The Evaluation of Knee Pain and Degenerative Changes at the Tissue, Cellular and Molecular Levels in a Rat Model of Post-Traumatic Knee Osteoarthritis

ASBMR 2022
Poster SUN-626

Jukka Morko[1], Jukka Vääräniemi[1], Anniina Lehto[1], Jaakko Lehtimäki[1], Laura Honkanen[1], Minna Auranen[1], Yvonne Konkol[1], Hanae Toyonaga[2], and Jukka P. Rissanen[1]

[1] Pharmatest Services, Turku, Finland; [2] Astellas Innovation Management LLC, Cambridge, MA, USA

E-mail correspondence to Jukka Morko (jukka.morko@pharmatest.com)

Introduction

Several experimental animal models have been developed for human osteoarthritis (OA) and used to study the preclinical efficacy of disease and symptom modifying OA drug candidates.[1,2] These animal models have been characterized and the preclinical efficacy of OA drug candidates has been studied by using various microscopic scoring systems and joint pain assessments. The histopathology initiative of Osteoarthritis Research Society International (OARSI) presented recommendations for histological OA assessment in different species in order to standardize the histopathological OA evaluation. Recommendations for the histopathological evaluation of knee joints in rat OA models focus on degenerative changes in articular cartilage, synovium, joint capsule and growth plate at the tissue and cellular levels and on one degenerative change in articular cartilage at the molecular level, namely a reduction in proteoglycan content.[3]

Aim of the Study

We have characterized OA-related knee pain and degenerative changes in knee joints in rat OA models used frequently in preclinical efficacy studies earlier.[4] In this study, these analyses were completed with immunohistochemistry (IHC) staining and semi-quantification of immunostaining distribution in articular cartilage in a rat model of post-traumatic knee OA. The post-traumatic knee OA was induced by surgical medial meniscal tear (MMT) and medial collateral ligament transection (MCLT) in young adult male rats. The distribution of type II collagen and transcription factor SOX9 was studied as an example for cartilaginous extracellular matrix and chondrocytic proteins.

Materials and Methods

Animal experimentation: Unilateral post-traumatic OA was induced by the surgical MMT + MCLT operation in male Lewis rats at 3 months of age (body weight 300-338 g).[5] OA-related knee pain was assessed before the operation and weekly after the operation for 5 weeks. Rats were terminated, and their knee joints were harvested for histological evaluation. This protocol was approved by the Project Authorisation Board, Regional State Administrative Agency for Southern Finland, Hämeenlinna, Finland.

Experimental analysis: OA-related knee pain was analyzed as a reduction in static weight bearing using Incapacitace Tester (Linton Instrumentation) and as static mechanical/tactile allodynia using von Frey monofilaments (1-15g; Aesthesio; DanMic Global). The histopathological assessment of OA was performed according to the recommendations of the OARSI histopathology initiative.[3] IHC was applied to detect the distribution of type II collagen (Cat #M2139; Hycult Biotech) and transcription factor SOX9 (Cat #EPR14335; Abcam) in articular cartilage. The area of type II collagen immunostaining was quantified as a fraction of articular cartilage area, and the number of chondrocytes exhibiting SOX9 immunostaining as a fraction of total number of chondrocytes.

Statistical analysis: All data is presented as mean +/- standard error of mean (n = 9-12 per group). If the assumptions of parametric tests were fulfilled as such or after data transformations, one-way analysis of variance (ANOVA) and linear contrasts of means were used for statistical evaluation. Otherwise, statistical significances were evaluated by non-parametric Kruskal-Wallis and Dunn's tests.

