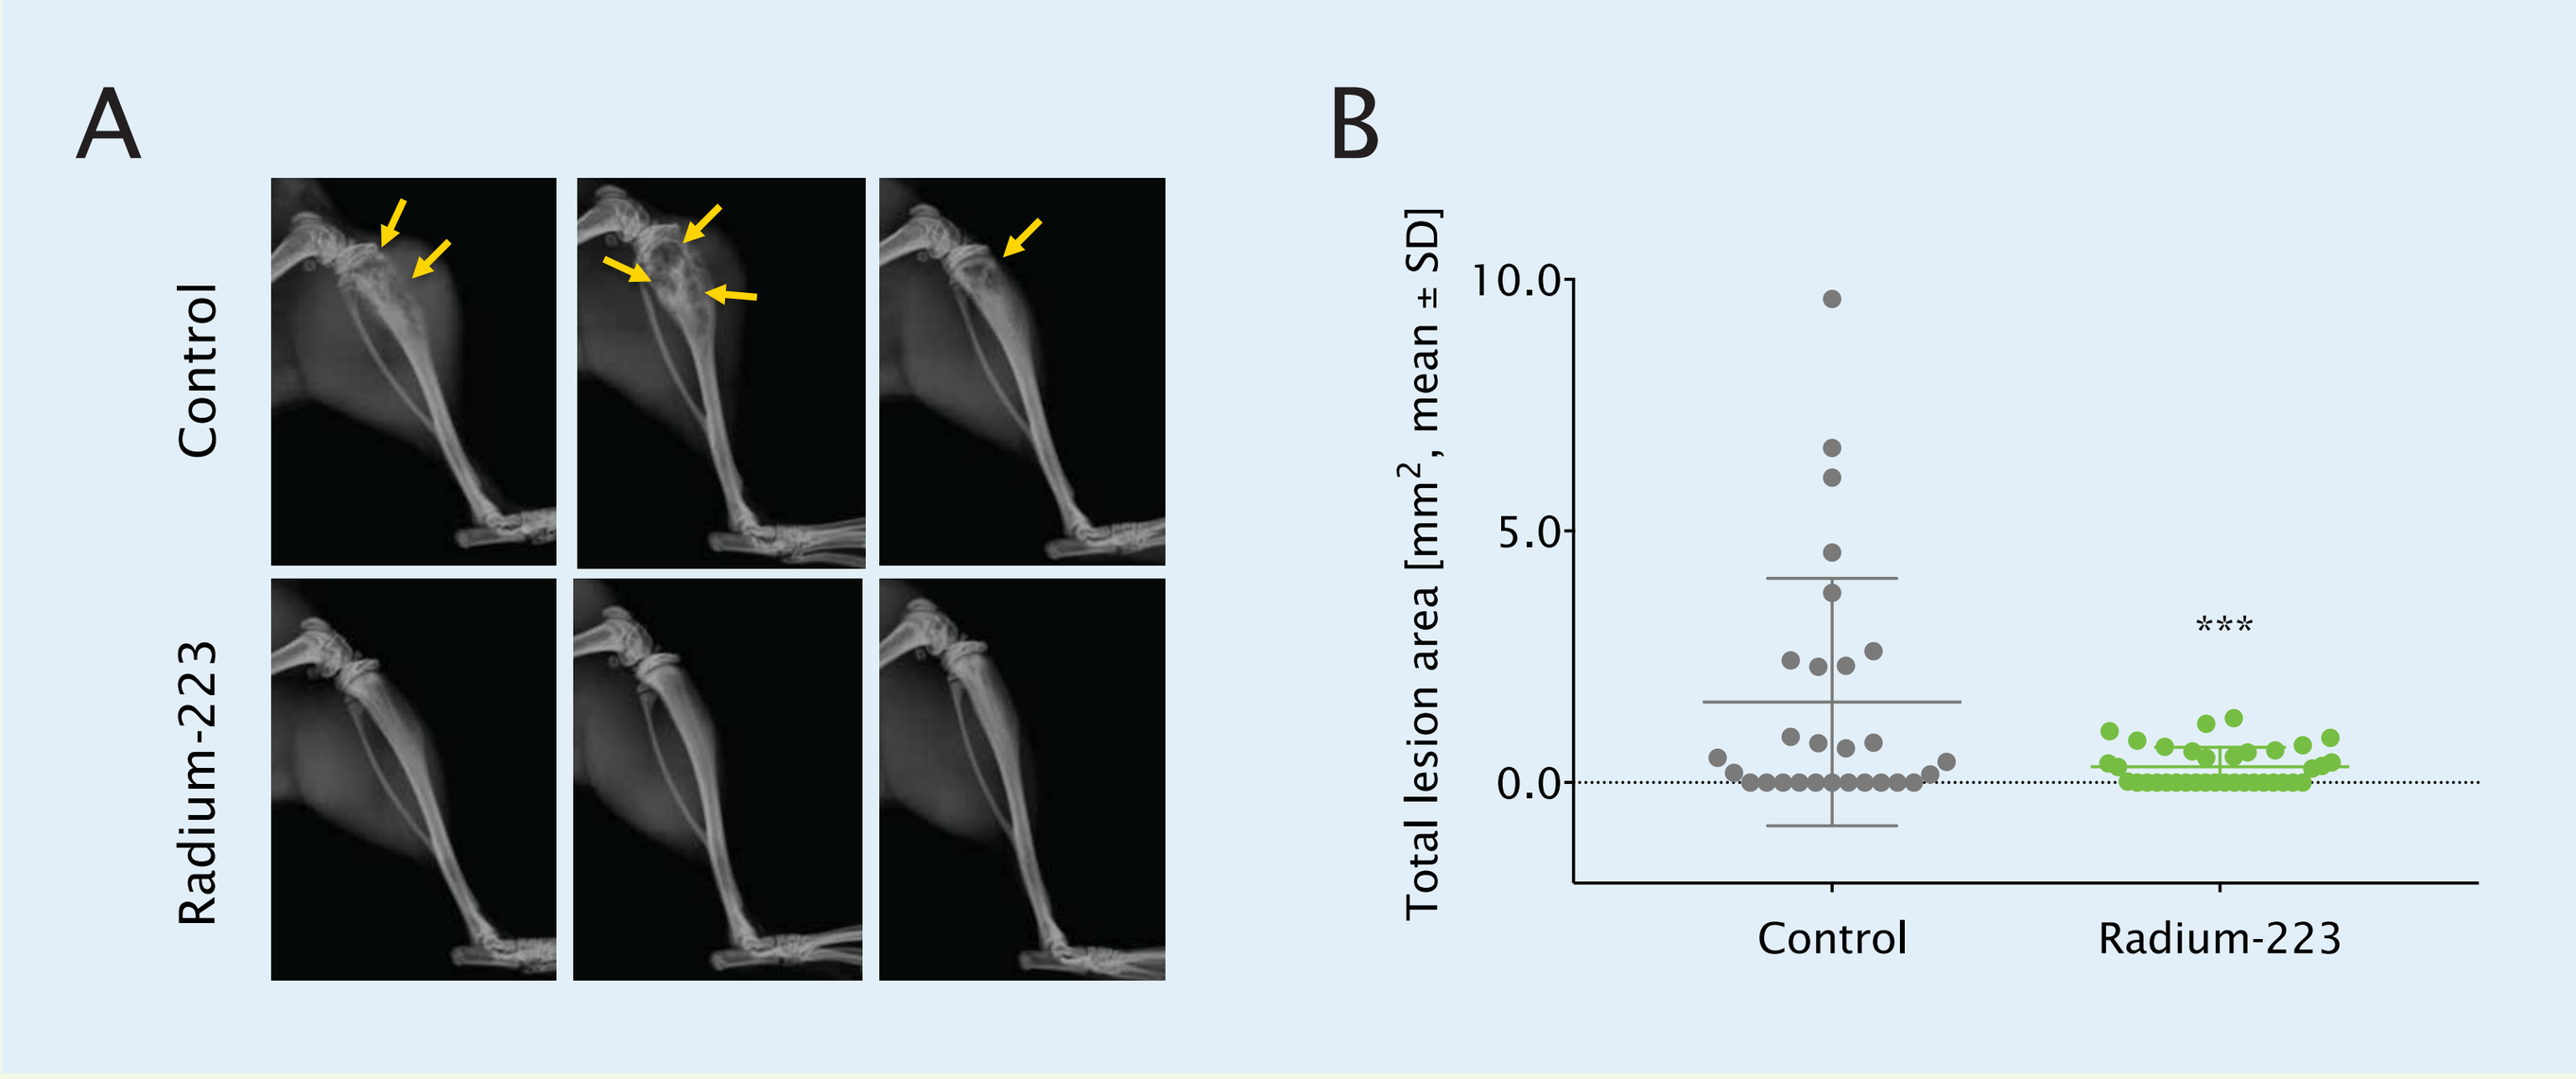
A
Control
Radium-223
B
Total lesion area [mm^2, mean ± SD]
10.0
5.0
0.0

