3318

PSMA-targeted thorium-227 conjugate (PSMA-TTC) inhibits tumor growth and abnormal bone changes in the intratibial LNCaP xenograft model of bone-metastatic prostate cancer

Christoph Schatz[1], Mari I. Suominen[2], Matias Knuuttila[3], Sabine Zitzmann-Kolbe[1], Jukka Rissanen[2], Sanna-Maria Käkönen[3,4], Urs Hagemann[1], Arne Scholz[1]

[1]Bayer AG, Pharmaceuticals, Berlin, Germany; [2]Pharmatest Services Ltd., Turku, Finland; [3]Aurexel Life Sciences Ltd., Askainen, Finland; [4]Institute of Biomedicine, University of Turku, Turku, Finland

INTRODUCTION

- Prostate-specific membrane antigen (PSMA) is a transmembrane glycoprotein specifically overexpressed on the cell membrane of prostate cancer cells.[1,2]
- PSMA-targeted thorium-227 conjugate (PSMA-TTC, ^{227}Th-pelgifatamab corixetan, BAY 2315497) consists of a human PSMA-targeting antibody covalently linked to a 3,2-HOPO (hydroxypyridinone) chelator moiety, radiolabeled with alpha-particle emitting thorium-227 (**Fig. 1**).[3]
 - PSMA-TTC induces difficult-to-repair clustered DNA double-strand breaks in PSMA-expressing cancer cells.
- Previous preclinical studies have demonstrated PSMA-TTC to be an attractive therapy option for patients with metastatic castration-resistant prostate cancer (mCRPC)[3-4], also in combination with other treatments for CRPC.[5]
- In this study, we investigated the efficacy of PSMA-TTC in the LNCaP intratibial model mimicking prostate cancer metastasized to bone.[6]
 - LNCaP cells are androgen-responsive, prostate-specific antigen (PSA)-secreting, and PSMA-expressing human prostate cancer cells[7], which form mixed osteoblastic/osteolytic lesions when inoculated intratibially into mice.[6]

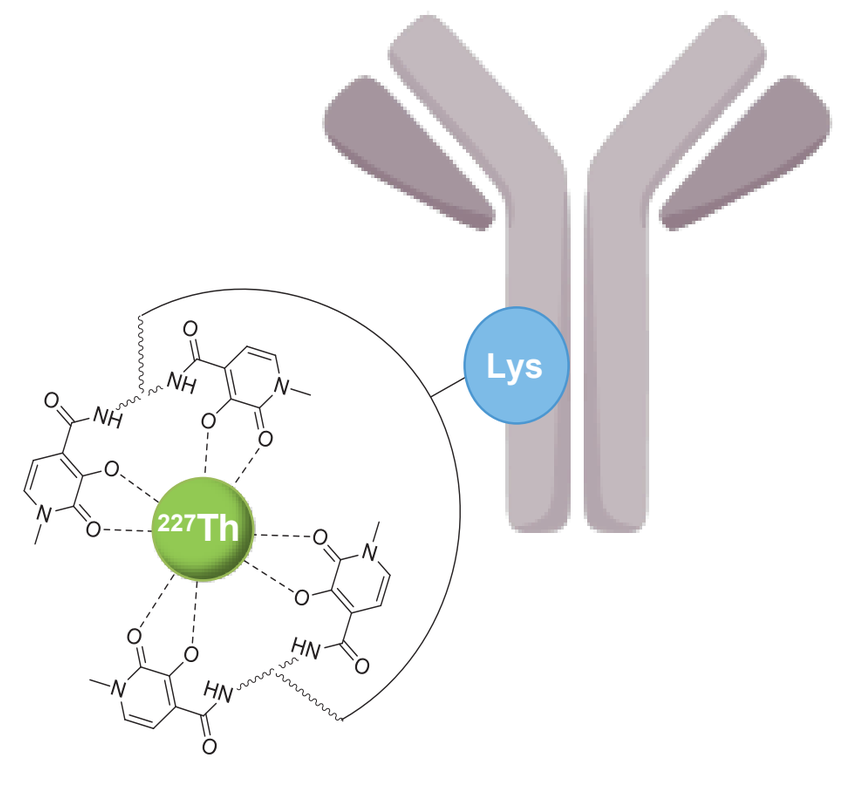
Lys
227Th

Figure 1. Structure of PSMA-TTC (BAY 2315497, ^{227}Th-pelgifatamab corixetan).

METHODS

- LNCaP cells (2 x 10^6 cells/mouse, ATCC) were inoculated into the bone marrow cavity of the right proximal tibia of 5–6-week-old male mice (NOD/SCID, Envigo). Seven weeks after inoculation, the mice were randomized into three treatment groups (n=6-12 mice/group) based on serum PSA levels and treated with a single dose of vehicle (**Group 1**) or PSMA-TTC (**Groups 2 and 3**; 300 kBq/kg, 0.75 mg/kg, i.v.) (**Fig. 2**).
- A human PSA ELISA assay (R&D Systems) and a VICTOR2 Multilabel Counter (PerkinElmer) were used for measuring serum PSA. Tumor-induced abnormal bone area was determined using the Faxitron Specimen Radiographic System MX-20 D12 (Faxitron).
- At sacrifice, hind limbs were collected for analyses of:
 - Radionuclide (PSMA-TTC) uptake in tumor-bearing and non-tumor-bearing tibiae using a gamma counter (**Group 2**, Hidex) or a germanium detector (**Group 3**, Mirion Technologies).
 - Bone microarchitecture, bone formation, and the osteoblast and osteoclast characteristics of tumor-bearing tibiae by bone histomorphometry using an OsteoMeasure7 histomorphometry system (OsteoMetrics). For bone labeling, injections of alizarin (30 mg/kg) and calcein green (10 mg/kg) were given to mice seven and two days before sacrifice, respectively.
 - Bone volume, cross-sectional dimensions, and the bone structure of tumor-bearing tibiae using a SkyScan 1276 high-resolution micro-computed tomography (micro-CT) scanner (Bruker).
- Statistical analyses were performed with the statistical software R (version 3.5.3). Longitudinal data (PSA, X-ray) were log-transformed and analyzed using mixed models and model contrasts. Endpoint measurements of microCT, radionuclide uptake by gamma counter, and bone histomorphometry were analyzed using ANOVA followed by contrasts, or Kruskal-Wallis test and Dunn's test.