Body Weight and OA-Related Knee Pain

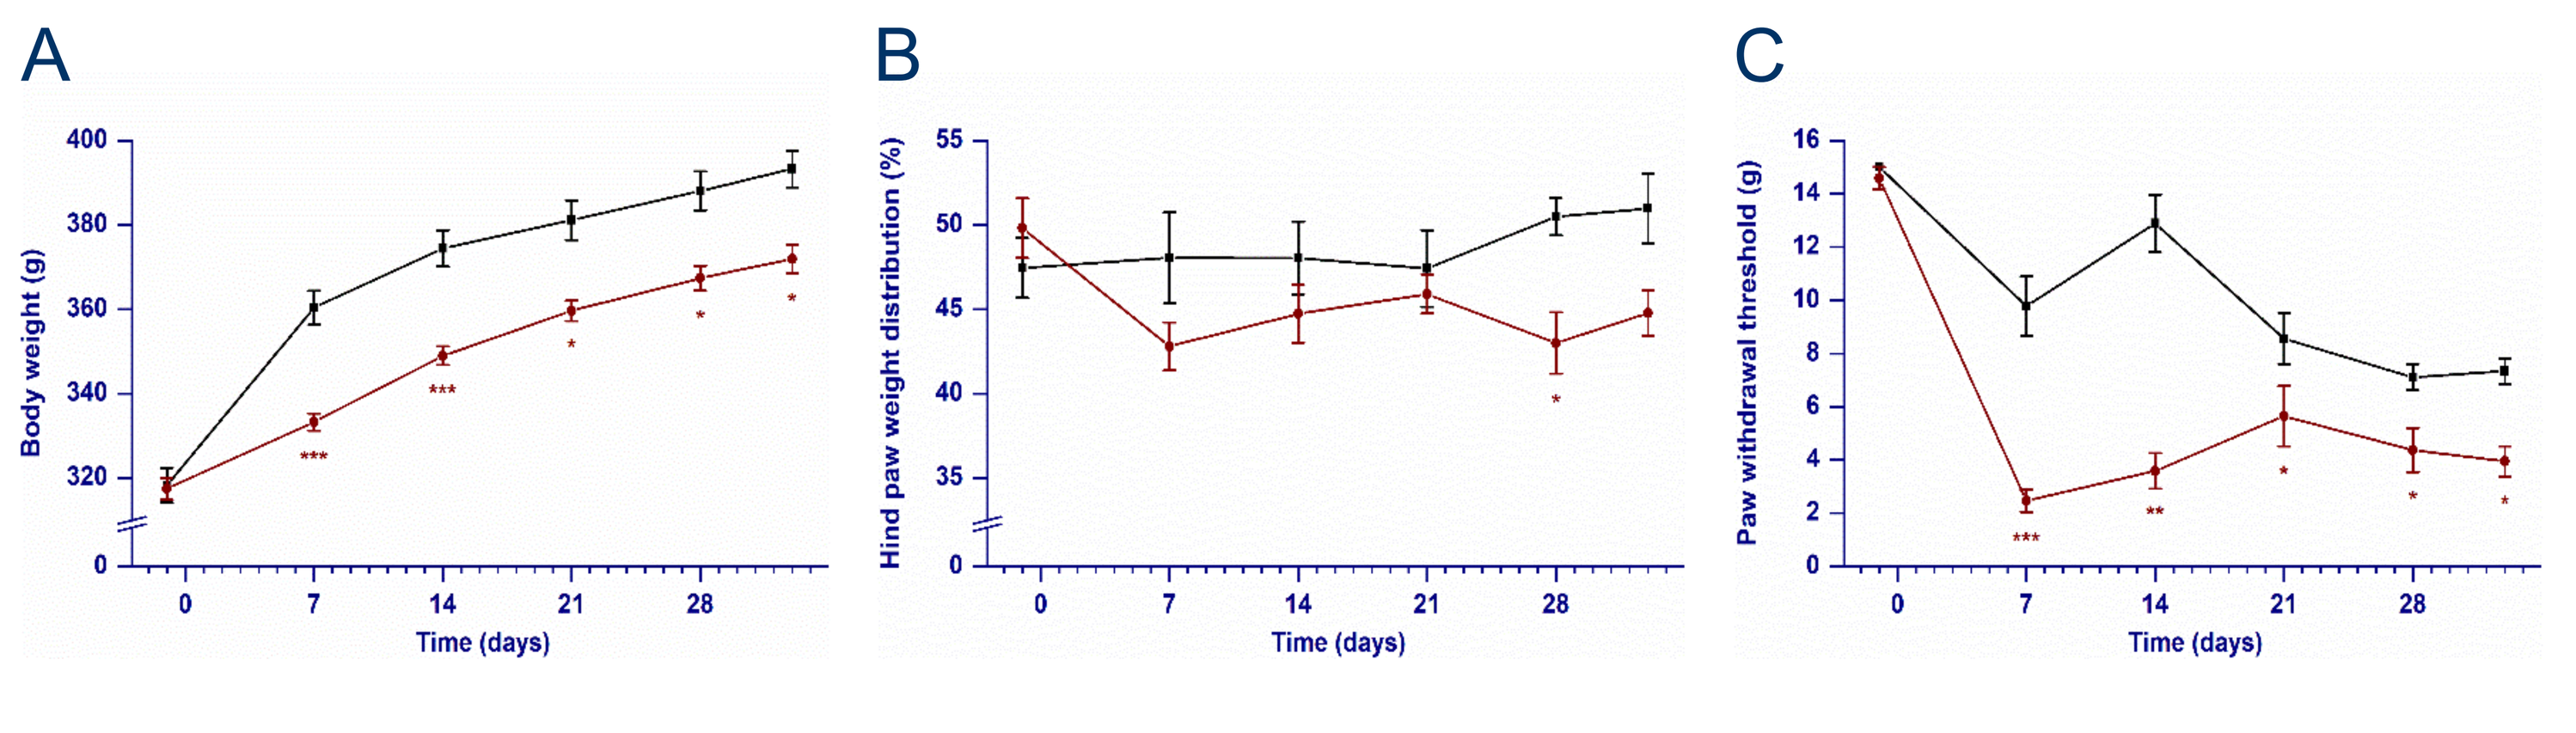
A
B
C
Body weight (g)
Hind paw weight distribution (%)
Paw withdrawal threshold (g)
Time (days)

FIGURE 1. Body weight, static weight bearing, and static mechanical/tactile allodynia over the in-life phase of the study. (A) Body weight (g). (B) Relative hind paw weight bearing on ipsilateral right hind limb (%). (C) Paw withdrawal threshold in ipsilateral right hind limb (g). Black line = intact control rats; Red line = MMT + MCLT operated rats; Red asterisks = significance between MMT + MCLT and intact control rats; * $p < 0.05$; ** $p < 0.01$; *** $p < 0.001$

