3311

Darolutamide potentiates the antitumor efficacy of a PSMA-targeted thorium-227 conjugate (PSMA-TTC) in a hormone-independent prostate cancer model

Christoph A. Schatz[1], Urs B. Hagemann[1], Sabine Zitzmann-Kolbe[1], Bernard Haendler[1], Hartwig Hennekes[1], Stefanie Hammer[1], Justyna Zdrojewska[2], Arne Scholz[1]

[1]Bayer AG, Pharmaceuticals, Berlin, Germany; [2]Pharmatest Services Ltd., Turku, Finland

INTRODUCTION

- Targeted alpha therapy (TAT) consists of a tumor-targeting molecule, a chelating agent, and an alpha emitter and it delivers alpha radiation with high linear energy transfer selectively to tumors.[1]
- The TAT prostate-specific membrane antigen (PSMA)-targeted thorium-227 conjugate PSMA-TTC (BAY 2315497, ^{227}Th-pelgifatamab corixetan) targets radiation to PSMA-positive cancer cells. It consists of a human PSMA-specific antibody covalently linked to a 3,2-HOPO chelator moiety radiolabeled with the alpha emitter thorium-227 (**Fig. 1**).[2]
- Darolutamide is a novel androgen receptor inhibitor:
 - Approved by the FDA for the treatment of non-metastatic castration-resistant prostate cancer (CRPC) based on the results of the phase III study ARAMIS, as reported in the NEJM in 2019.[3]
 - Darolutamide in combination with androgen deprivation therapy (ADT) and the standard-of-care docetaxel significantly improved overall survival (primary endpoint), with a reduction in risk of death by 32.5% compared with ADT and docetaxel alone (HR 0.675; 95% CI 0.568–0.801; *P*<0.0001).[4]
- Although most patients initially respond well to ADT, eventually the disease becomes androgen-independent.
- We have previously demonstrated that PSMA-TTC has potent antitumor activity in PSMA-expressing prostate cancer models.[2,5-6] Moreover, we have shown that PSMA-TTC in combination with darolutamide shows synergistic antitumor efficacy in androgen-dependent *in vivo* prostate cancer models with no overlapping toxicities.[7]
- Here, we evaluated the antitumor efficacy of PSMA-TTC in combination with darolutamide in androgen-independent prostate cancer xenograft models with low or high PSMA baseline expression.

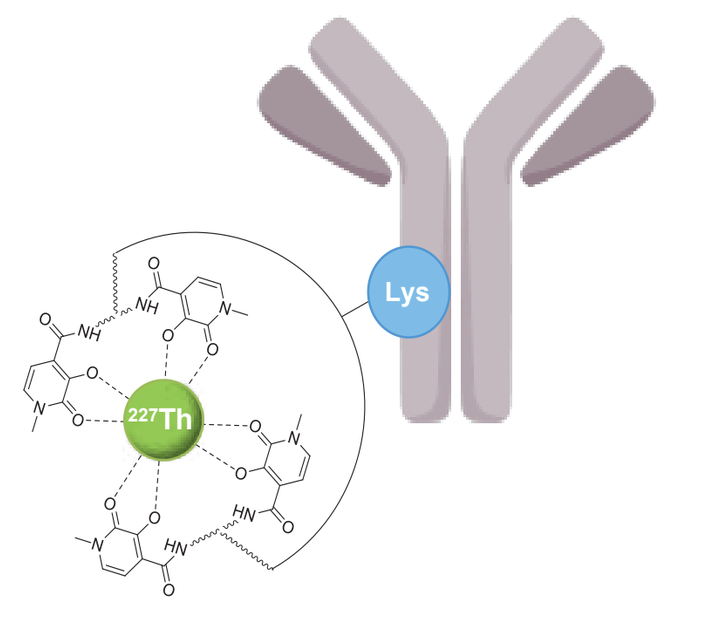
Lys
^{227}Th

Figure 1. Structure of PSMA-TTC (BAY 2315497, ^{227}Th-pelgifatamab corixetan).

METHODS

- The *in vitro* effect of fixed-ratio combinations of PSMA-TTC and darolutamide was assessed in human 22Rv1 and C4-2 prostate cancer cells. Cell viability was assessed after a 5-day exposure to PSMA-TTC (CellTiter-Glo®, Promega). Darolutamide was added 3 days before PSMA-TTC. IC_{50} values, isobolograms, and combination indices (CI) were calculated according to the median-effect model of Chou-Talalay[8] with CI<0.8 defined as synergistic effect.
- Cell surface PSMA expression was determined by flow cytometry using the murine PSMA-specific mAb J591 (prepared in-house) after a 7-day treatment with 5 µM darolutamide.
- *XRCC2*, *XRCC3*, and *CDKN1A* gene expression in 22Rv1 and C4-2 cells was analyzed by qRT-PCR after a 48-h treatment with 2 µM darolutamide, 5 kBq/mL PSMA-TTC, or their combination.
- The *in vivo* antitumor efficacy of PSMA-TTC in combination with darolutamide was evaluated in two androgen-independent prostate cancer models: low PSMA-expressing 22Rv1 and high PSMA-expressing C4-2. Male nude mice (n=10–12/group, 22Rv1: nude (nu/nu), C4-2: Balb/c nude) were treated with a single injection of PSMA-TTC (150 or 300 kBq/kg, i.v., total protein dose 0.14 mg/kg) and/or darolutamide (100 mg/kg, BID, p.o.) starting from day 10 or 15 after 22Rv1 or C4-2 tumor cell inoculation, respectively.
- PSMA expression was determined by immunohistochemistry (IHC) using the murine PSMA-specific mAb GCP-04 (Novus Biologicals) in formalin-fixed, paraffin-embedded (FFPE) tissue sections of isolated 22Rv1 and C4-2 tumors.[7] In addition, 22Rv1 tumors were analyzed for PSMA expression in dissociated tumor cells by flow cytometry using the J591 antibody.

RESULTS

Darolutamide shows differential effects in combination with PSMA-TTC in 22Rv1 and C4-2 prostate cancer cells *in vitro*

- The combination of PSMA-TTC and darolutamide showed synergistic antitumor efficacy in low PSMA-expressing 22Rv1 but not in high PSMA-expressing C4-2 cells *in vitro* (**Fig. 2A-B**).
- Darolutamide induced cell surface PSMA protein expression in 22Rv1 cells and, despite high baseline levels of PSMA, also in C4-2 cells (**Fig. 2C-D**).
- Darolutamide reduced the expression of the DNA damage response (DDR) gene *XRCC3* in 22Rv1 but not in C4-2 cells (**Fig. 3A-B**).
- PSMA-TTC decreased the expression of *XRCC2* and *XRCC3* (**Fig. 3A-D**) and increased the expression of *CDKN1A*, a p53 pathway gene linking DNA damage to cell cycle arrest, in both 22Rv1 and C4-2 cells (**Fig. 3E-F**). Adding darolutamide to the treatment regimen had no additional effects on the expression of these genes.