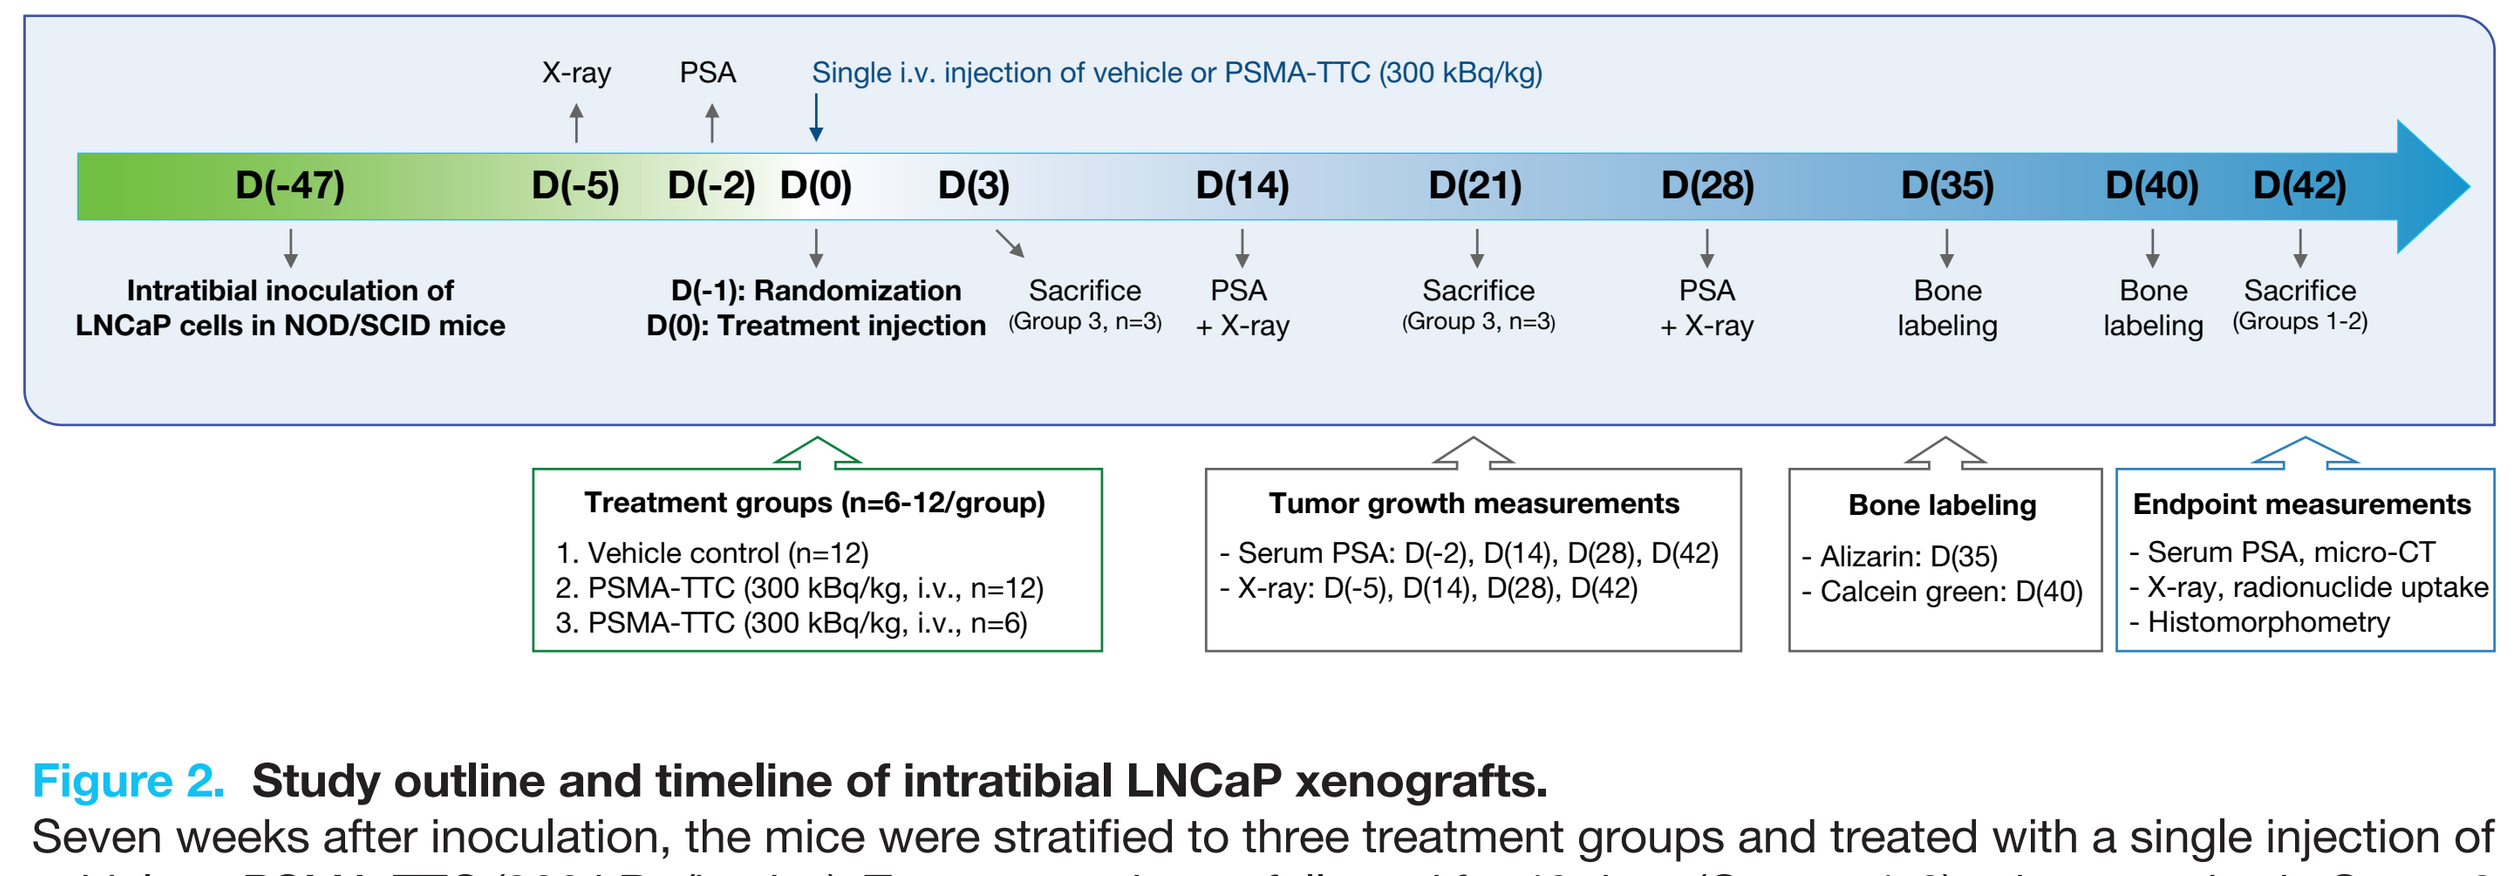
X-ray
PSA
Single i.v. injection of vehicle or PSMA-TTC (300 kBq/kg)
D(-47) D(-5) D(-2) D(0) D(3) D(14) D(21) D(28) D(35) D(40) D(42)
Intratibial inoculation of LNCaP cells in NOD/SCID mice
D(-1): Randomization
D(0): Treatment injection
Sacrifice (Group 3, n=3)
PSA + X-ray
Sacrifice (Group 3, n=3)
PSA + X-ray
Bone labeling
Bone labeling
Sacrifice (Groups 1-2)
Treatment groups (n=6-12/group)
1. Vehicle control (n=12)
2. PSMA-TTC (300 kBq/kg, i.v., n=12)
3. PSMA-TTC (300 kBq/kg, i.v., n=6)
Tumor growth measurements
- Serum PSA: D(-2), D(14), D(28), D(42)
- X-ray: D(-5), D(14), D(28), D(42)
Bone labeling
- Alizarin: D(35)
- Calcein green: D(40)
Endpoint measurements
- Serum PSA, micro-CT
- X-ray, radionuclide uptake
- Histomorphometry

Figure 2. Study outline and timeline of intratibial LNCaP xenografts.
Seven weeks after inoculation, the mice were stratified to three treatment groups and treated with a single injection of vehicle or PSMA-TTC (300 kBq/kg, i.v.). Tumor growth was followed for 42 days (Groups 1-2), whereas mice in Group 3 were sacrificed either 3 or 21 days post injection. D() indicates days after the treatment injection.

RESULTS

A single i.v. injection of PSMA-TTC inhibits intratibial LNCaP tumor growth efficiently

- PSMA-TTC inhibited intratibial LNCaP tumor growth in mice (p<0.001) as evidenced by an 82.6% decrease from the baseline serum PSA value, whereas the relative serum PSA level in the vehicle group increased by up to 380% (**Fig. 3A**).
- At the end of the study on day 42, the mean serum PSA values for vehicle and PSMA-TTC-treated groups were 15.2 ng/mL and 0.7 ng/mL, respectively (**Fig. 3B**).
- PSMA-TTC treatment was well tolerated as no marked decrease in body weight was observed during the study (**Fig. 3C**).