Control
Radium-223

Figure 4. Quantitation of bone lesion area in the syngeneic MBT-2 bladder cancer bone growth model. (A) Representative X-ray images from control and radium-223 treated mice on day 25. Yellow arrows indicate bone lesions. **(B)** Total osteolytic lesion area on day 25. At this time-point, 39% (11/28) of the control animals and 50% (18/36) of the radium-223 treated mice were tumor-free. The data were modeled using a linear fixed-effects model and the comparison was carried out using model contrasts. Asterisks indicate statistical significance compared to the control (***p<0.001).

- Based on X-ray imaging 25 days after cancer cell inoculation, radium-223 treatment reduced the total bone lesion area compared to the control treatment (p<0.001) (**Fig. 4**).
- Radium-223 decreased the tumor-bearing tibia weight compared to the control (p=0.0040) suggesting an inhibitory effect on tumor growth (**Fig. 5**).

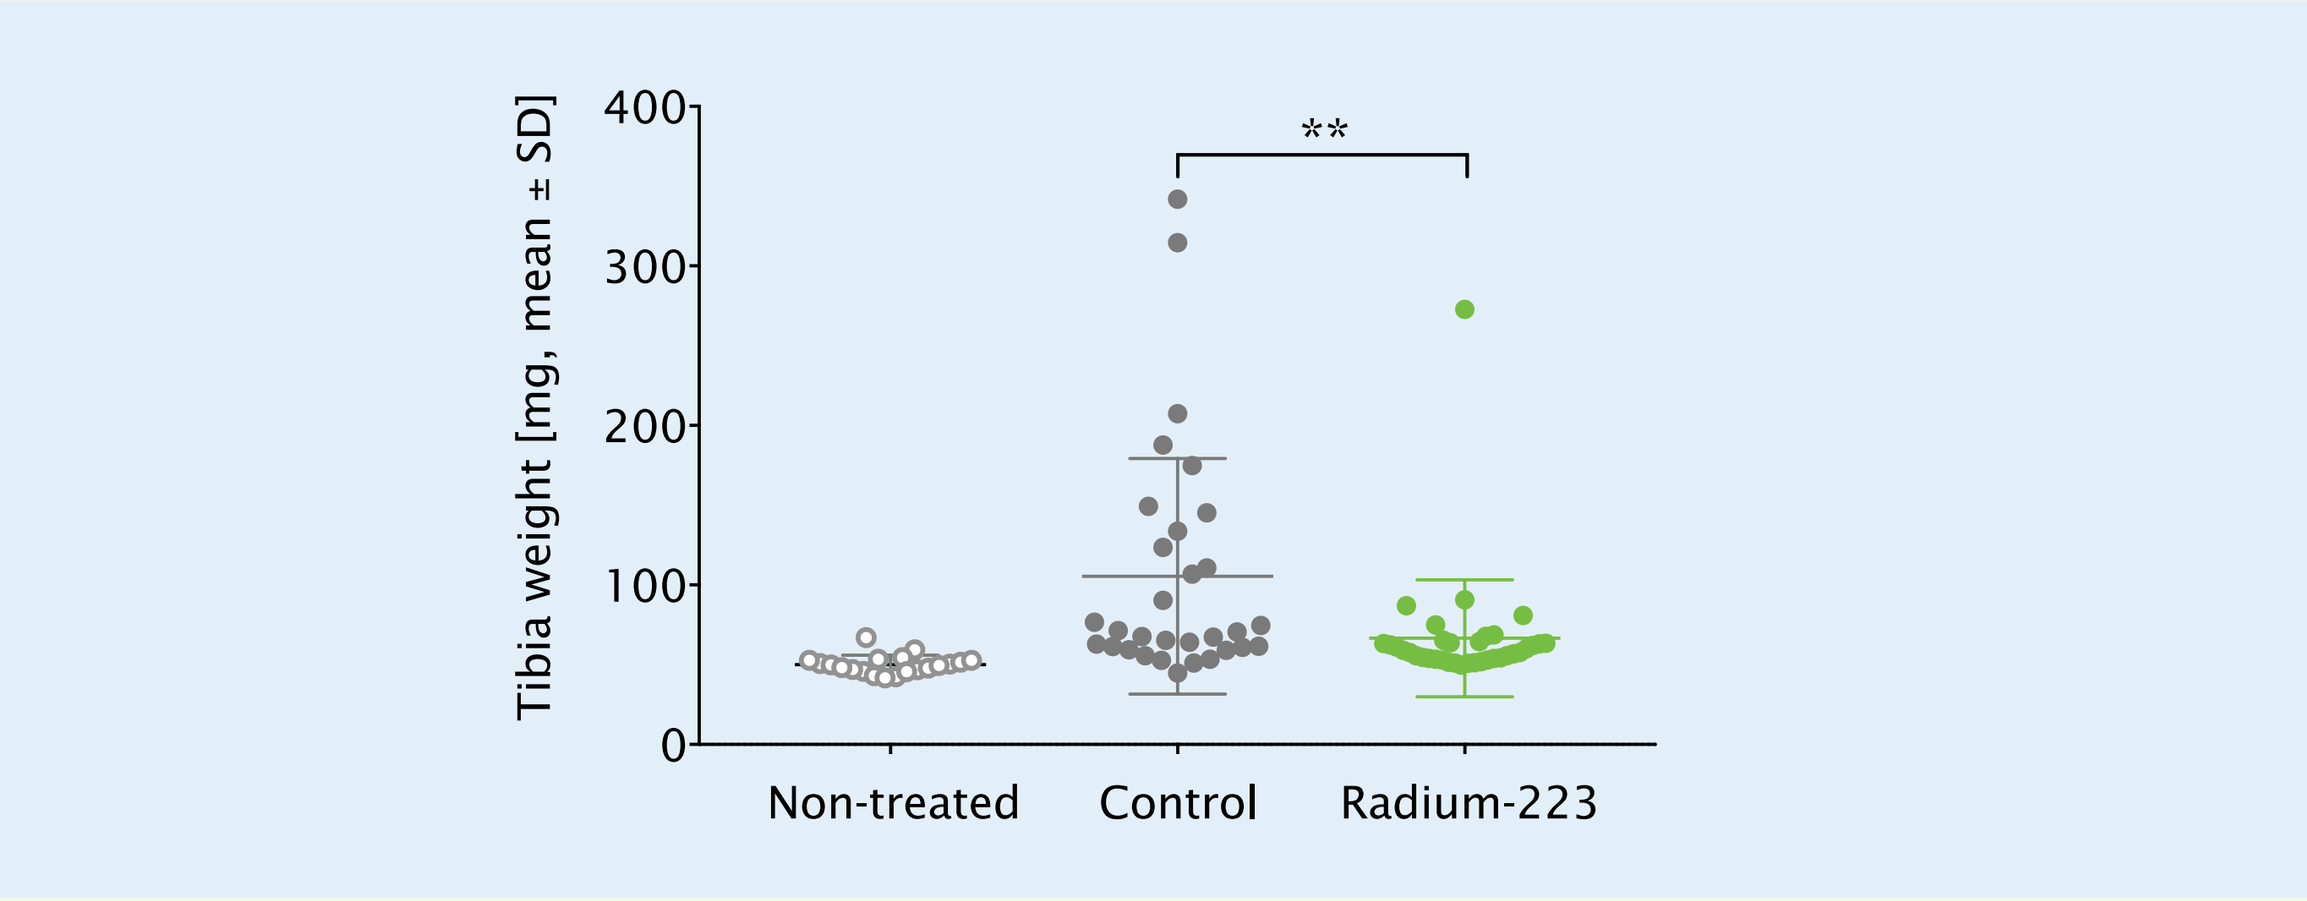
Tibia weight [mg, mean ± SD]
400
300
200
100
0
**
Non-treated
Control
Radium-223