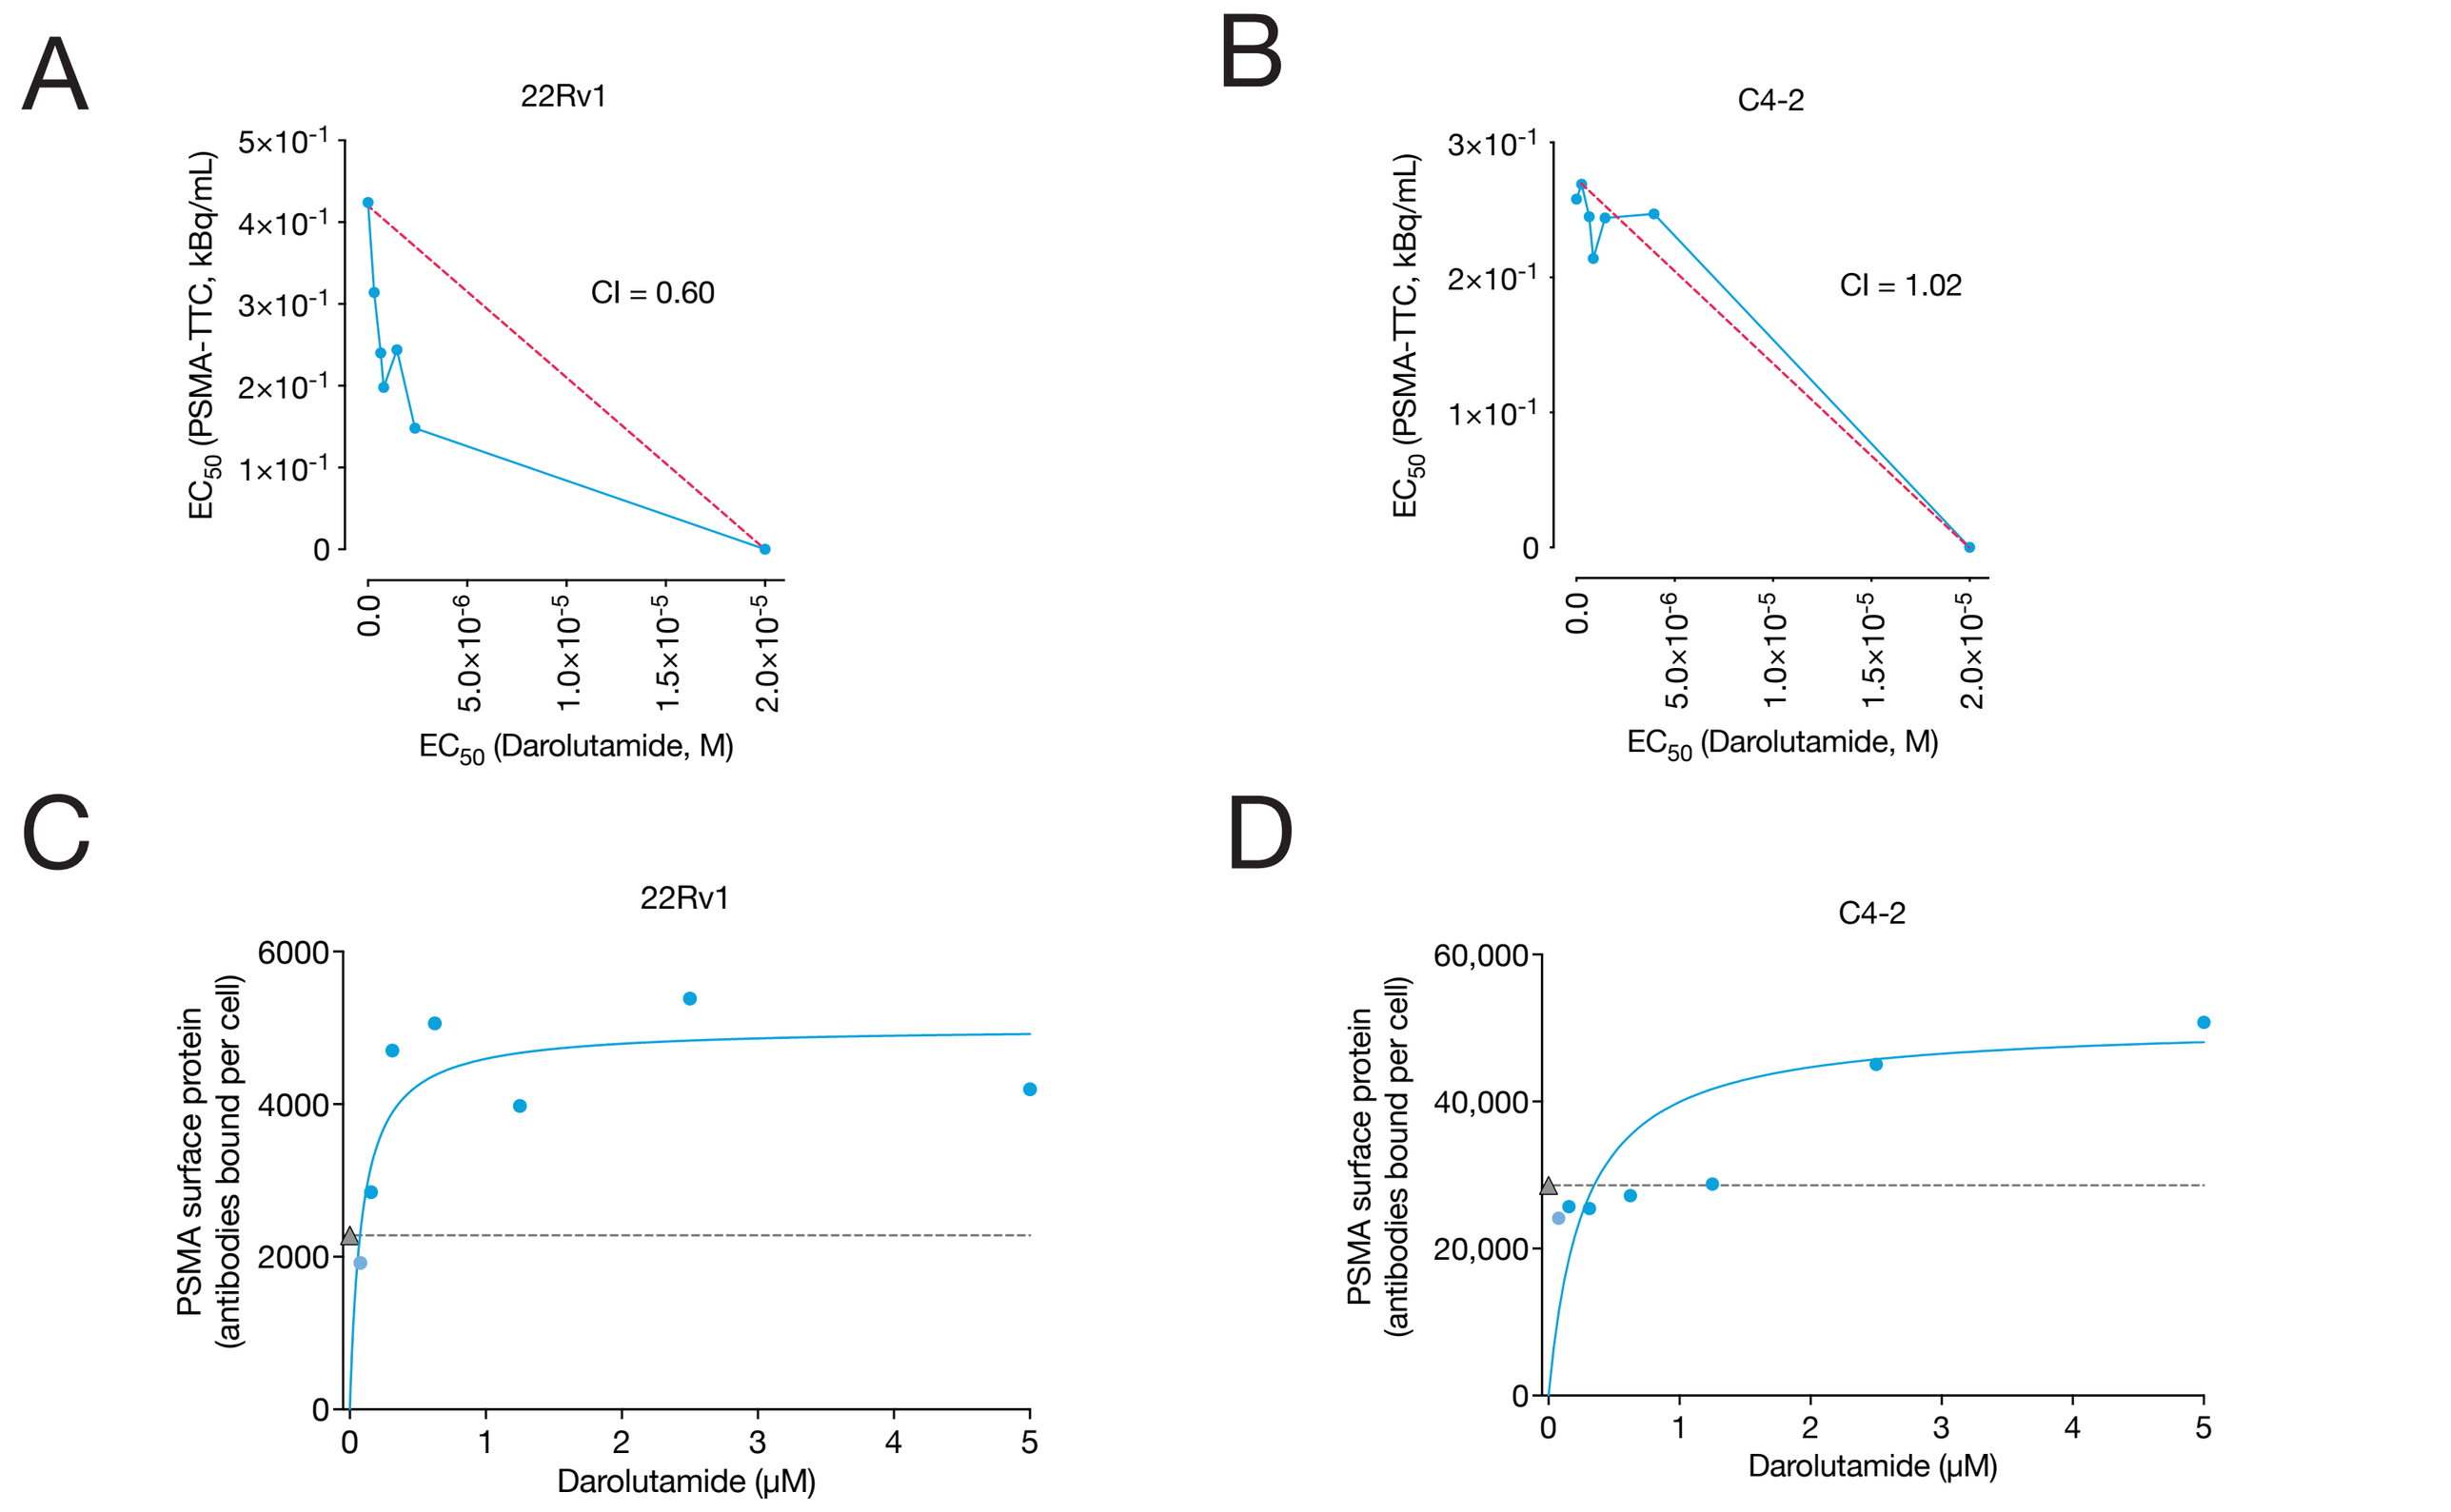
A
22Rv1
CI = 0.60
EC50 (PSMA-TTC, kBq/mL)
EC50 (Darolutamide, M)
B
C4-2
CI = 1.02
C
22Rv1
PSMA surface protein (antibodies bound per cell)
Darolutamide (µM)
D
C4-2