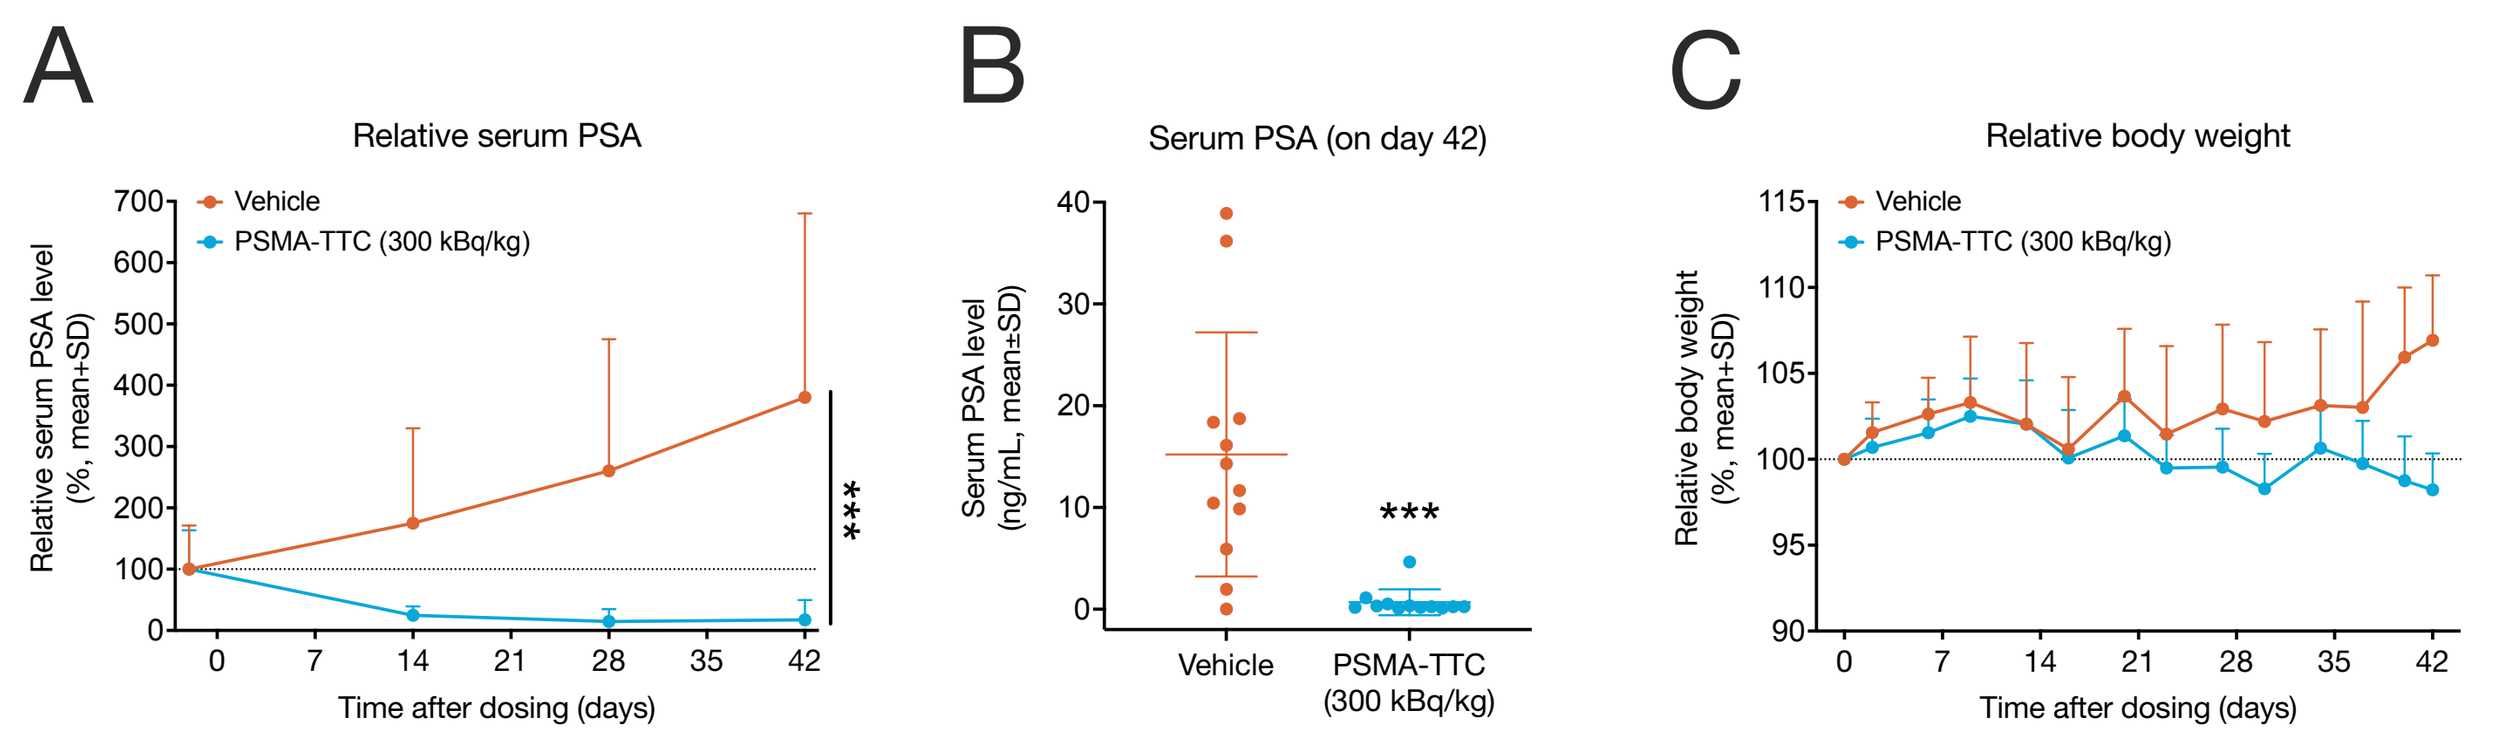
A
Relative serum PSA
Vehicle
PSMA-TTC (300 kBq/kg)
Relative serum PSA level (%, mean+SD)
Time after dosing (days)

B
Serum PSA (on day 42)
Serum PSA level (ng/mL, mean±SD)

Vehicle
PSMA-TTC (300 kBq/kg)
C
Relative body weight
Vehicle
PSMA-TTC (300 kBq/kg)
Relative body weight (%, mean+SD)
Time after dosing (days)

Figure 3. A single i.v. injection of PSMA-TTC inhibits intratibial LNCaP tumor growth efficiently.
The mice were treated with either vehicle or PSMA-TTC (300 kBq/kg, single dose on day 0, i.v.). Blood samples were collected from the saphenous vein of mice (n=12/group) to measure serum PSA levels (**A**) during and (**B**) at the end of the study. (**C**) Relative body weights of mice recorded twice weekly during the study. Dashed lines represent pre-treatment levels. The statistical analyses were performed using mixed models and model contrasts: ***, p<0.001, vs. vehicle.

PSMA-TTC decreases tumor-induced abnormal bone area in tumor-bearing tibiae

- PSMA-TTC treatment decreased tumor-induced abnormal bone area in the tumor-bearing tibiae of mice compared with the vehicle group (43.5% of the vehicle, p<0.001) (**Fig. 4A-B**).
- As seen in the X-ray images, the area of abnormal bone (yellow outlines) was larger in the vehicle-treated group compared with the PSMA-TTC-treated group (**Fig. 4C**).
- Furthermore, in the vehicle group, the tumor-bearing tibiae were heavier (138%, mean weight 87.5 mg, p=0.011) than the non-tumor-bearing tibiae (mean weight 63.2 mg). In the PSMA-TTC-treated mice, no such difference was observed in the tumor-bearing tibiae (103%, 67.2 mg) (**Fig. 4D**).