Figure 5. Tibia weights in the syngeneic MBT-2 bladder cancer bone growth model. At sacrifice, the tumor-bearing tibias were dissected and weighted. Statistical analysis was performed using Kruskal-Wallis test. Radium-223 decreased the tibia weight compared to control (**p=0.0040).

The most common causes for mortality: local outgrowth from bone and lung metastases

- The largest bone lesions were seen 25 days after inoculation of MBT-2 cancer cells. This was also the time point where most of the mice were sacrificed due to tumor outgrowth from bone.
- When the mice with extensive bone lesions were sacrificed, the tumor growth was stabilized.
- Intratibial inoculation of MBT-2 cells resulted in the formation of lung metastases in some of the animals.The main reason for sacrifice at a later stage was breathing difficulties, and of the radium-223-treated mice, only one mouse was sacrificed due to bone metastases.

Radium-223 treatment is well-tolerated

- Radium-223 treatment was well-tolerated in terms of fatal toxicities, cachexia and body weight loss.

METHODS

Female 5-7-week-old C3H/HeNHsd mice were injected intratibially with 5×10^5 MBT-2 cells on day 0. The mice were stratified based on body weight (n(non-treated)=20/group; n(treated)=40/group) on day 5 and the non-treated mice were sacrificed for evaluation of tumor take rate as assessed by histology (**Fig. 2**). Radium-223 (300 kBq/kg, i.v.) or vehicle control was administered on days 5, 33, and 61 (**Fig. 3**).

The tumor-induced osteolytic lesion area was imaged using radiography on days 10, 18, 25, 76, and at sacrifice. The mice with no detectable active bone disease (total bone lesion area below 0.5 mm^2 during the study) that survived until the end of the study (day 158) were excluded from all analyses. Nine mice in the control group and three mice in the radium-223 treatment group were excluded based on this criterion.

The mice were sacrificed when they met the pre-defined sacrifice criteria (> 20% body weight loss, palpable tumor outgrowth from the bone or general deterioration of health status). The study was terminated on day 158.