Figure 2. Differential effects of darolutamide and PSMA-TTC combination treatment in 22Rv1 and C4-2 prostate cancer cells *in vitro*. Isobolograms for the combination effect of PSMA-TTC and darolutamide on the proliferation of **(A)** 22Rv1 and **(B)** C4-2 cells. Cell surface PSMA expression as determined by flow cytometry in darolutamide-treated **(C)** 22Rv1 and **(D)** C4-2 cells. DMSO served as baseline control and is depicted with a dashed line. CI, combination index.

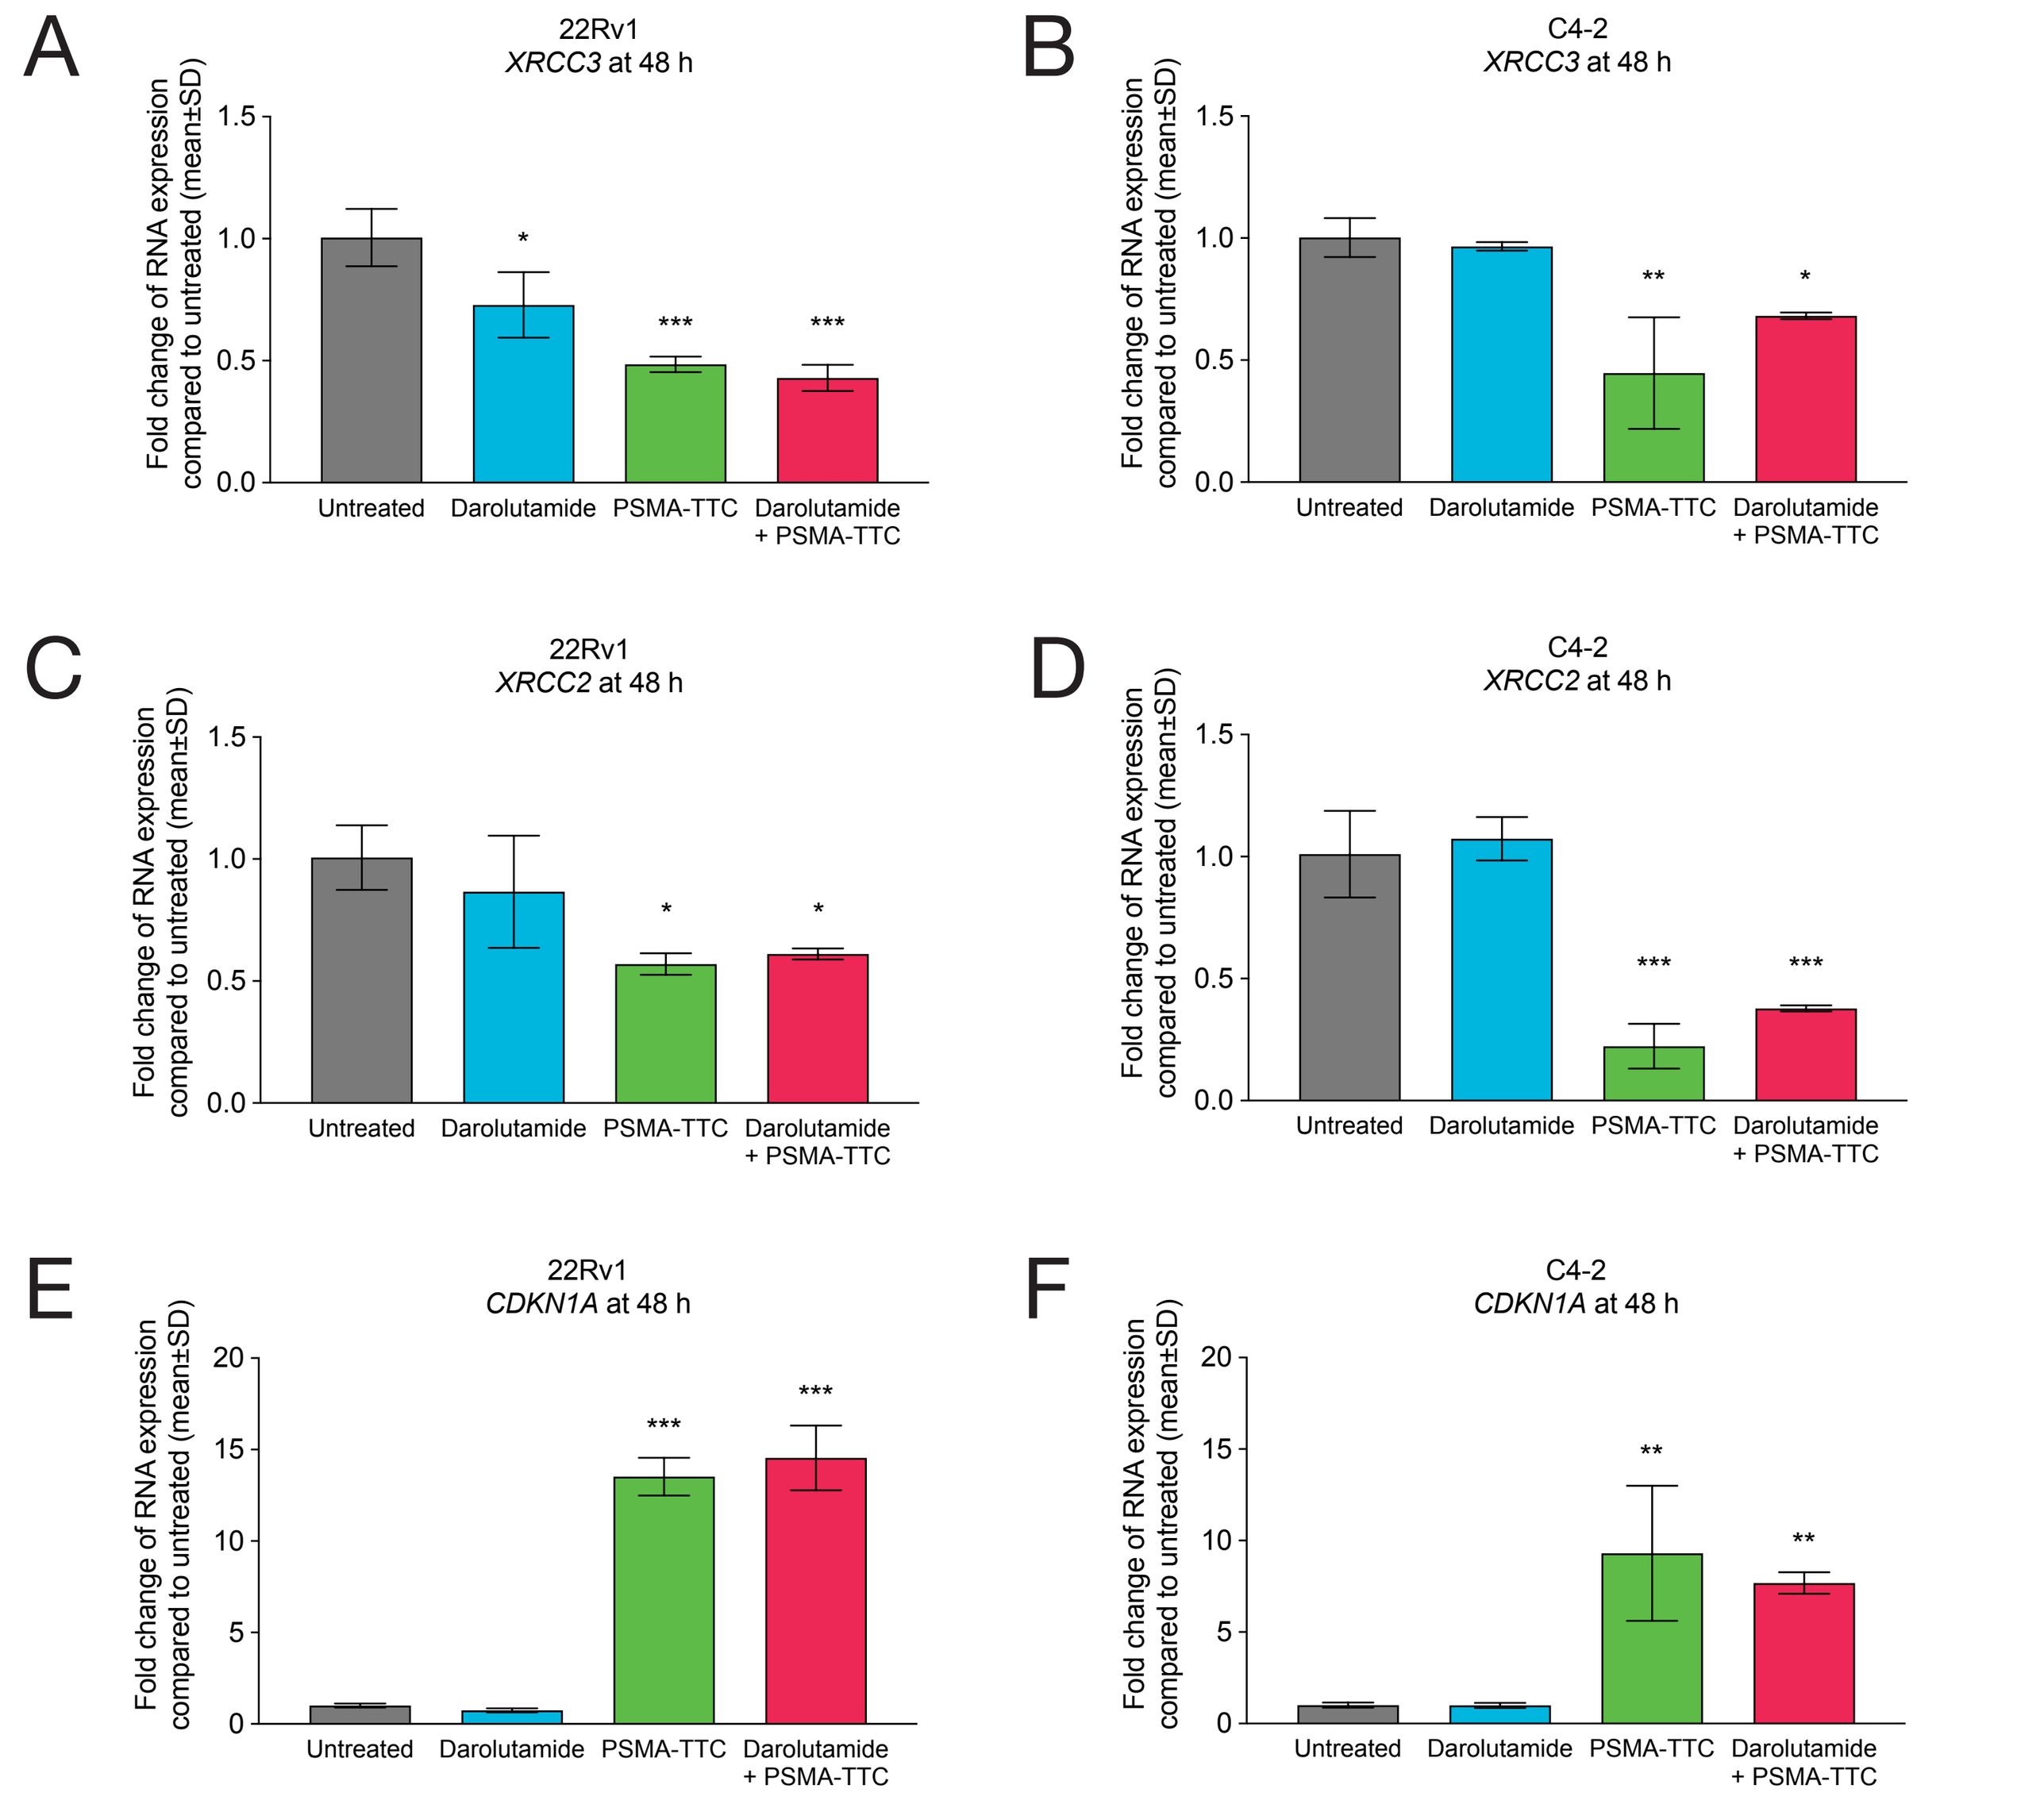
A
22Rv1
XRCC3 at 48 h
B
C4-2
XRCC3 at 48 h
C
22Rv1
XRCC2 at 48 h
D
C4-2
XRCC2 at 48 h
E
22Rv1
CDKN1A at 48 h
F
C4-2
CDKN1A at 48 h
Fold change of RNA expression compared to untreated (mean±SD)
Untreated
Darolutamide
PSMA-TTC
Darolutamide + PSMA-TTC