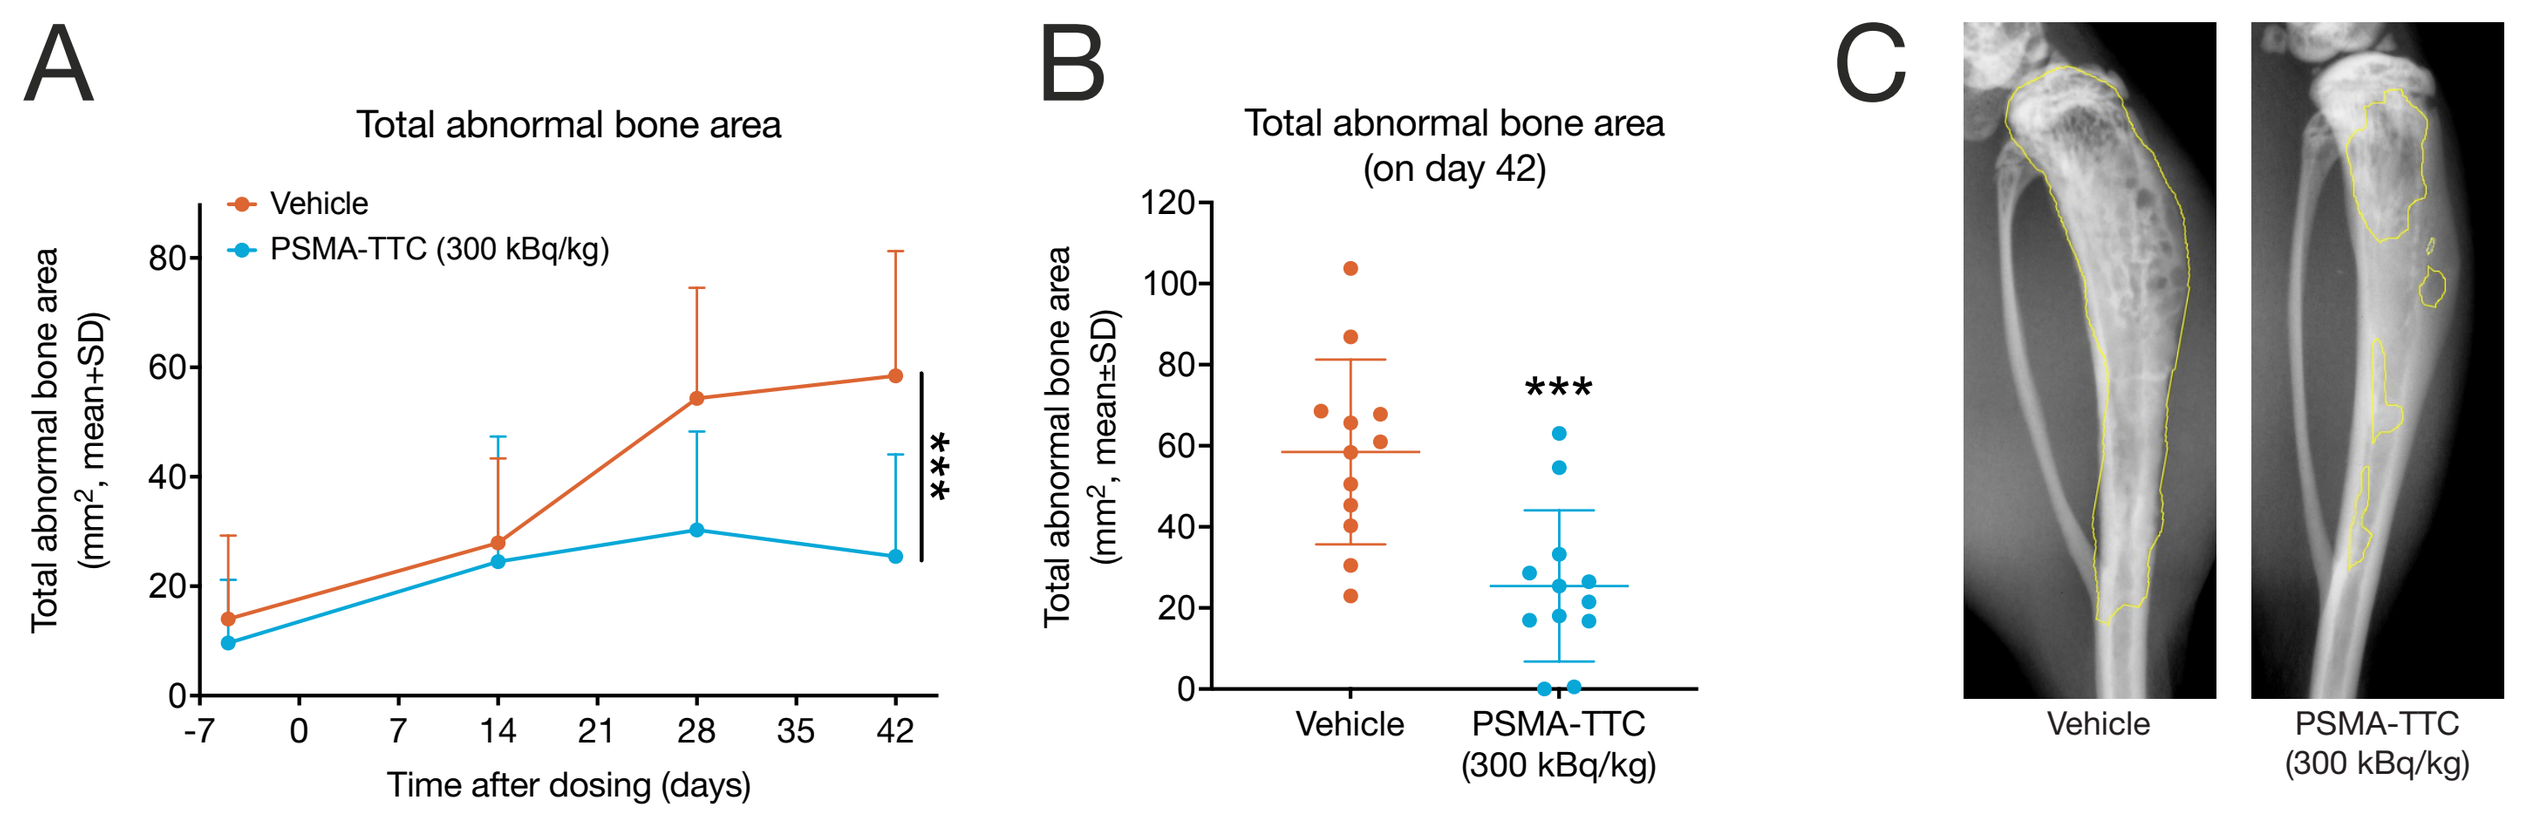
A
Total abnormal bone area
Vehicle
PSMA-TTC (300 kBq/kg)
Total abnormal bone area (mm2, mean+SD)
Time after dosing (days)

B
Total abnormal bone area (on day 42)
Total abnormal bone area (mm2, mean±SD)

Vehicle
PSMA-TTC (300 kBq/kg)
C
Vehicle
PSMA-TTC (300 kBq/kg)

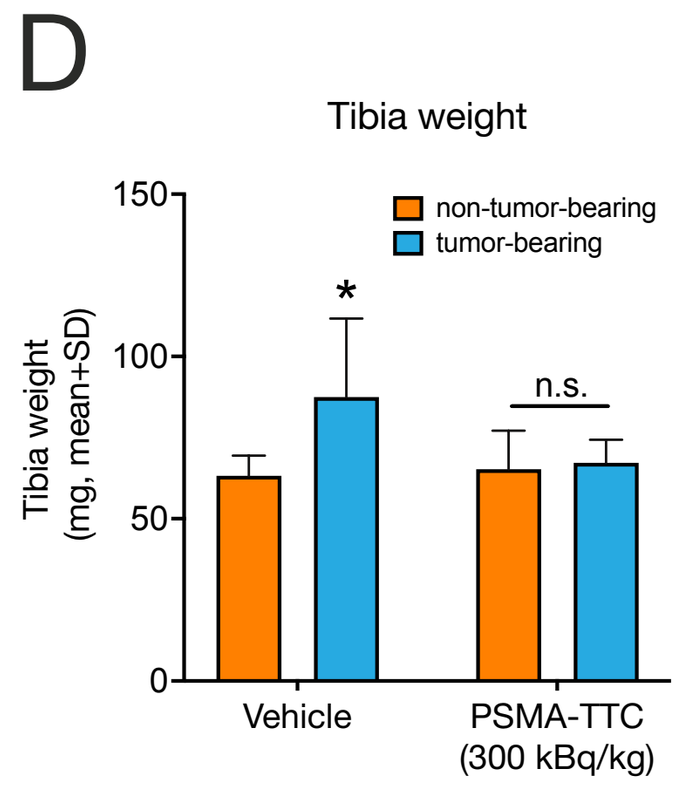
D
Tibia weight
non-tumor-bearing
tumor-bearing
Tibia weight (mg, mean+SD)
*
n.s.
Vehicle
PSMA-TTC (300 kBq/kg)