Histological OA Assessment

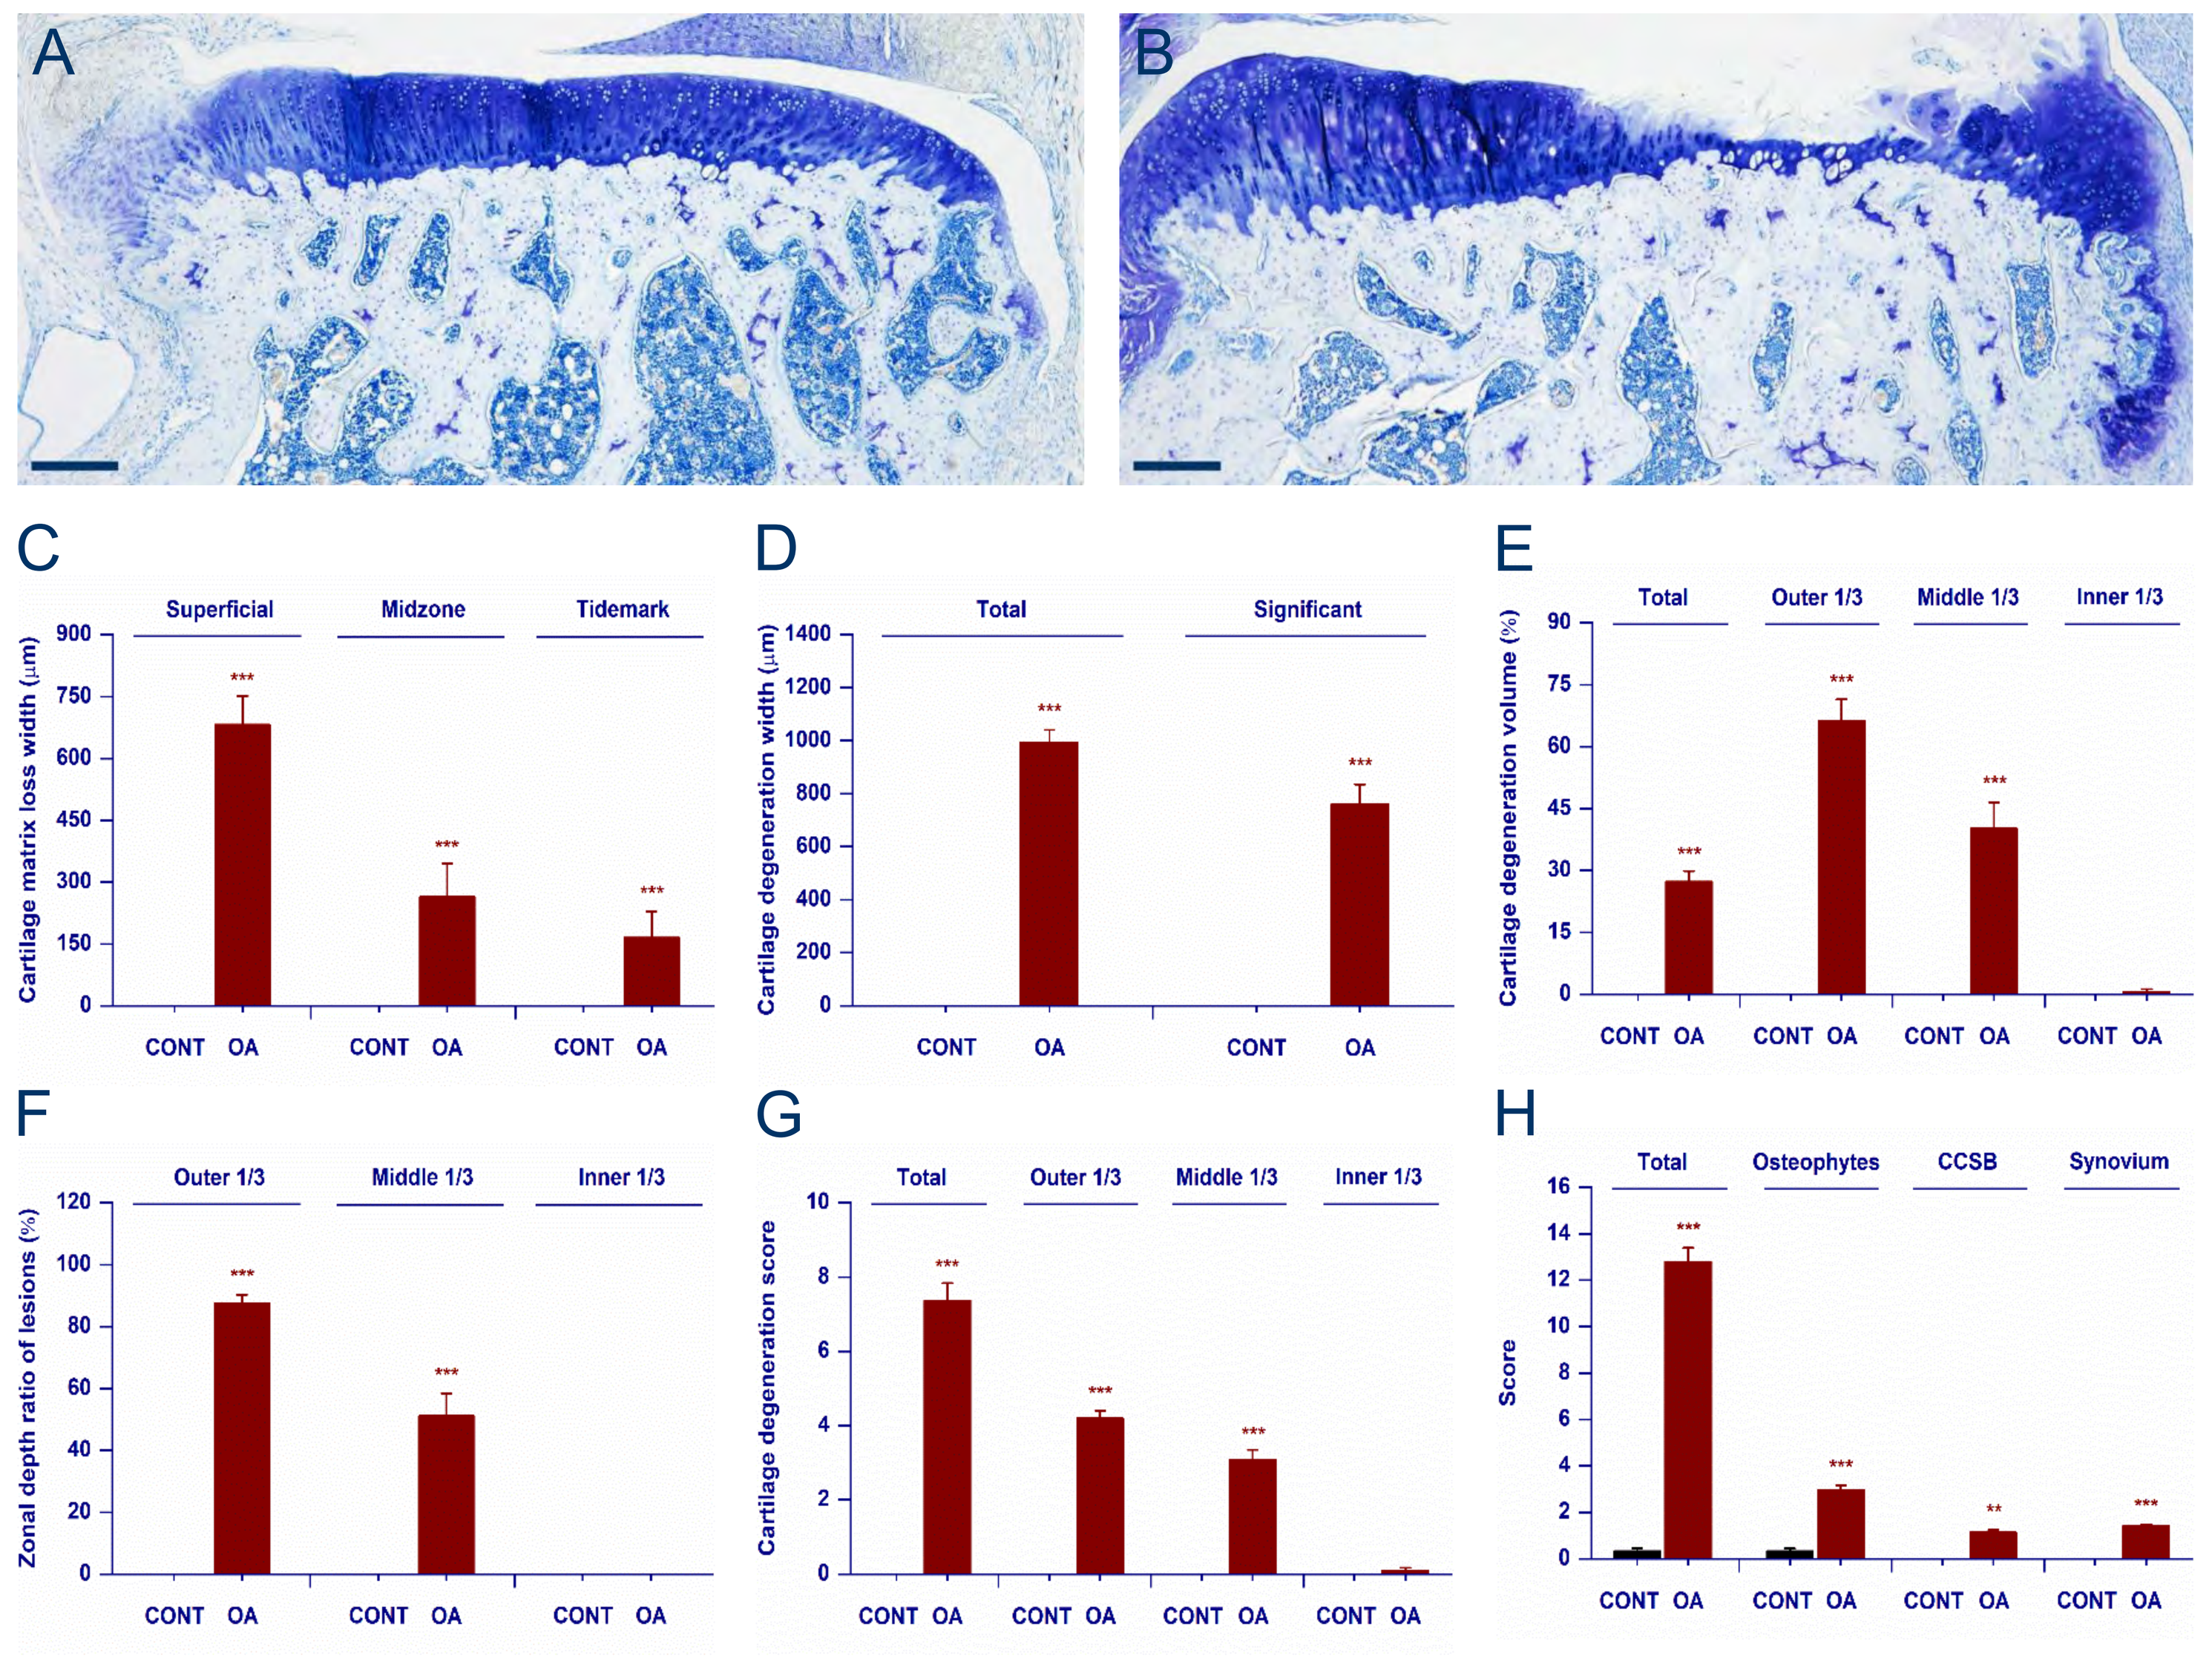
A
B
C
Superficial
Midzone
Tidemark
Cartilage matrix loss width (µm)
D
Total
Significant
Cartilage degeneration width (µm)
E
Total
Outer 1/3
Middle 1/3
Inner 1/3
Cartilage degeneration volume (%)
F
Zonal depth ratio of lesions (%)
G
Cartilage degeneration score
H
Osteophytes
CCSB
Synovium
Score
CONT
OA