Figure 3. Expression of DNA damage response genes and cell cycle-related genes upon darolutamide and PSMA-TTC treatment in 22Rv1 and C4-2 prostate cancer cells. Relative RNA expression of the DNA damage response genes **(A-B)** *XRCC3* and **(C-D)** *XRCC2* and **(E-F)** the p53 pathway gene *CDKN1A* in 22Rv1 and C4-2 cells treated with 2 µM darolutamide and/or 5 kBq/mL PSMA-TTC, as determined by qRT-PCR (n=3). Statistical analyses were performed using one-way ANOVA combined with Dunnett's test. *, p<0.05; **, p<0.01; ***, p<0.001 vs. untreated cells.

Darolutamide induces PSMA expression in low PSMA-expressing 22Rv1 prostate cancer xenografts

- In line with the *in vitro* results (**Fig. 2C-D**), 22Rv1 tumor xenografts were shown to express only low levels of PSMA (**Fig. 4A**), whereas high PSMA expression levels were detected in C4-2 tumor xenografts (**Fig. 4B**).
- Darolutamide treatment resulted in a 4-fold increase of PSMA expression compared with the baseline control in 22Rv1 tumors (**Fig. 4C**).

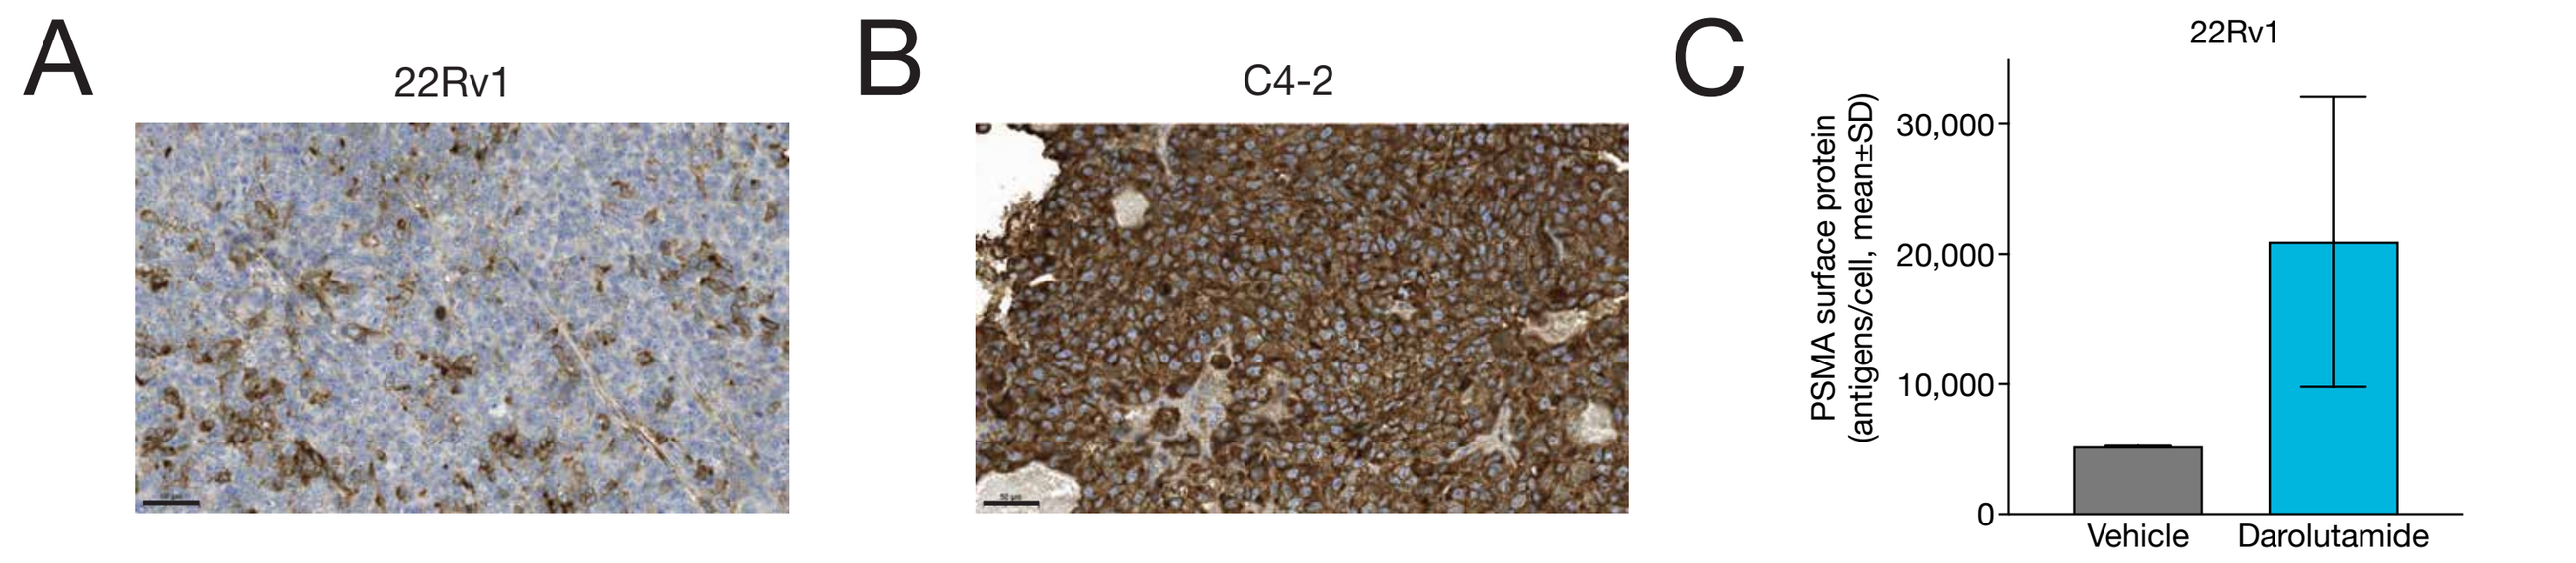
A
22Rv1
B
C4-2
C
22Rv1
PSMA surface protein (antigens/cell, mean±SD)
Vehicle
Darolutamide