Figure 4. PSMA-TTC decreases tumor-induced abnormal bone area in tumor-bearing tibiae.
The mice were treated either with vehicle or PSMA-TTC (300 kBq/kg, single dose on day 0, i.v.). Total abnormal bone area of tumor-bearing tibiae (n=12/group) (**A**) during and (**B**) at the end of the study as measured by planar X-ray. (**C**) Representative X-ray images of tumor-bearing tibiae on day 42. Areas of abnormal bone growth are marked with yellow outlines. (**D**) The weights of non-tumor-bearing and tumor-bearing tibiae (n=12/group) at the end of the study. The statistical analyses were performed using (**A-B**) mixed models and model contrasts or (**D**) Kruskal-Wallis test and Dunn's multiple comparisons test: *, p<0.05; ***, p<0.001; n.s., non-significant; vs. vehicle or non-tumor-bearing tibia.

PSMA-TTC targets and accumulates specifically in the tumor microenvironment

- PSMA-TTC uptake in relation to tibia weight was higher (p=0.002) in the tumor-bearing tibiae compared with the non-tumor-bearing tibiae at the end of the study (**Fig. 5A**).
- Similarly, the presence of thorium-227 and its daughter isotope radium-223 was detected in the tumor-bearing tibiae as mearused 3 days and 3 weeks after PSMA-TTC injection. In contrast, the radionuclides were hardly detectable in the non-tumor-bearing tibiae (**Fig. 5B**).

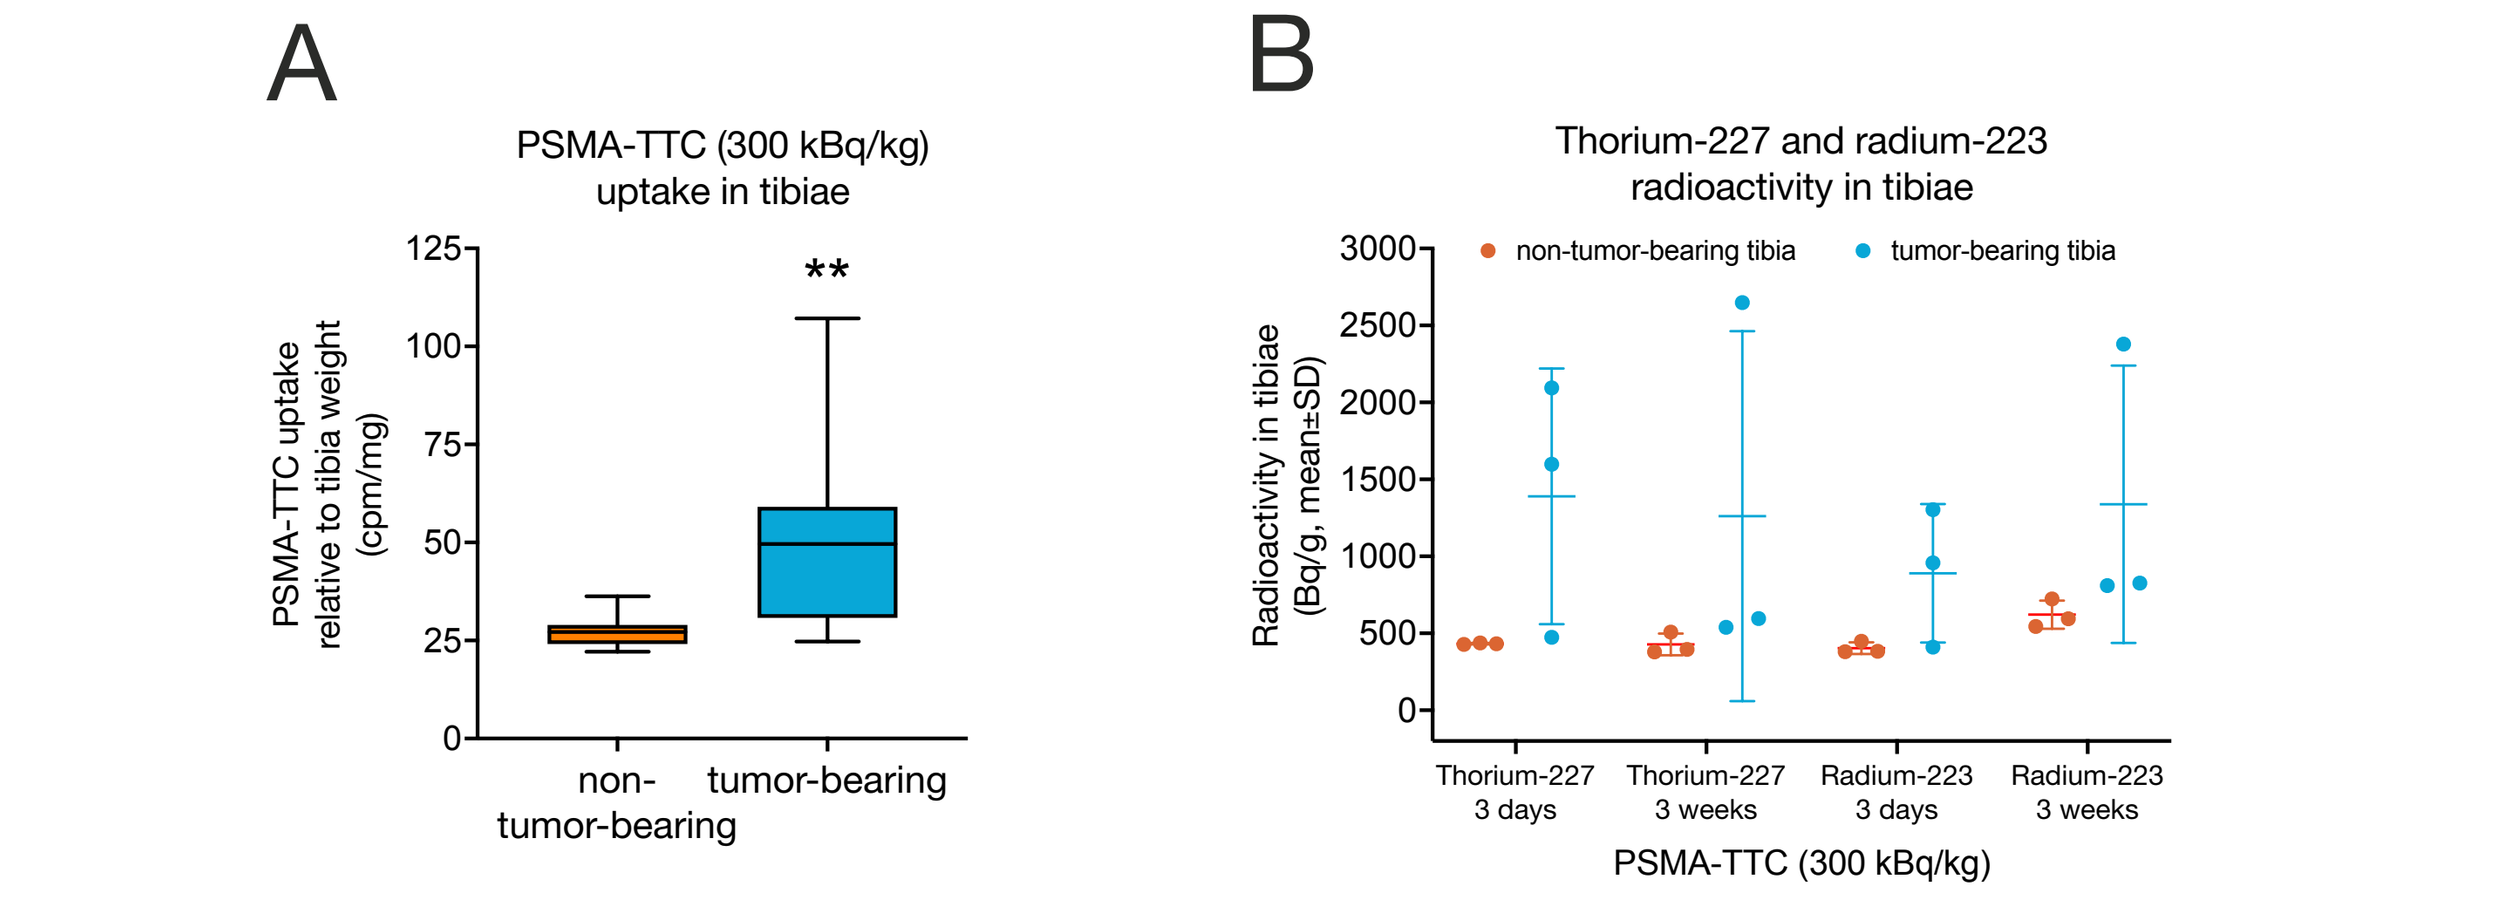
A
PSMA-TTC (300 kBq/kg) uptake in tibiae
PSMA-TTC uptake relative to tibia weight (cpm/mg)
**
non-tumor-bearing
tumor-bearing
B
Thorium-227 and radium-223 radioactivity in tibiae
non-tumor-bearing tibia
tumor-bearing tibia
Radioactivity in tibiae (Bq/g, mean±SD)
Thorium-227 3 days
Thorium-227 3 weeks
Radium-223 3 days
Radium-223 3 weeks
PSMA-TTC (300 kBq/kg)