FIGURE 2. Histological assessment of OA changes in the medial tibial plateau of ipsilateral right knee. (A) Representative histological image of intact control rats and (B) MMT + MCLT operated rats. A bar in panels A and B corresponds to 250 µm. (C) Cartilage matrix loss width (µm) in superficial layer, midzone and tidemark. (D) Total and significant cartilage degeneration width (µm). (E) Cartilage degeneration volume (%) in medial tibial plateau and in its outer, middle and inner 1/3. (F) Zonal depth ratio of lesions (%) in outer, middle and inner 1/3. (G) Cartilage degeneration score in medial tibial plateau and in its outer, middle and inner 1/3. (H) Total, osteophyte, calcified cartilage and subchondral bone (CCSB), and synovial score. CONT = intact control rats; OA = MMT + MCLT operated rats; Red asterisks = significance between MMT + MCLT and intact control rats; ** $p < 0.01$; *** $p < 0.001$

Type II Collagen Immunostaining

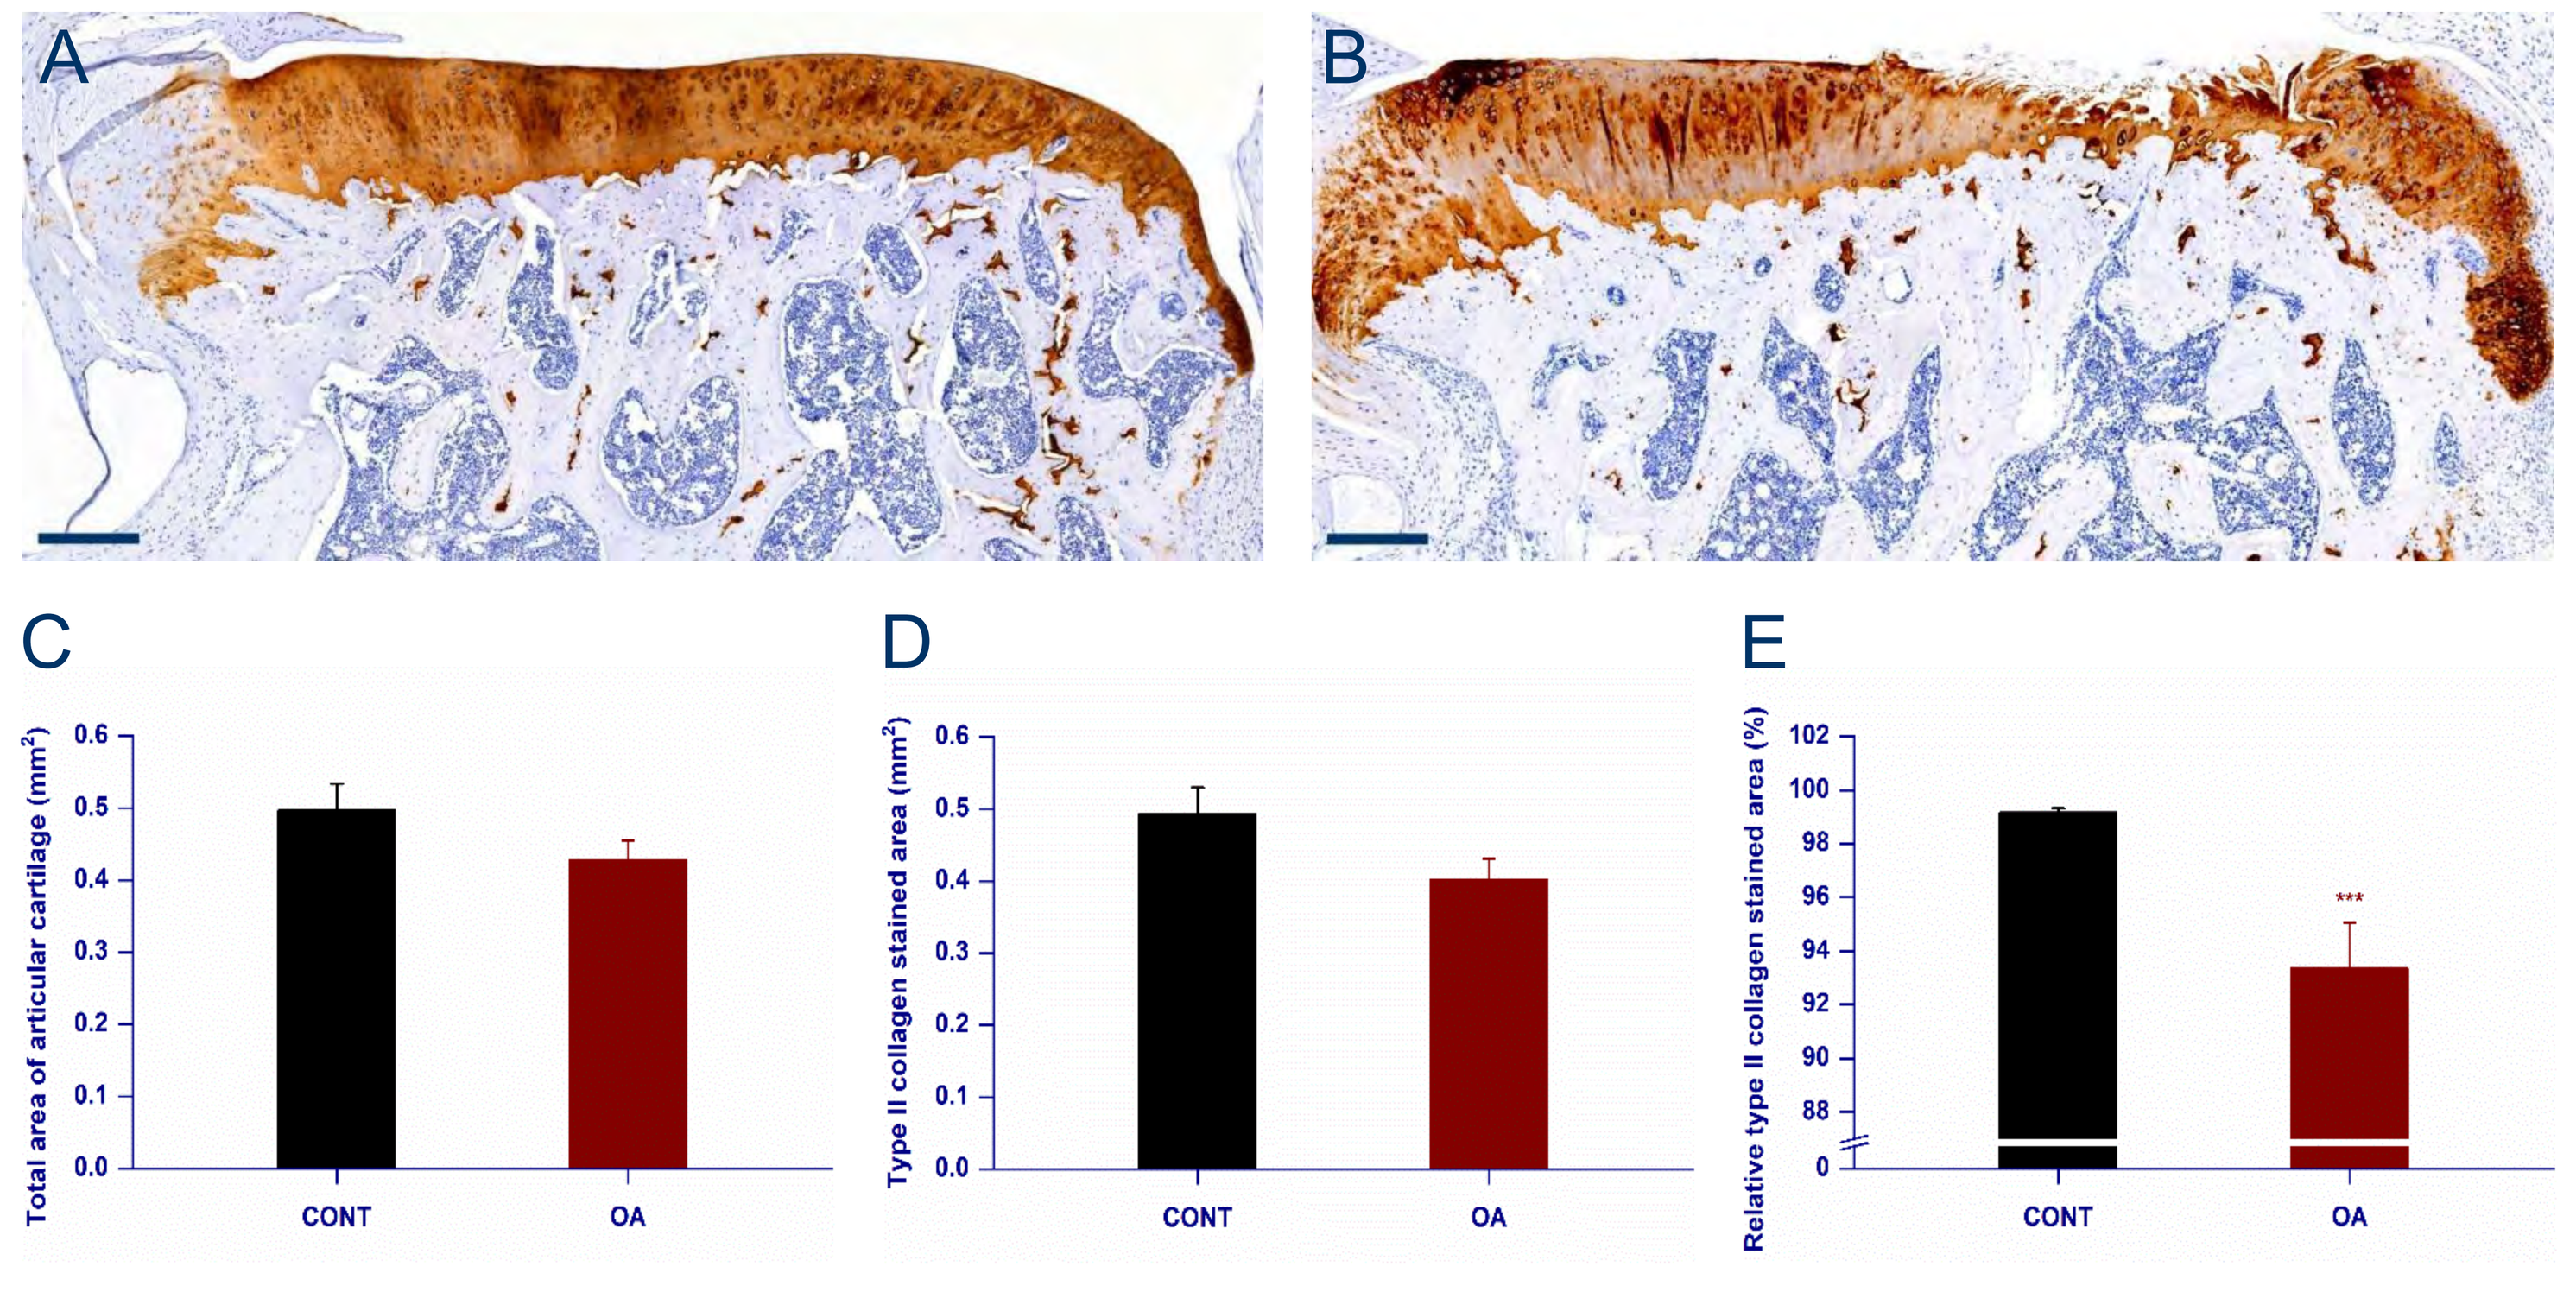
A
B
C
Total area of articular cartilage (mm²)
D
Type II collagen stained area (mm²)
E
Relative type II collagen stained area (%)
CONT
OA