Figure 4. PSMA expression in 22Rv1 and C4-2 tumors *ex vivo*. PSMA expression in **(A)** 22Rv1 and **(B)** C4-2 tumor xenografts detected by IHC using the PSMA antibody GCP-04. Brown color indicates PSMA staining. Scale bars indicate 50 µm. **(C)** PSMA expression in dissociated 22Rv1 cancer cells obtained from 22Rv1 tumor-bearing mice treated with vehicle or darolutamide for 18 days before sacrifice (n=2), as determined by flow cytometry.

PSMA-TTC in combination with darolutamide shows additive antitumor efficacy in the low PSMA-expressing 22Rv1 prostate cancer xenograft model

- PSMA-TTC monotherapy at 150 and 300 kBq/kg showed dose-dependent antitumor efficacy with T/C ratios of 0.54 and 0.30, respectively, on day 19 (the last day of the vehicle group) (**Fig. 5A-B, Table 1**).
- In contrast, darolutamide was not efficacious at 100 mg/kg (BID) compared with vehicle on day 19 (**Fig. 5A-B, Table 1**).
- PSMA-TTC (150 or 300 kBq/kg) in combination with darolutamide showed additive antitumor efficacy compared with the respective monotherapy groups on day 19, with T/C ratios of 0.30 and 0.14, respectively (**Fig. 5A-B, Table 1**).
- Combination treatment with 150 or 300 kBq/kg PSMA-TTC and darolutamide markedly extended the time for tumors to reach a volume of 500 mm³ compared with the respective monotherapies (**Fig. 5C, Table 1**).
- All treatments were well tolerated as indicated by body weight loss of less than 5% (**Fig. 5D**).