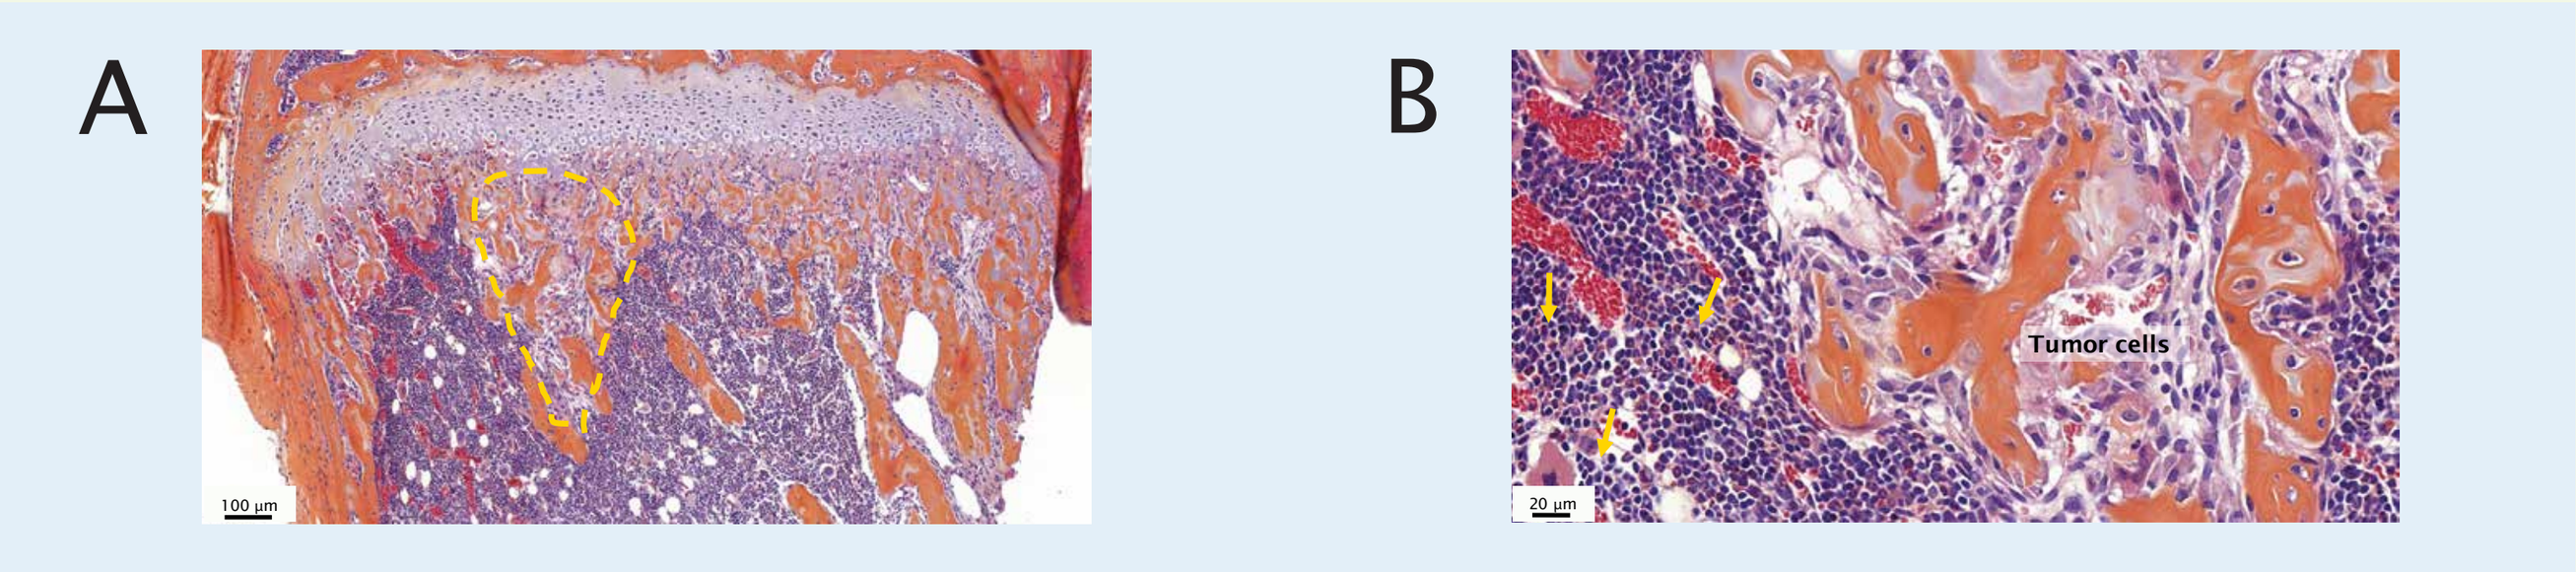
A
B
Tumor cells

Figure 2. Representative images of hematoxylin and eosin (HE) stained tibias of non-treated mice 5 days after cancer cell inoculation. (A) 10x magnification: the area marked in yellow contains tumor cells. **(B)** 40x magnification: lymphocytes are indicated with asterisks. The non-treated mice were sacrificed 5 days after cancer cell inoculation and the tumor take rate was evaluated by histology. All the non-treated mice (n=20) had detectable tumor cells or small tumor islets on day 5 in the inoculated tibias.