Figure 5. PSMA-TTC targets and accumulates specifically in the tumor microenvironment.
The mice were treated either with vehicle or PSMA-TTC (300 kBq/kg, single dose on day 0, i.v.). (**A**) PSMA-TTC (300 kBq/kg) uptake as counts per minute (cpm) relative to tibia weight (mg) (Group 2, n=12) and (**B**) thorium-227 and radium-223 radioactivity (Bq) relative to tibia weight (g) (Group 3, n=3 for 3 days or 3 weeks) in non-tumor-bearing and tumor-bearing tibiae were measured using either a gamma counter (**A**) or a germanium detector (**B**), respectively. Error bars (**A**) represent median, 25/75% quartiles, and minimum/maximum). The statistical analyses (**A**) were performed using paired t-test: **, p<0.01, vs. non-tumor-bearing tibia.

PSMA-TTC inhibits trabecular bone formation in tumor-bearing tibiae

- PSMA-TTC decreased osteoblast number in the tumor-bearing tibiae (**Fig. 6A**, p<0.001), but the number of osteoclasts remained unaffected (**Fig. 6B**).
- Dynamic bone histomorphometric analyses demonstrated that PSMA-TTC reduced trabecular mineral apposition rate (**Fig. 6C**, p=0.007), trabecular mineralizing surface (**Fig. 6D**, p=0.006) and trabecular bone formation rate (**Fig. 6E**, p<0.001) in the tumor-bearing tibiae.

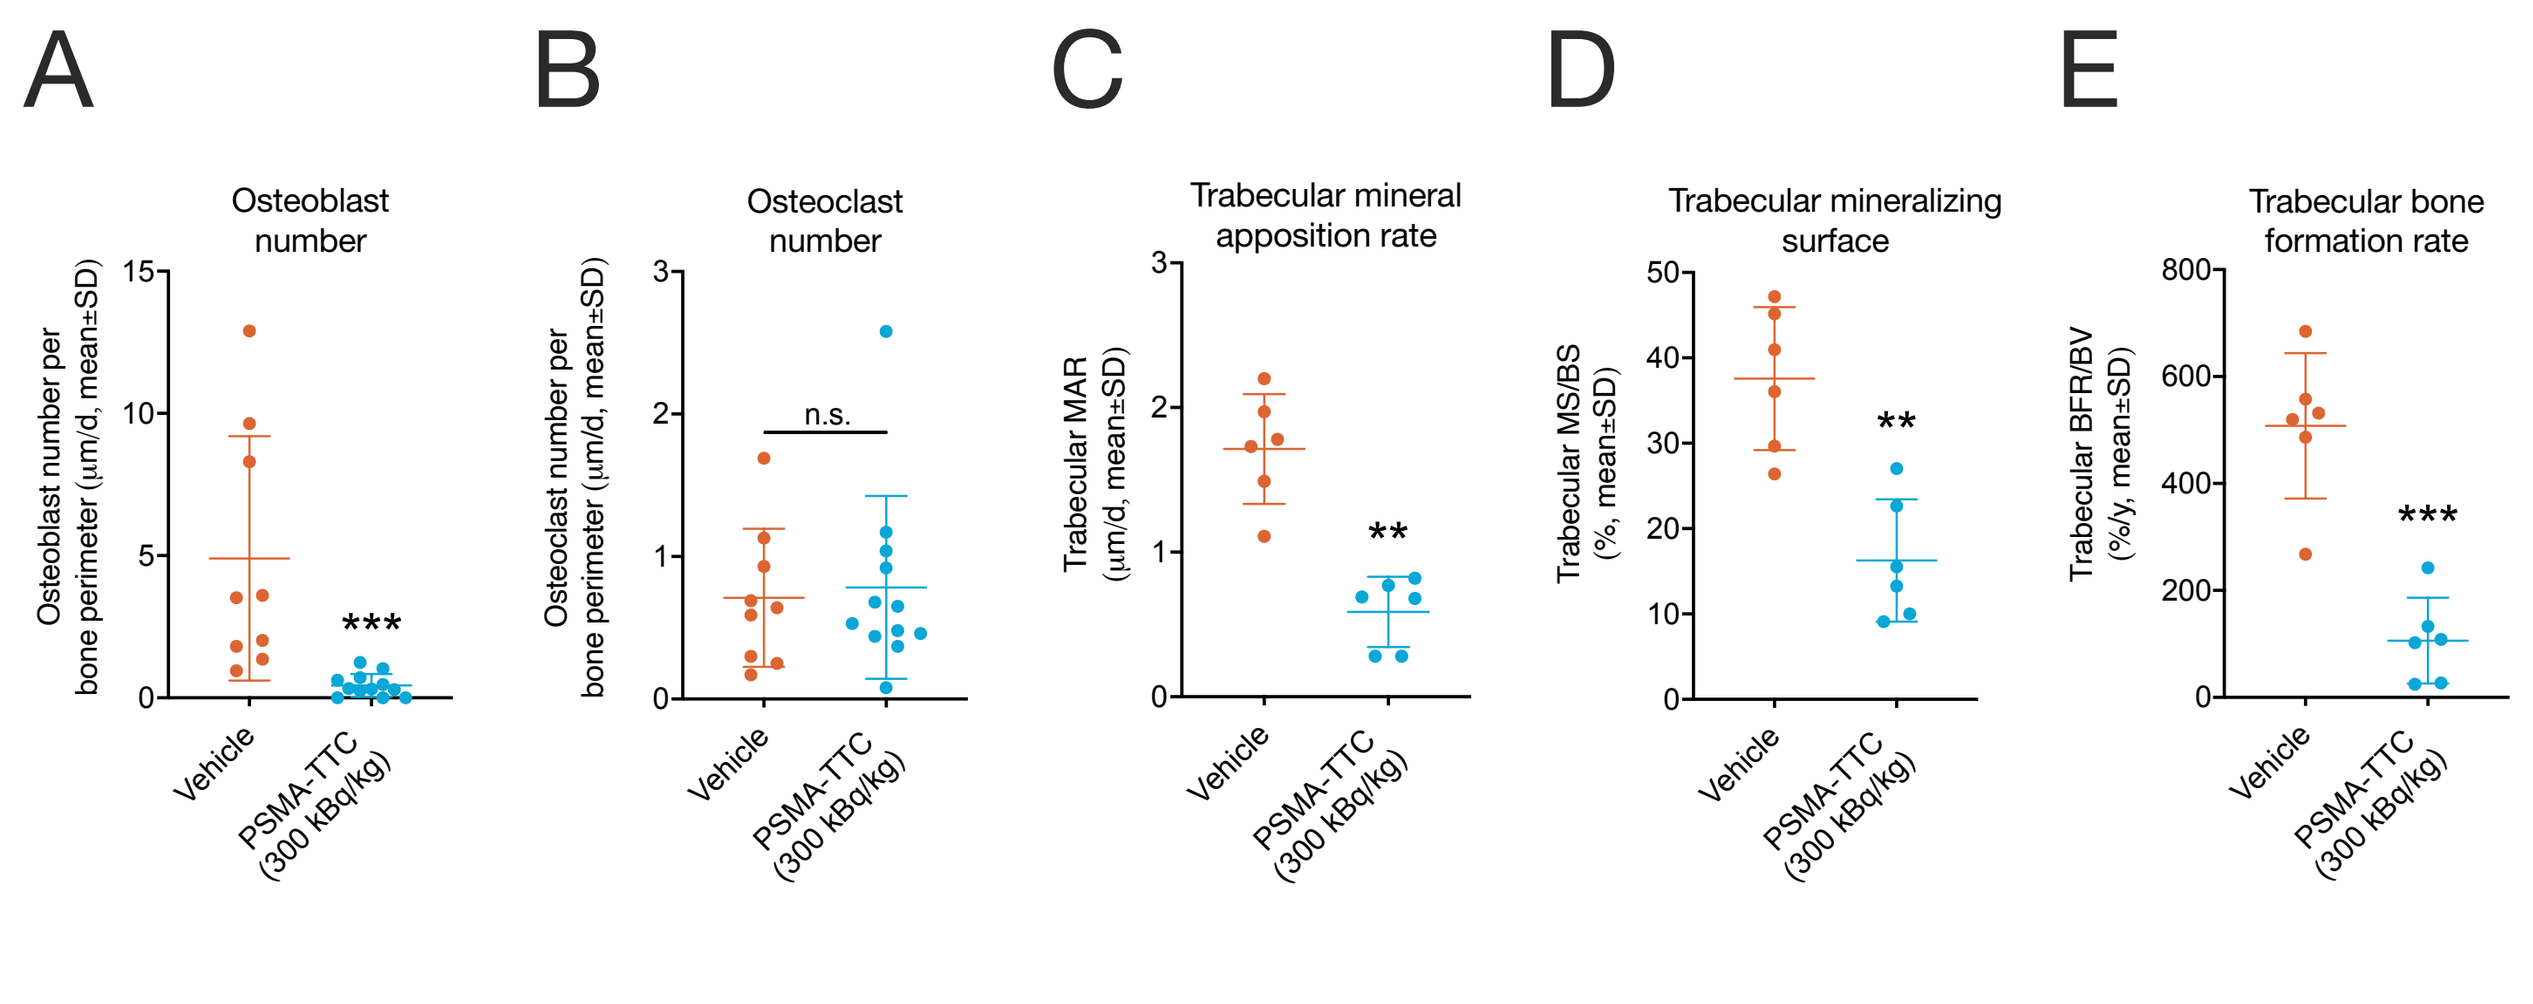
A
Osteoblast number
Osteoblast number per bone perimeter (µm/d, mean±SD)