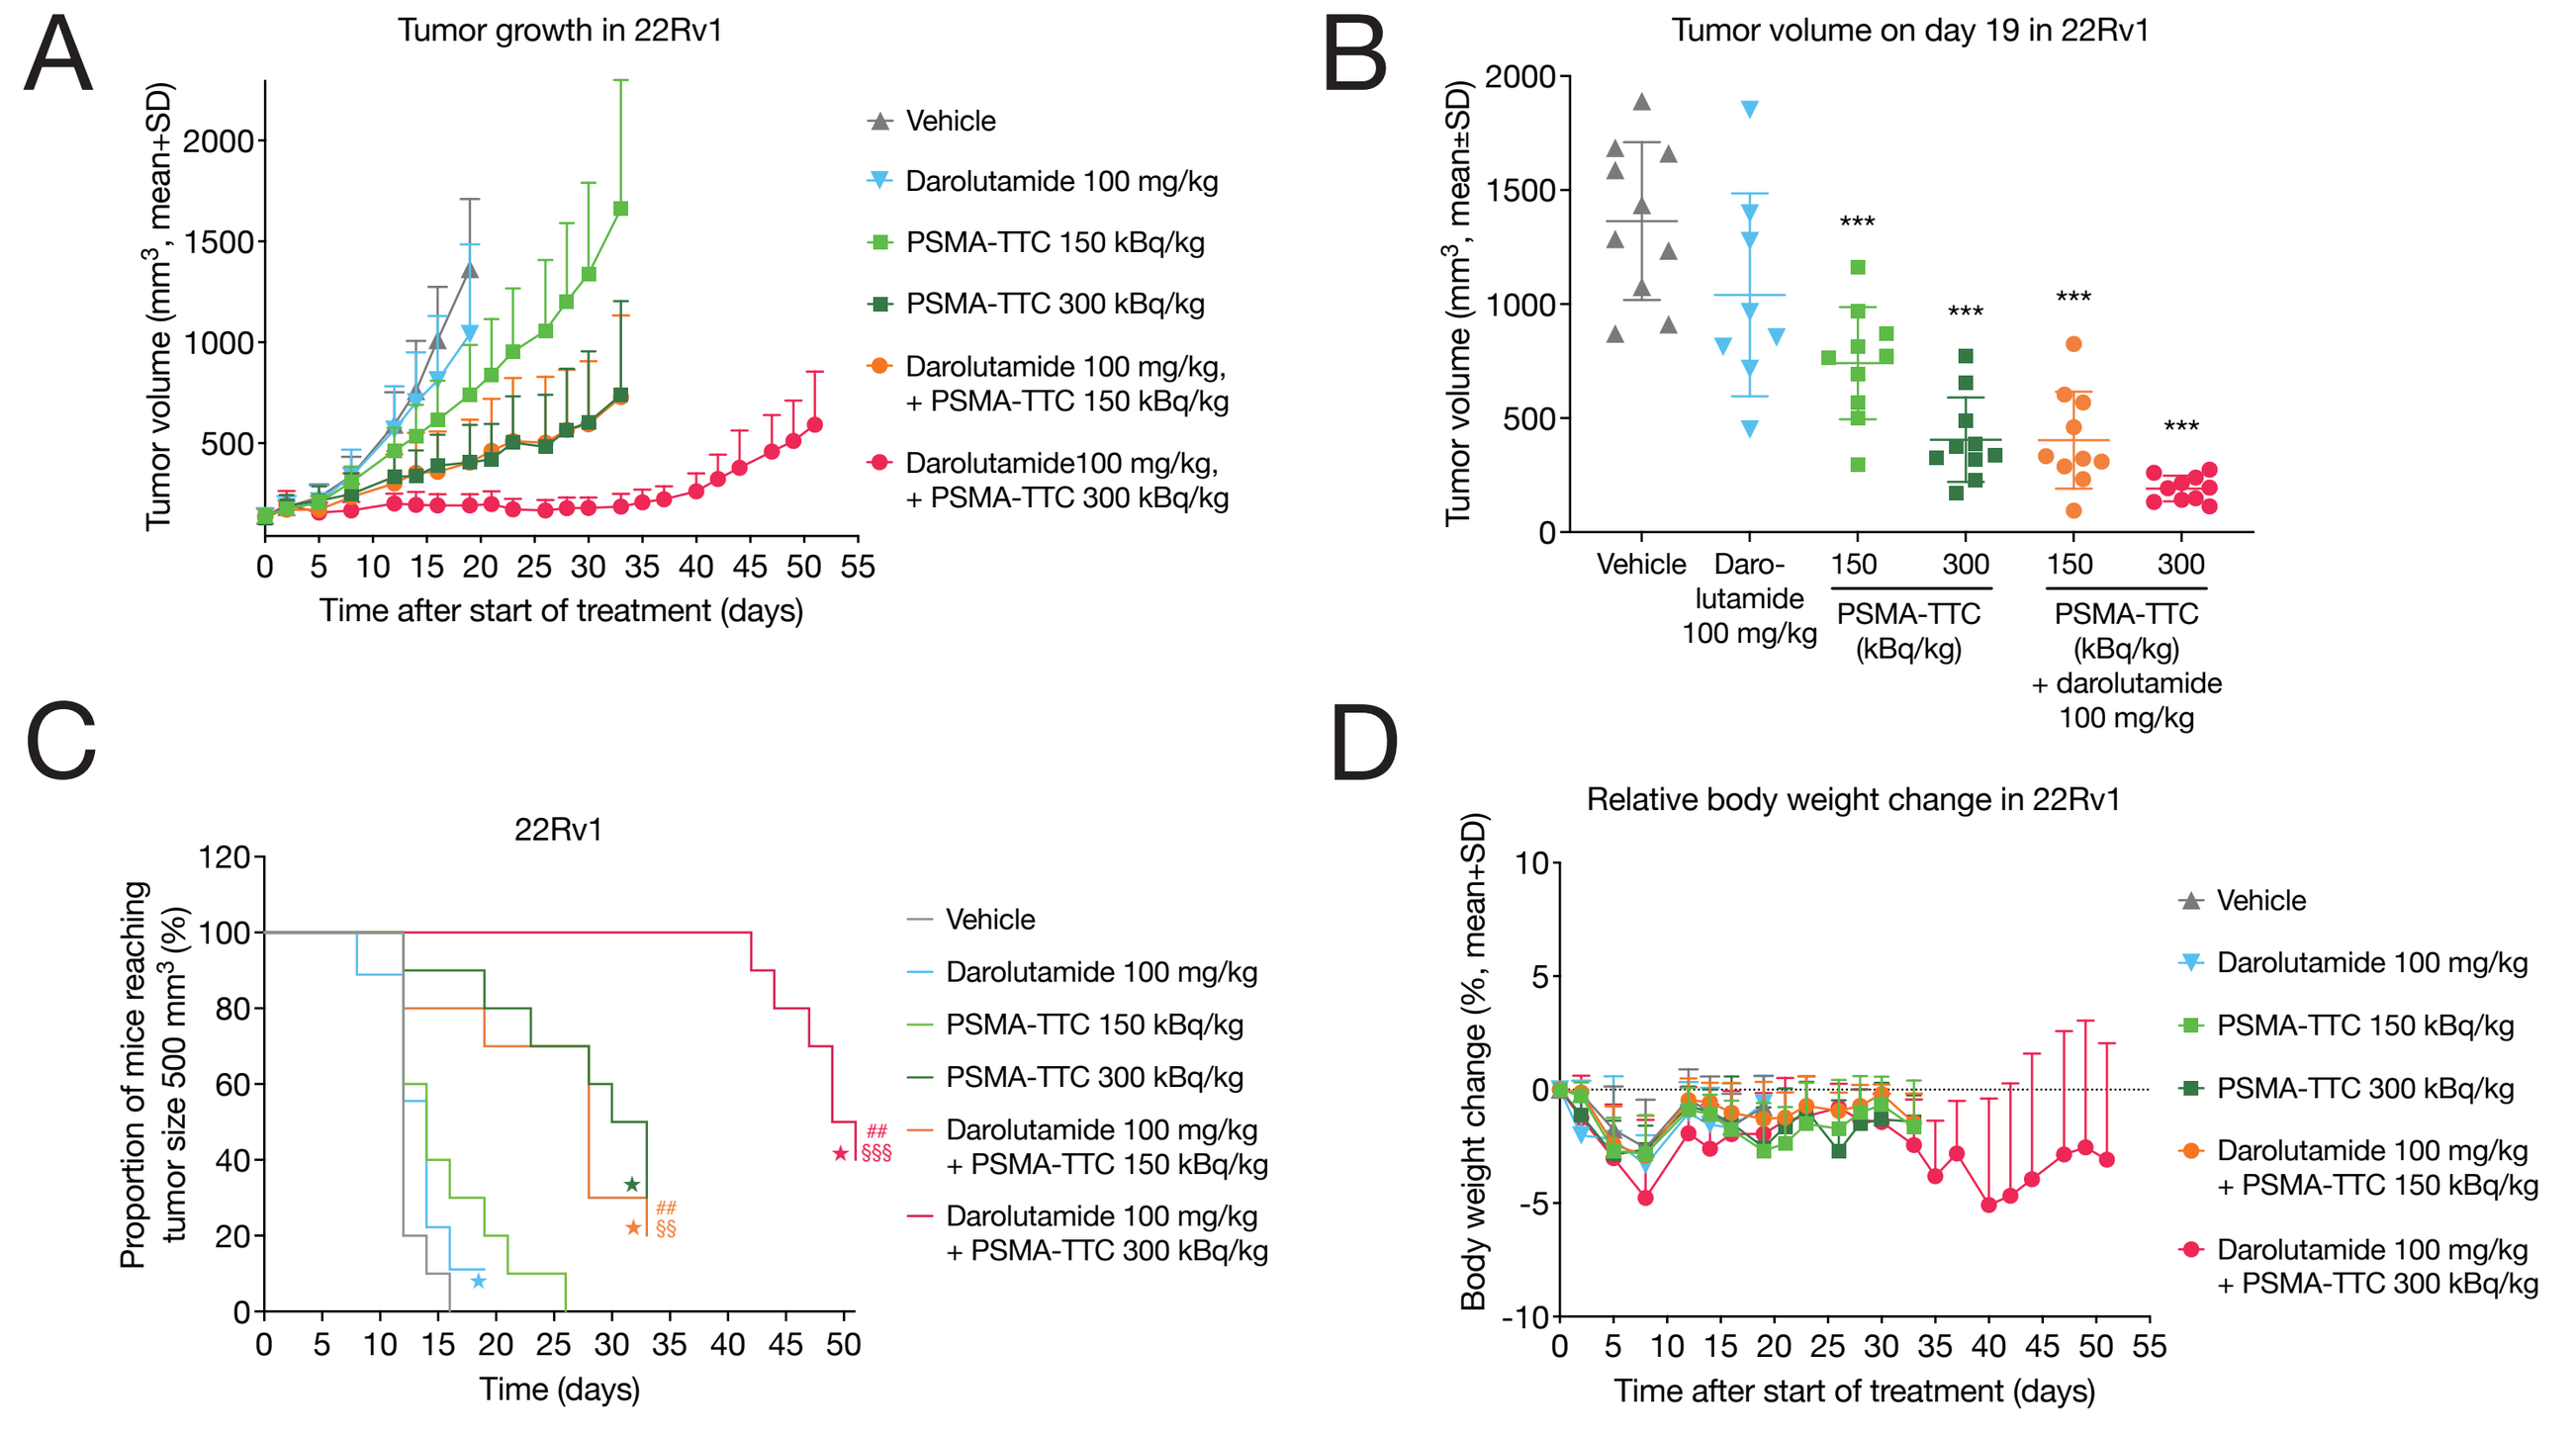
A
Tumor growth in 22Rv1
Tumor volume (mm³, mean+SD)
Time after start of treatment (days)
Vehicle
Darolutamide 100 mg/kg
PSMA-TTC 150 kBq/kg
PSMA-TTC 300 kBq/kg
Darolutamide 100 mg/kg, + PSMA-TTC 150 kBq/kg
Darolutamide100 mg/kg, + PSMA-TTC 300 kBq/kg
B
Tumor volume on day 19 in 22Rv1
Vehicle
Daro-lutamide 100 mg/kg
PSMA-TTC (kBq/kg)
PSMA-TTC (kBq/kg) + darolutamide 100 mg/kg
C
22Rv1
Proportion of mice reaching tumor size 500 mm³ (%)
Time (days)
D
Relative body weight change in 22Rv1
Body weight change (%, mean+SD)

Figure 5. Antitumor efficacy of PSMA-TTC and darolutamide in the low PSMA-expressing, androgen-independent 22Rv1 prostate cancer xenograft model. (A) Growth curves of 22Rv1 tumors treated with vehicle, darolutamide (100 mg/kg, BID, p.o.), and/or PSMA-TTC (150 or 300 kBq/kg, single dose, i.v., total protein dose 0.14 mg/kg). **(B)** Volumes of individual 22Rv1 tumors shown in (A) on day 19. Statistical analysis was performed using one-way ANOVA and Šidák's test. ***, p<0.001 vs. vehicle. **(C)** Time to reach a tumor volume of 500 mm³ in the 22Rv1 tumors shown in (A). The darolutamide group was stopped on day 19, the PSMA-TTC 300 kBq/kg and PSMA-TTC 150 kBq/kg + darolutamide groups on day 33, and the PSMA-TTC 300 kBq/kg + darolutamide group on day 51 with 1, 3, 2, and 4 mice with tumor volumes < 500 mm³ at the time of stopping, respectively (indicated with ★ in the graph). Statistical analysis was performed using the log-rank test. ##, p<0.01 vs. respective PSMA-TTC monotherapy; §§, p<0.01 and §§§, p<0.001 vs. darolutamide monotherapy. **(D)** Relative body weight change of the mice shown in (A). BID, twice daily, i.v., intravenously, p.o., per os.