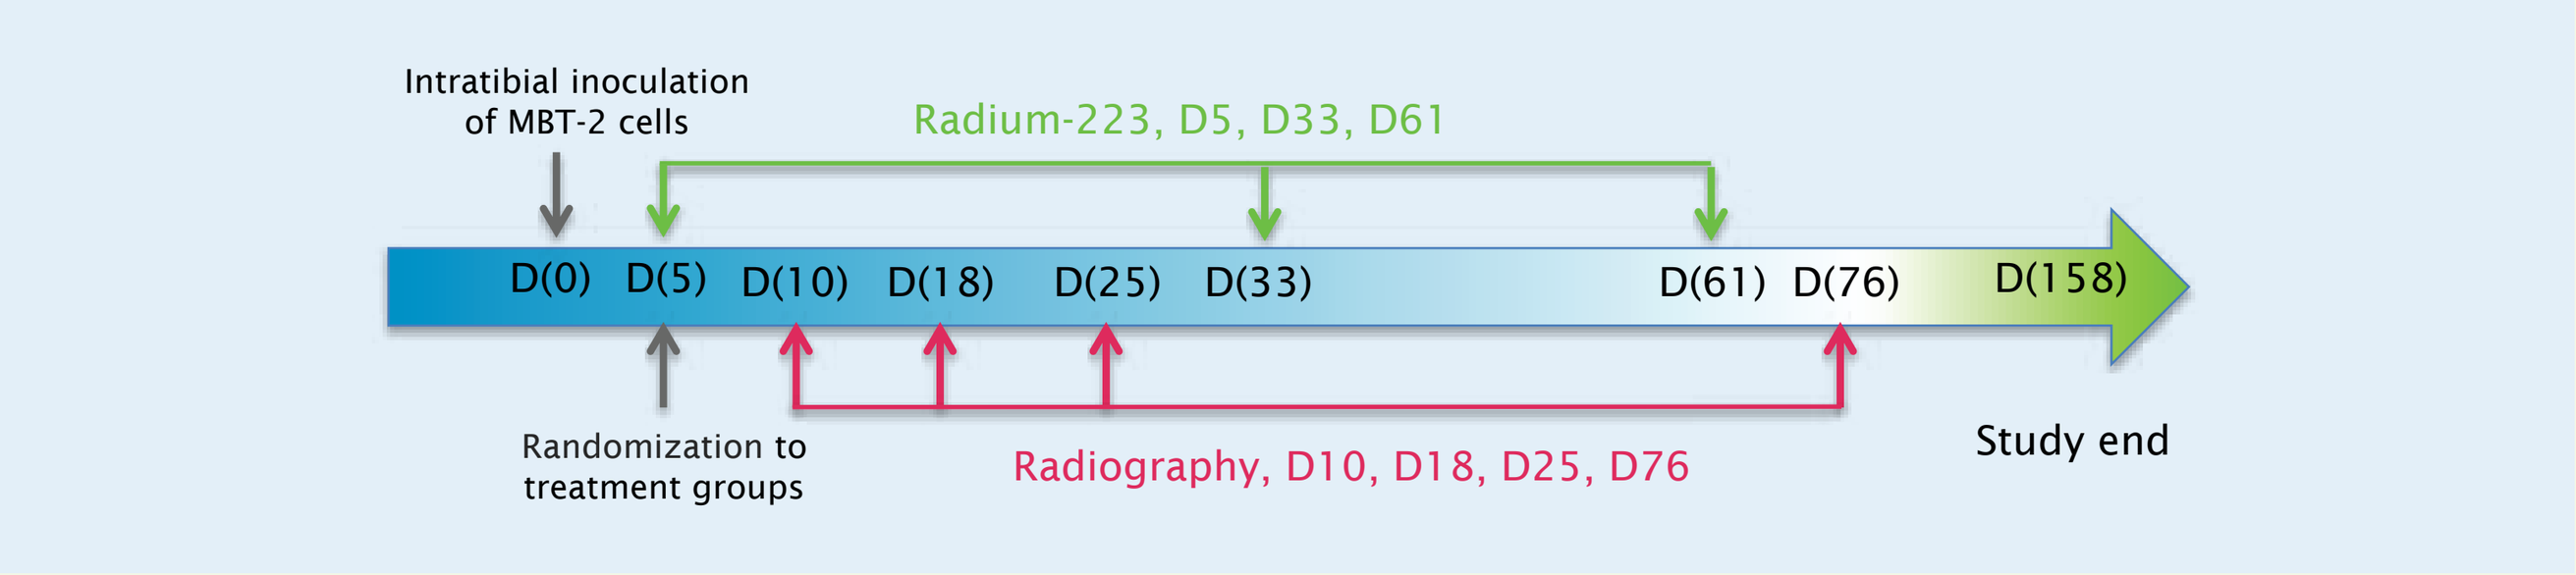
Intratibial inoculation of MBT-2 cells
Radium-223, D5, D33, D61
D(0) D(5) D(10) D(18) D(25) D(33) D(61) D(76) D(158)
Randomization to treatment groups
Radiography, D10, D18, D25, D76
Study end

Figure 3. Design for radium-223 treatment in the syngeneic MBT-2 bladder cancer bone growth mouse model.