B
Osteoclast number
Osteoclast number per bone perimeter (µm/d, mean±SD)
n.s.
C
Trabecular mineral apposition rate
Trabecular MAR (µm/d, mean±SD)
**
D
Trabecular mineralizing surface
Trabecular MS/BS (%, mean±SD)
**
E
Trabecular bone formation rate
Trabecular BFR/BV (%/y, mean±SD)

Vehicle
PSMA-TTC (300 kBq/kg)

Figure 6. PSMA-TTC inhibits trabecular bone formation in tumor-bearing tibiae.
The mice were treated either with vehicle or PSMA-TTC (300 kBq/kg, single dose on day 0, i.v.). (**A**) Osteoblast number and (**B**) osteoclast number per bone perimeter in tumor-bearing tibiae (n=9-12/group) analyzed by static bone histomorphometry. Bone formation described as (**C**) trabecular mineral apposition rate (MAR), (**D**) trabecular mineralizing surface per trabecular bone surface (MS/BS), and (**E**) trabecular bone formation rate per bone volume (BFR/BV) in tumor-bearing tibiae (n=6/group) analyzed by dynamic bone histomorphometry. The statistical analyses were performed using Kruskal-Wallis test and Dunn's multiple comparisons test: **, p<0.01; ***, p<0.001; n.s., non-significant; vs. vehicle.

The effect of PSMA-TTC on the bone structure of tumor-bearing tibiae

- PSMA-TTC treatment decreased total bone volume compared with the vehicle group (**Fig. 7A**).
- However, no differences were observed between the groups in trabecular percent bone volume or cortical bone volume (**Fig. 7B-C**).