PSMA-TTC shows antitumor efficacy in the high PSMA-expressing C4-2 prostate cancer xenograft model with minimal additional efficacy in combination with darolutamide

- PSMA-TTC monotherapy at 300 kBq/kg showed potent antitumor efficacy in the high PSMA-expressing C4-2 model with a T/C ratio of 0.29 on day 19. Combination treatment with darolutamide did not result in further enhanced antitumor efficacy (T/C 0.33) (**Fig. 6A-B, Table 1**).
- The combination of 150 kBq/kg PSMA-TTC and darolutamide resulted in only slightly better antitumor efficacy than PSMA-TTC monotherapy (**Fig. 6A-B, Table 1**).
- Combination treatment with 300 kBq/kg PSMA-TTC and darolutamide did not extend the time for tumors to reach a volume of 500 mm³ compared with PSMA-TTC monotherapy. A small benefit was observed with 150 kBq/kg PSMA-TTC and darolutamide combination treatment compared with the respective monotherapies (**Fig. 6C, Table 1**).
- All treatments were well tolerated as indicated by increasing or stable mean body weight in all groups throughout the study (**Fig. 6D**).

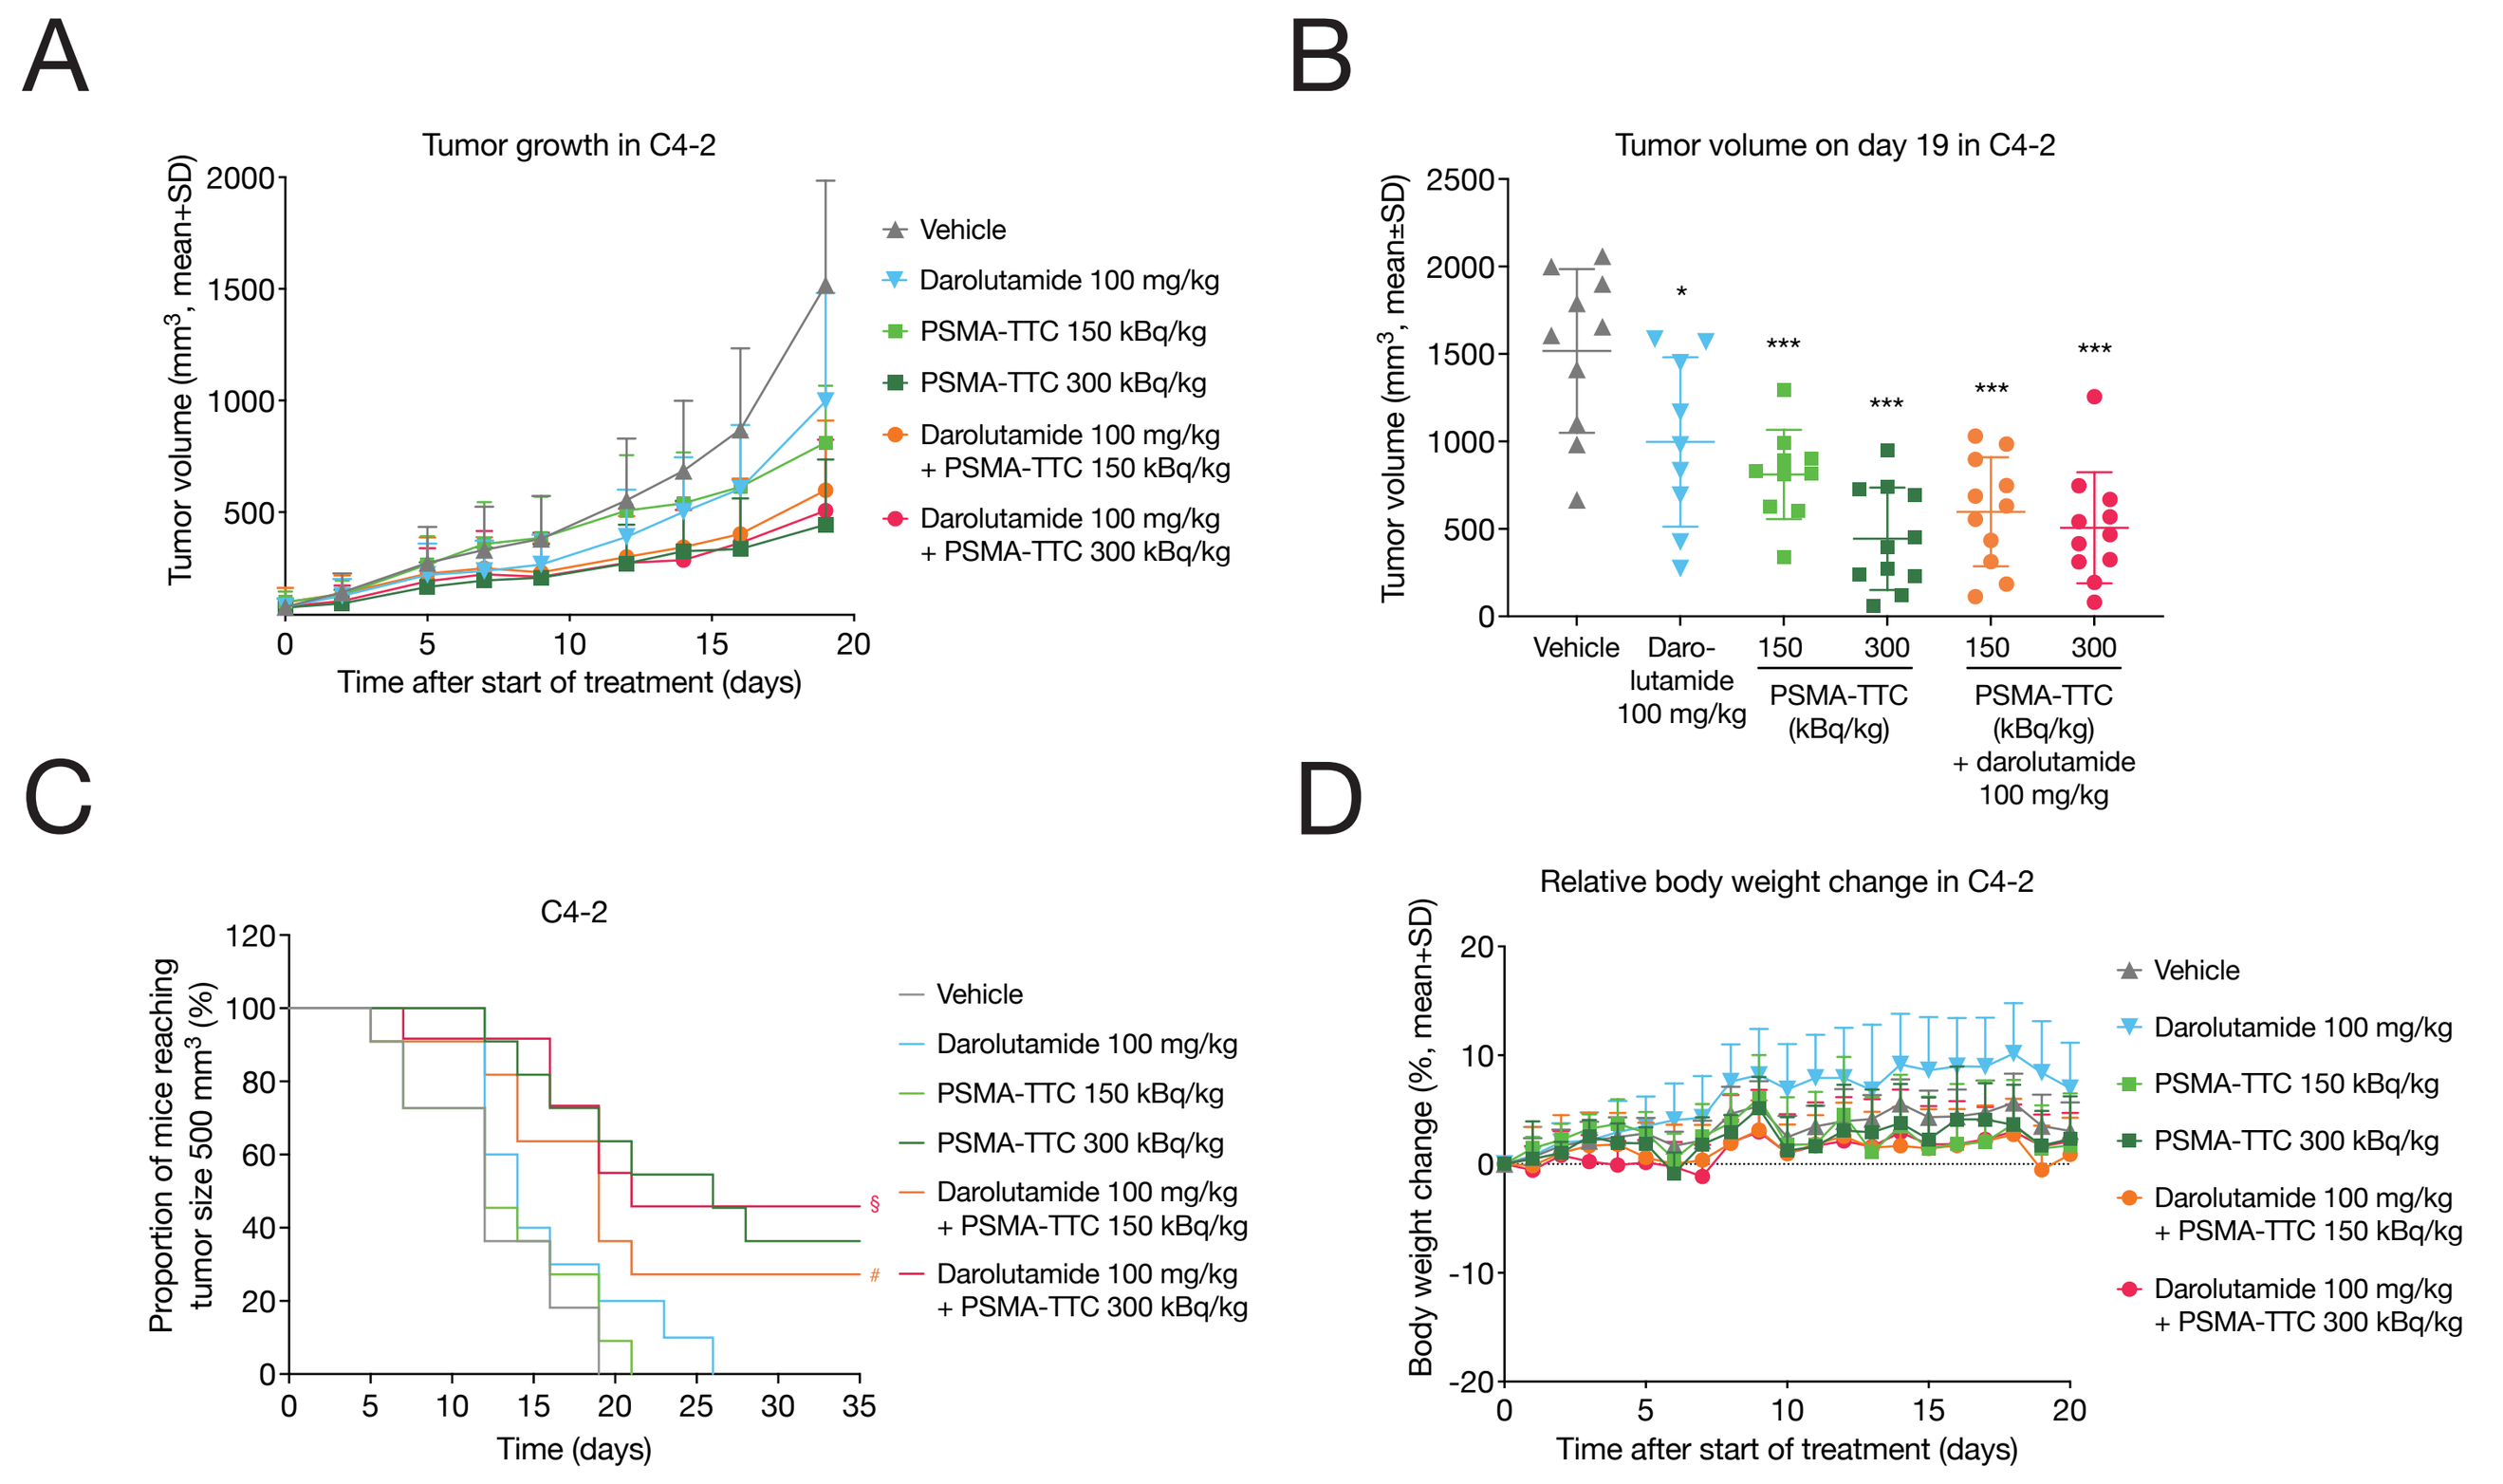
A
Tumor growth in C4-2
Tumor volume (mm³, mean+SD)
Time after start of treatment (days)
Vehicle
Darolutamide 100 mg/kg
PSMA-TTC 150 kBq/kg
PSMA-TTC 300 kBq/kg
Darolutamide 100 mg/kg + PSMA-TTC 150 kBq/kg
Darolutamide 100 mg/kg + PSMA-TTC 300 kBq/kg
B
Tumor volume on day 19 in C4-2
Vehicle
Daro-lutamide 100 mg/kg
PSMA-TTC (kBq/kg)
PSMA-TTC (kBq/kg) + darolutamide 100 mg/kg
C
C4-2
Proportion of mice reaching tumor size 500 mm³ (%)
Time (days)
D
Relative body weight change in C4-2
Body weight change (%, mean+SD)