FIGURE 3. Distribution of type II collagen immunostaining in the articular cartilage of medial tibial plateau in ipsilateral right knee. (A) Representative histological image of intact control rats and (B) MMT + MCLT operated rats. A bar in panels A and B corresponds to 250 µm. (C) Total area of existing articular cartilage (mm^2). (D) Area of articular cartilage stained with type II collagen antibodies (mm^2). (E) Relative area of articular cartilage stained with type II collagen antibodies (%). CONT = intact control rats; OA = MMT + MCLT operated rats; Red asterisks = significance between MMT + MCLT and intact control rats; *** $p < 0.001$

Transcription Factor SOX9 Immunostaining

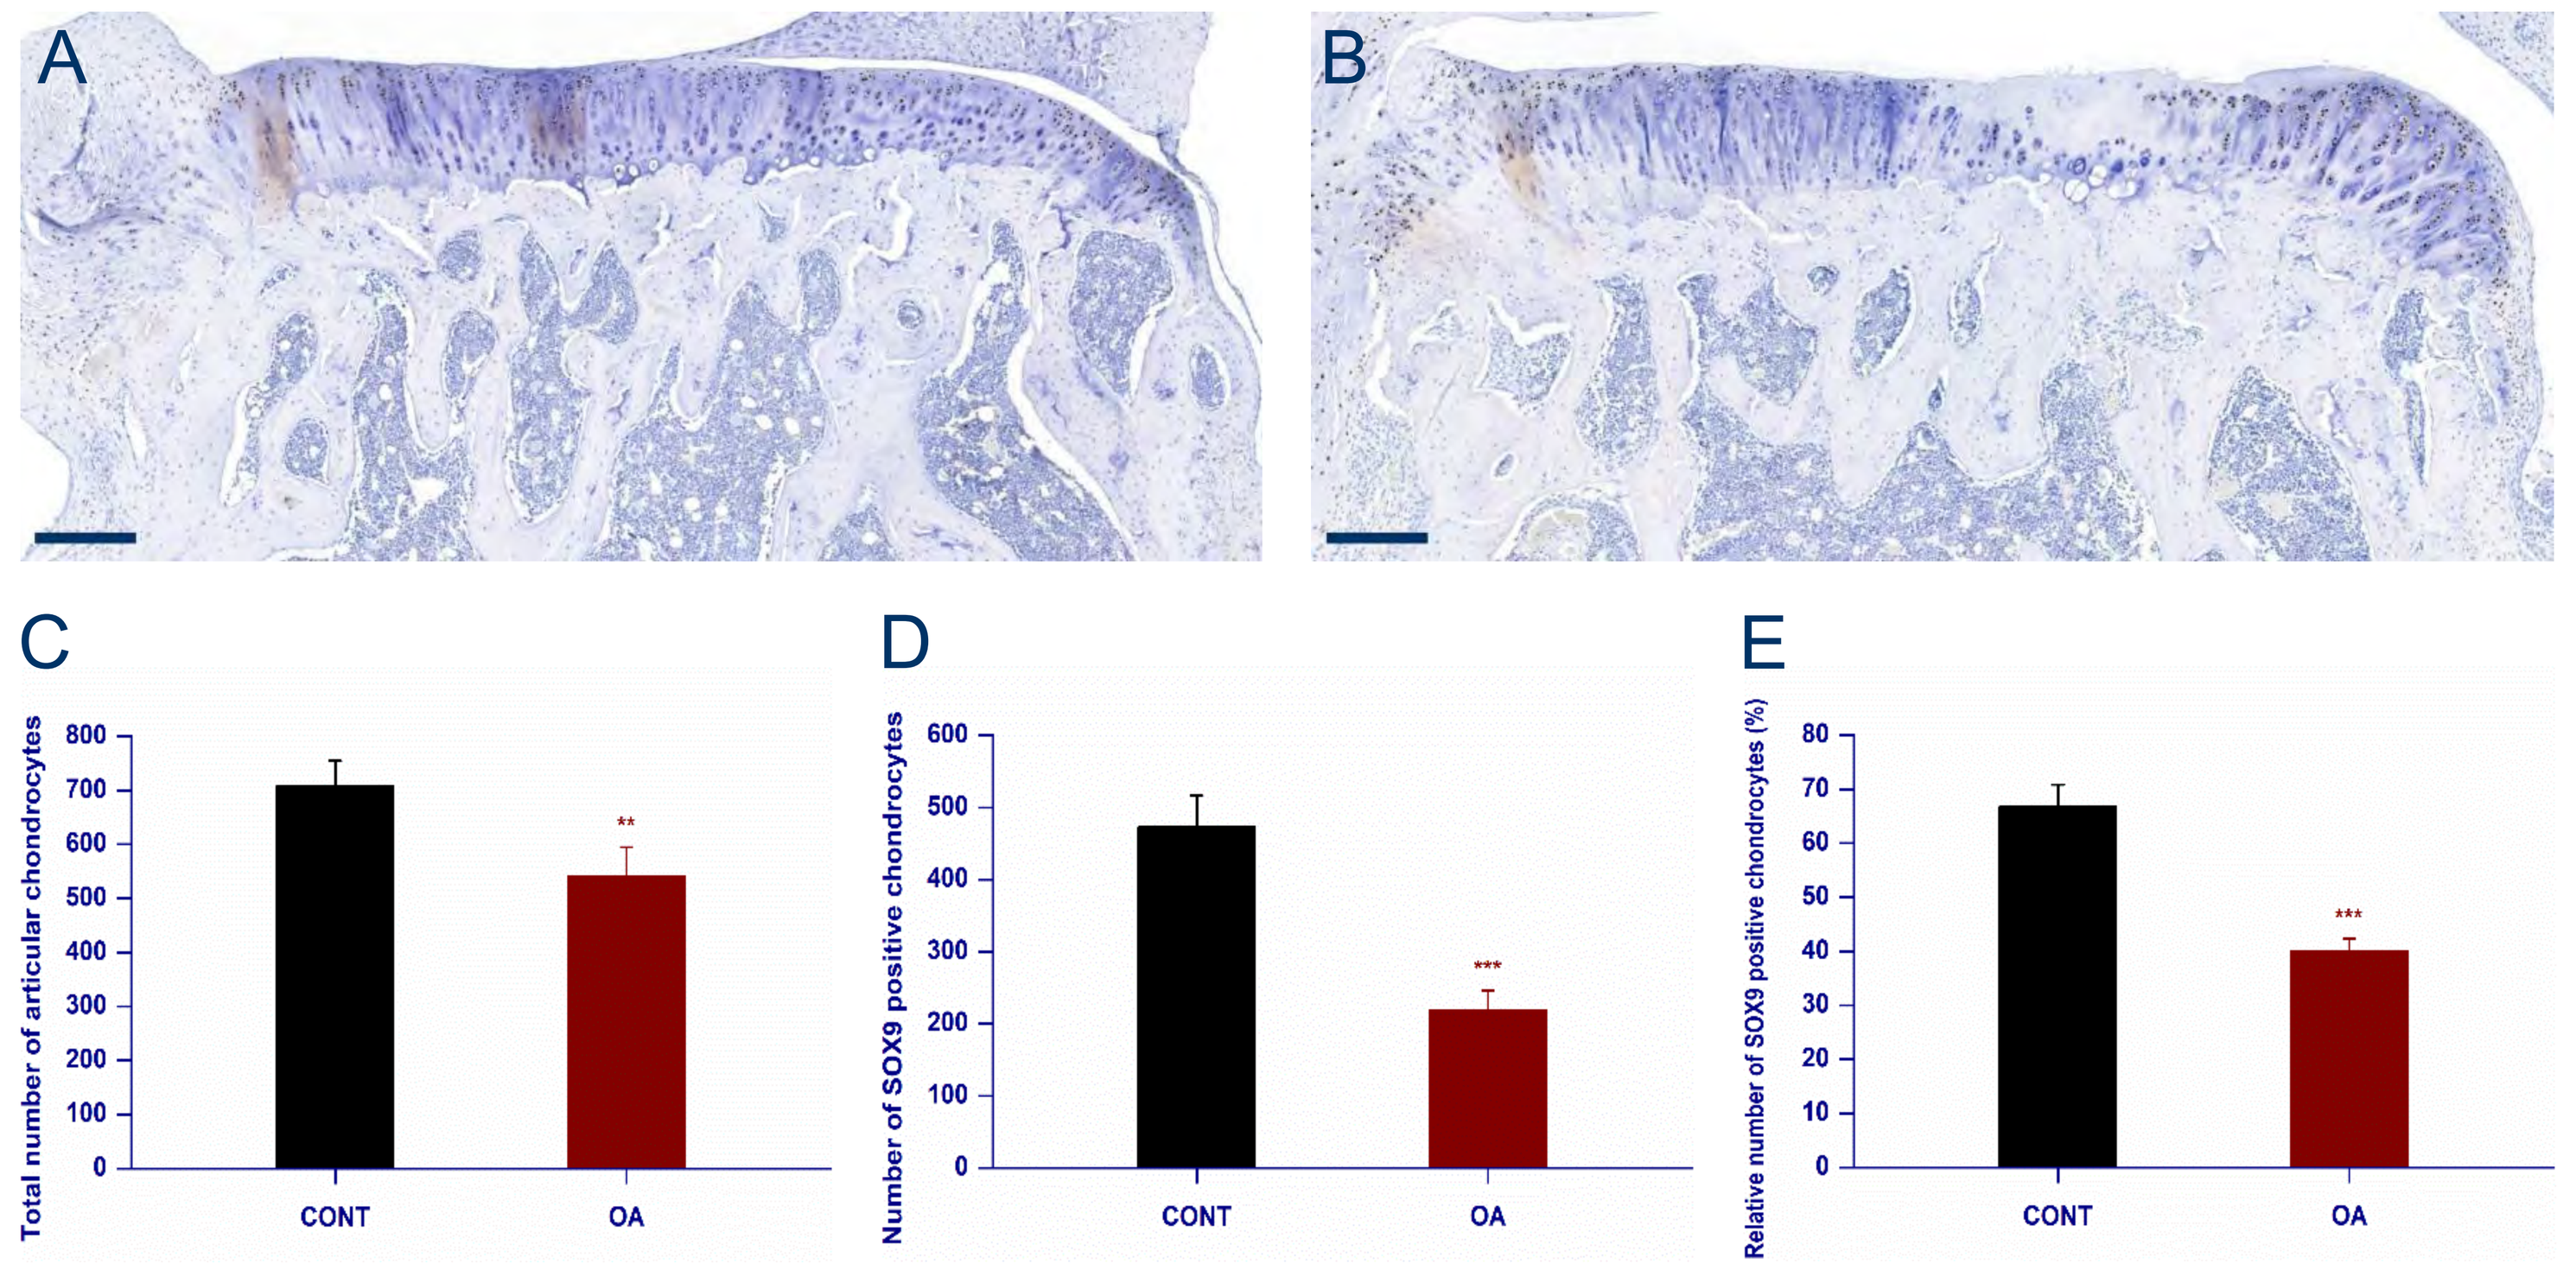
A
B
C
Total number of articular chondrocytes
D
Number of SOX9 positive chondrocytes
E
Relative number of SOX9 positive chondrocytes (%)
CONT
OA