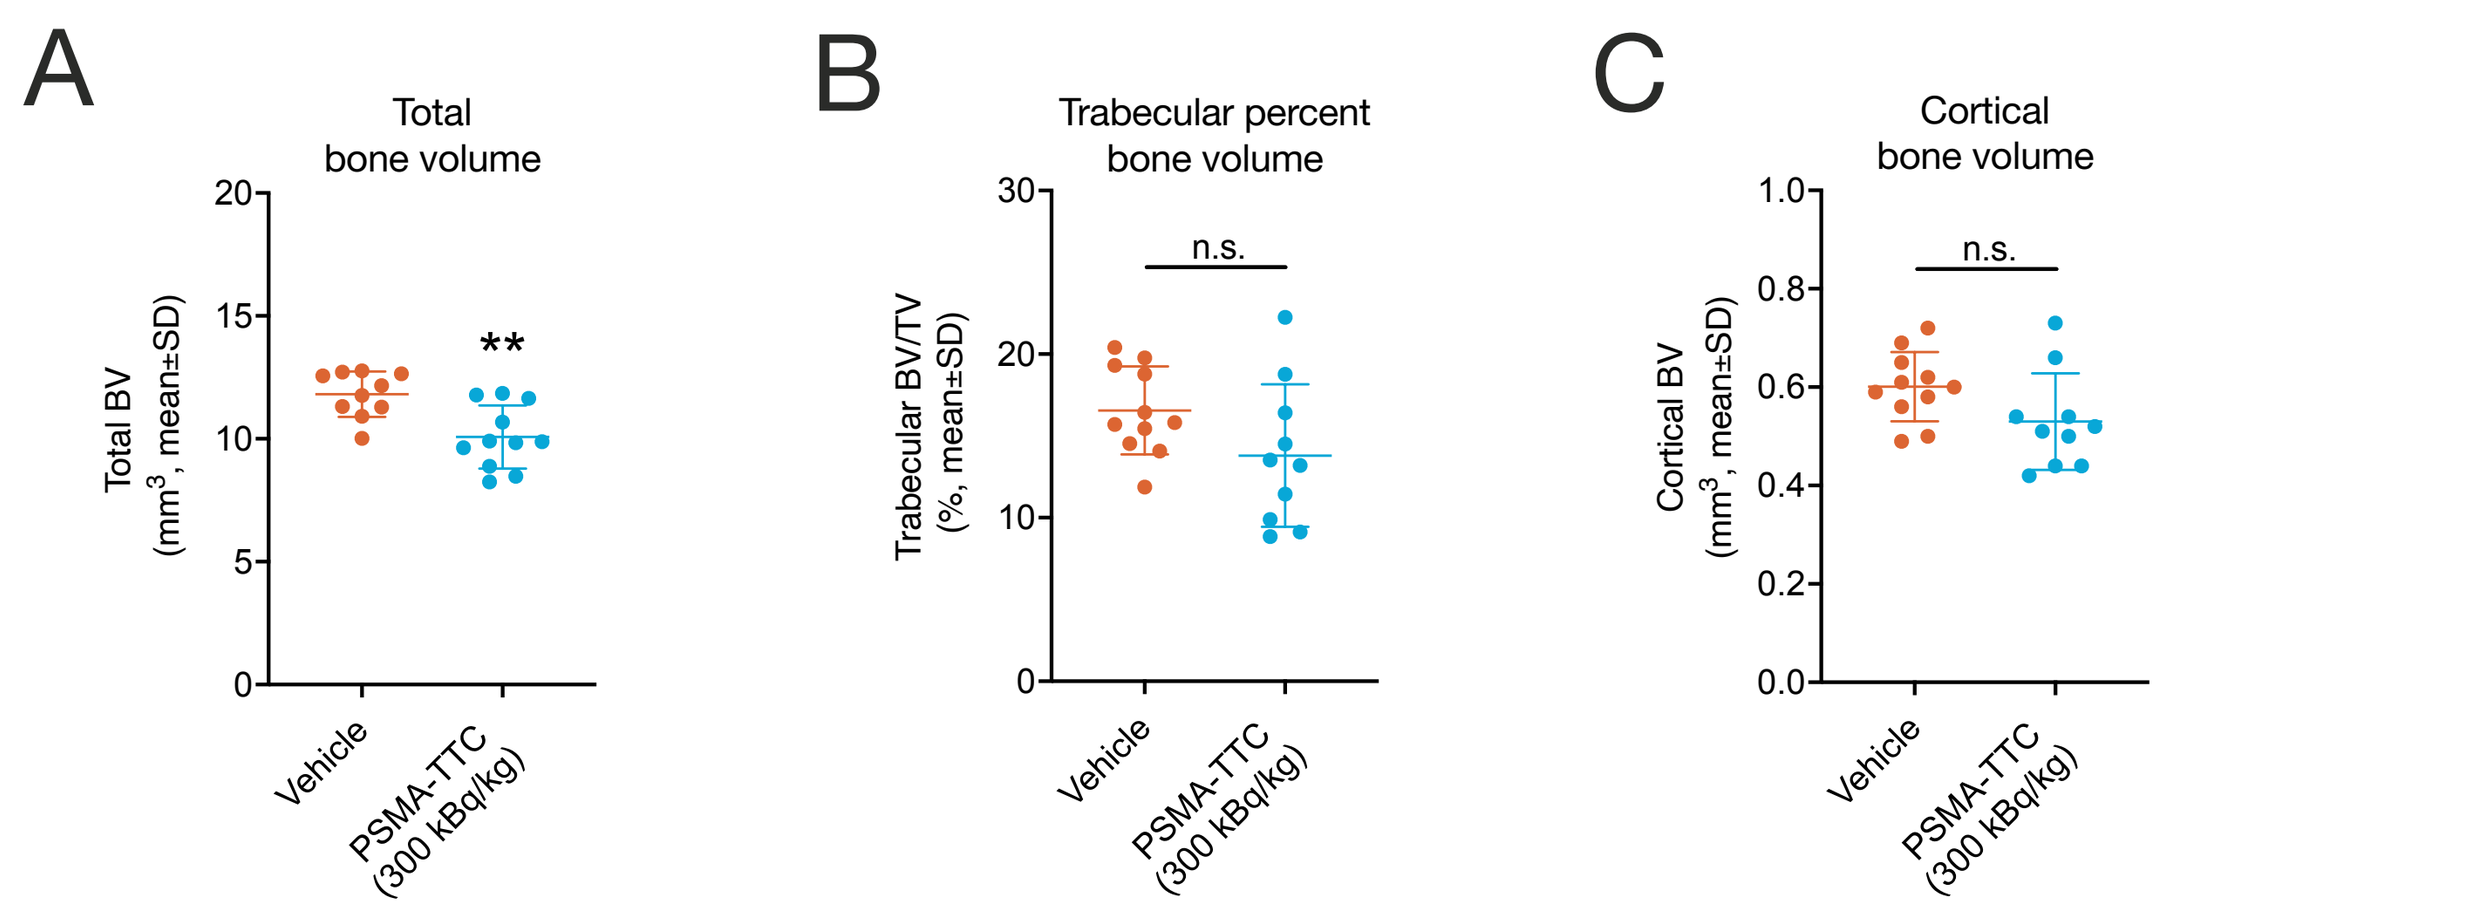
A
Total bone volume
Total BV (mm3, mean±SD)
**
B
Trabecular percent bone volume
Trabecular BV/TV (%, mean±SD)
n.s.
C
Cortical bone volume
Cortical BV (mm3, mean±SD)
n.s.
Vehicle
PSMA-TTC (300 kBq/kg)

Figure 7. The effect of PSMA-TTC on the bone structure of tumor-bearing tibiae.
The mice were treated either with vehicle or PSMA-TTC (300 kBq/kg, single dose on day 0, i.v.). Bone structure described as (**A**) total bone volume, (**B**) trabecular percent bone volume (percent of bone volume with respect to total bone volume, BV/TV), and (**C**) cortical bone volume in tumor-bearing tibiae (n=10-11/group). The statistical analyses were performed using ANOVA followed by contrasts, or Kruskal-Wallis test and Dunn's test: **, p<0.01; n.s., non-significant; vs. vehicle.

CONCLUSIONS

- PSMA-TTC showed robust antitumor efficacy in a mouse model mimicking prostate cancer metastasized to bone. In comparison with the vehicle group, this was evidenced by:
 - a decrease in serum PSA levels
 - reduced tumor-induced abnormal bone area
 - decreased tumor-bearing tibia weight
 - a decrease in disease-driving osteoblasts in tumor-bearing bone
- PSMA-TTC binds to PSMA-expressing cancer cells, delivering a cytotoxic dose of alpha radiation. The decay product of thorium-227, radium-223, is similarly retained in tumor-bearing bone and, thus, may also contribute to the antitumor efficacy in this bone-metastatic model.
- PSMA-TTC as monotherapy and in combination with darolutamide, a recently approved androgen receptor inhibitor for the treatment of nonmetastatic CRPC[8], is currently being investigated in patients with mCRPC in a phase 1 clinical trial (NCT03724747).

References

1. Silver DA *et al. Clin Cancer Res*. 1997; 3(1):81-85.
2. Ruigrok EAM *et al. Pharmaceutics*. 2019; 11(11):560.
3. Hammer S *et al. Clin Cancer Res*. 2019; 26(8):1985-1996.
4. Zitzmann-Kolbe S *et al. J Med Imaging Radiat Sci*. 2019; 50(4):Suppl1:S69-70.
5. Hammer S *et al. Clin Cancer Res*. 2021; 27(15):4367-4378.
6. Suominen MI *et al. Clin Cancer Res*. 2017; 23(15):4335-4346.
7. Ghosh A *et al. Cancer Res*. 2005; 65(3):727-731.
8. Fizazi K *et al. N Engl J Med*. 2009; 380(13):1235-1246.

Acknowledgments

Pharmatest personnel is thanked for the technical performance of the study. Meike Fehder and Vu Tung Le are acknowledged for helping in logistics and data analysis. The antibody moiety used in BAY 2315497 is licensed from Lantheus (www.lantheus.com). Aurexel Life Sciences Ltd. (www.aurexel.com) is thanked for editorial assistance in the preparation of this poster funded by Bayer AG.

FPO

Presented at the AACR Annual Meeting, April 8 – 13, 2022, New Orleans, LA, USA

Presenting author: Arne Scholz (arne.scholz@bayer.com)