Figure 6. Antitumor efficacy of PSMA-TTC and darolutamide in the high PSMA-expressing, androgen-independent C4-2 prostate cancer xenograft model. (A) Growth curves of C4-2 tumors treated with vehicle, darolutamide (100 mg/kg, BID, p.o.) and/or PSMA-TTC (150 or 300 kBq/kg, single dose, i.v., total protein dose 0.14 mg/kg). **(B)** Volumes of individual C4-2 tumors shown in (A) on day 19. Statistical analysis was performed using one-way ANOVA and Šidák's test. *, p<0.05; ***, p<0.001 vs. vehicle. **(C)** Time to reach a mean tumor volume of 500 mm³ in C4-2 tumors shown in (A). Statistical analysis was performed using the log-rank test. #, p<0.015 vs. respective PSMA-TTC monotherapy; §, p<0.05 vs. darolutamide monotherapy. **(D)** Relative body weight change of the mice shown in (A). BID, twice daily, i.v., intravenously, p.o., per os.

Table 1. *In vivo* efficacy of PSMA-TTC, darolutamide and their combination in the low PSMA-expressing 22Rv1 and the high PSMA-expressing C4-2 prostate cancer xenograft models on day 19.

Treatment, dose	Treatment/control ratio on day 19		Time to reach tumor size 500 mm³ (median, days)	
	22Rv1	C4-2	22Rv1	C4-2
Vehicle	1	1	12	12
Darolutamide 100 mg/kg	0.76	0.66*	14	14
PSMA-TTC 150 kBq/kg	0.54***	0.53***	14	12
PSMA-TTC 300 kBq/kg	0.30***	0.29***	31.5	26
PSMA-TTC 150 kBq/kg + darolutamide 100 mg/kg	0.30***	0.39***	28	19
PSMA-TTC 300 kBq/kg + darolutamide 100 mg/kg	0.14***	0.33***	50	21

Statistical analysis was performed using one-way ANOVA and Šidák's test. *, p<0.05; ***, p<0.001 vs. vehicle.

CONCLUSIONS

- Darolutamide induces PSMA expression in the low PSMA-expressing, androgen-independent 22Rv1 prostate cancer model *in vitro* and *in vivo*. Combination treatment with PSMA-TTC and darolutamide shows synergistic *in vitro* antitumor efficacy and additive *in vivo* efficacy in this model.
- Monotherapy with PSMA-TTC was highly active in the high PSMA-expressing, androgen-independent C4-2 prostate cancer model. Antitumor activity was only minimally increased in combination with darolutamide *in vitro* and *in vivo*.
- These studies provide a strong rationale for further investigation of the combination of PSMA-TTC and darolutamide in patients with androgen-independent prostate cancer.
- PSMA-TTC is currently under clinical investigation in combination with darolutamide in patients with metastatic CRPC (Phase I, NCT03724747).

References

1. Hagemann *et al. Cancer Biother Radiopharm.* 2020;35:497-510.
2. Hammer *et al. Clin Cancer Res.* 2020;26(8):1985-96.
3. Fizazi *et al. N Engl J Med.* 2019;380(13):1235-46.
4. Smith *et al. J Clin Oncol.* 2022;40:6_suppl:13.
5. Hammer *et al. Cancer Res.* 2017;77(13 Suppl):Abstract nr 5200.
6. Hammer *et al. Cancer Res.* 2018;78(13 Suppl):Abstract nr 844.
7. Hammer *et al. Clin Cancer Res.* 2021;27(15):4367-78.
8. Chou *et al. Pharmacol Rev.* 2006;58:621-81.

FPO

Acknowledgments

Pharmatest is thanked for performing the C4-2 *in vivo* study funded by Bayer AG. We would like to thank Stefan Stargard, Monika Klotz, and Meike Fehder for excellent technical support. Darolutamide (Nubeqa™) is a product of Bayer (bayerhealthcare.com). The antibody moiety used in BAY 2315497 is licensed from Lantheus (www.lantheus.com). Aurexel Life Sciences Ltd. (www.aurexel.com) is thanked for editorial assistance in the preparation of this poster, funded by Bayer AG.

Presented at the AACR Annual Meeting, April 8–13, 2022, New Orleans, LA, USA

Corresponding author: Christoph Schatz, christoph.schatz@bayer.com