FIGURE 4. Distribution of transcription factor SOX9 immunostaining in the articular cartilage of medial tibial plateau in ipsilateral right knee. (A) Representative histological image of intact control rats and (B) MMT + MCLT operated rats. A bar in panels A and B corresponds to 250 µm. (C) Total number of chondrocytes in existing articular cartilage. (D) Number of chondrocytes stained with SOX9 antibodies in articular cartilage. (E) Relative number of chondrocytes stained with SOX9 antibodies in articular cartilage (%). CONT = intact control rats; OA = MMT + MCLT operated rats; Red asterisks = significance between MMT + MCLT and intact control rats; ** $p < 0.01$; *** $p < 0.001$

Summary

- MMT + MCLT operation decreased body weight and paw withdrawal threshold of operated hind limb during the in-life phase indicating static mechanical/tactile allodynia (Fig. 1*A*,*C*).
- MMT + MCLT operation resulted in the marked degeneration of articular cartilage in the outer 1/3 of medial tibial plateau and in the moderate degeneration in the middle 1/3 (Fig. 2*E*,*F*,*G*).
- MMT + MCLT induced degeneration of articular cartilage was observed in the superficial layer, midzone and tidemark of medial tibial plateau, and included decreased intensity of Toludine blue staining indicating decreased proteoglycan content, the loss of cartilaginous matrix, and a mild damage of calcified cartilage and subchondral bone (Fig. 2*C*,*D*,*H*).
- MMT + MCLT operation led to the development of large osteophytes and mild synovial inflammation in the medial compartment of operated knee (Fig. 2*H*).
- MMT + MCLT operation resulted in decreased relative area of type II collagen immunostaining in existing articular cartilage indicating decreased amount of the major cartilaginous collagen in the medial tibial plateau of operated knee (Fig. 3*E*).
- MMT + MCLT operation led to decreased relative number of SOX9 positive chondrocytes indicating decreased expression of the major regulator of chondrocyte differentiation in articular cartilage in the medial tibial plateau of operated knee (Fig. 4*E*).

Conclusions

This study demonstrated neuropathic pain and various degenerative changes at the tissue, cellular and molecular levels in the rat model of post-traumatic knee OA. In preclinical efficacy studies, the assessment of OA-related knee pain and the histopathological OA evaluation could be completed with IHC and semi-quantification of immunostaining distribution to evaluate the effects of treatments at the molecular level in more detailed.

Disclosures

This work was funded by Astellas Pharma Inc.

References

1. Poole R, Blake S, Buschmann M, Goldring S, Laverty S, *et al*. Osteoarthritis Cartilage 2010;18:S10-6.
2. Martel-Pelletier J, Lukas M, Pelletier J.-P. Bone 2012:51:297-311.
3. Gerwin N, Bendele AM, Glasson S, Carlson CS. Osteoarthritis Cartilage 2010;18:S24-34.
4. Morko J, Peng ZQ, Vääräniemi J, *et al*. J Orthop Res. 2016;34:Suppl1:0533 (abstract).
5. Janusz MJ, Bendele AM, Brown KK, Taiwo YO, *et al*. Osteoarthritis Cartilage. 2002;10:785-91.

www.pharmatest.com