Radium-223 improves survival

- The first mice in the control group met the sacrifice criteria on study day 24 (**Fig. 6**).
- Survival was 97% in the radium-223 treatment group compared to 65% in the control group (p< 0.001) on day 40 (**Fig. 6**).
- The survival curves plateaued at 52% after 84 days in the control group and at 68% after 112 days in the radium-223 treatment group. The study was terminated on day 158.
- At the end of the study, the difference in survival between the control and the radium-223 treatment group was not statistically significant.

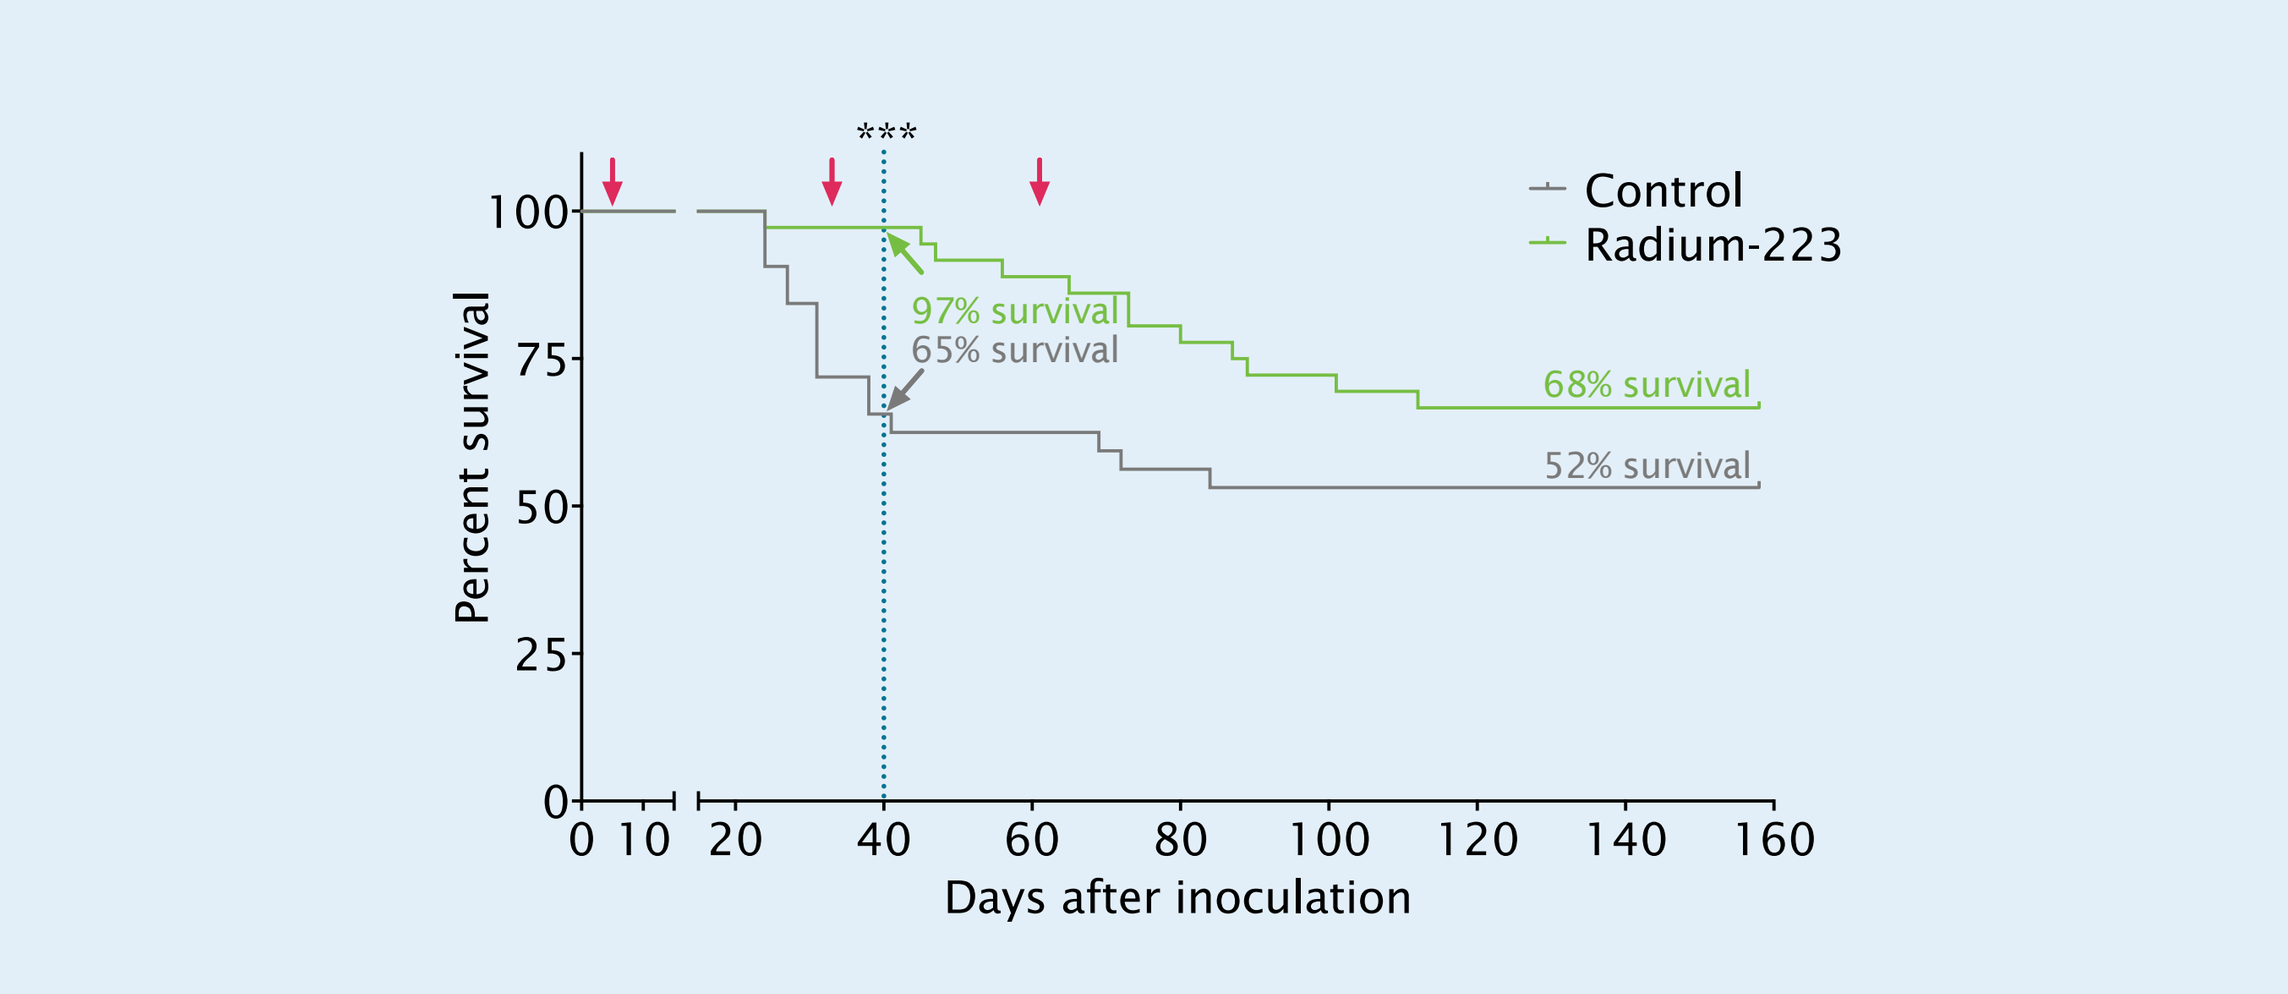

Control
Radium-223
Percent survival
100
75
50
25
0
97% survival
65% survival
68% survival
52% survival
0 10 20 40 60 80 100 120 140 160
Days after inoculation

Figure 6. Effect of radium-223 treatment on survival in the syngeneic MBT 2 bladder cancer bone growth model. The time from cancer cell inoculation to sacrifice was recorded and reported as a Kaplan-Meier survival plot. The statistical analysis was performed using log-rank test. Survival on day 40 was significantly improved in the radium-223 treatment group compared to the control (***p< 0.001), but the effect was not statistically significant at the end of the study. Red arrows indicate treatment days.

CONCLUSIONS

- Radium-223 dichloride (Xofigo®) inhibits cancer growth in bone and ameliorates tumor-induced bone destruction resulting in a beneficial effect on survival in the intratibial MBT-2 murine bladder cancer model of osteolytic bone metastasis.
- The single agent *in vivo* efficacy of radium-223 results in a moderate beneficial effect on survival.
- The promising results obtained in this immune-competent model warrant further investigation of radium-223 in combination with immuno-oncological treatments such as checkpoint inhibitors.

REFERENCES

1. Parker C, Nilsson S, Heinrich D, Helle SI, O'Sullivan JM, *et al.*, Alpha emitter radium-223 and survival in metastatic prostate cancer. *Engl J Med.* 2013; 369:213-23.

2. Suominen MI, Rissanen JP, Käkönen R, Fagerlund KM, Alhoniemi E., *et al.* Survival benefit with radium-223 dichloride in a mouse model of breast cancer bone metastasis. *J Natl Cancer Inst.* 2013; 105:908-16.

2. Suominen MI, Fagerlund KM, Rissanen JP, Konkol YM, Morko JP, *et al.* Radium 223 inhibits osseous prostate cancer growth by dual targeting of cancer cells and bone microenvironment in mouse models. *Clin Cancer Res.* 2017; 23:4335-46.

ACKNOWLEDGMENTS

Aurexel Life Sciences Ltd. (www.aurexel.com) is thanked for editorial assistance in the preparation of this poster, funded by Bayer AG.

Placeholder for QR code

Poster presented at the AACR Annual Meeting, March 29-April 3, 2019, Atlanta, GA, USA